Induction of immunogenic cell death and tumor regression in murine animal models by a novel cytolytic compound, LTX-401

5128

BRYNJAR MAUSETH[1,2,9], JI-HUA SHI[2], KETIL CAMILIO[3], LIV-MARIE EIKE[4], HENG ZHOU[5], ALLAN SAUVAT[5], LÍGIA C GOMES-DA-SILVA[5], SYLVÈRE DURAND[5], SABRINA FORVEILLE[5], KRISTINA IRIBARREN[5], TAKAHIRO YAMAZAKI[6], SYLVIE SOUQUERE[7], LUCILLIA BEZU[5], KEVIN MÜLLER[5], MARION LEDUC[5], PENG LIU[5], LIWEI ZHAO[5], AURÉLIEN MARABELLE[8], LAURENCE ZITVOGEL[6], OLIVER KEPP[5], GUIDO KROEMER[5], ØYSTEIN REKDAL[4,9], PÅL-DAG LINE[1,2], BALDUR SVEINBJØRNSSON[4,9]

[1]Institute of Clinical Medicine, University of Oslo, Oslo, Norway, [2]Division of Cancer, Surgery and Transplantation, Oslo University Hospital, Rikshospitalet, Oslo, Norway, [3]Institute for Cancer Research, Department of Tumor Biology, Oslo University Hospital, Oslo, Norway, [4]Department of Medical Biology, University of Tromsø, Norway, [5]Metabolomics and Cell Biology Platforms, Gustave Roussy Comprehensive Cancer Institute, Paris, France [6]University of Paris Sud XI, Paris, France, [7]Gustave Roussy Comprehensive Cancer Center, Paris, France, [8]Institut National de la Santé et de la Recerche Medicale, Paris, France, [9]Lytix Biopharma, Oslo, Norway

Lytix Biopharma

Background

Structure-activity relationship studies on host defense peptides have allowed us to design smaller derivatives with bulky and lipophilic moieties for improved anticancer activity. This work has culminated in the engineering of LTX-401, a cytolytic immunotherapeutic agent designed for intratumoral injection. Intratumoral drug administration offers several advantages over standard systemically given therapies, including increased drug penetration, avoidance of circulatory degradation, less systemic exposure/toxicity and potential depletion of intratumoral immunosuppressive cells.

Because of the hydrophobicity and positive charge, LTX-401 may interact electrostatically with anionic cell membrane components of cancer cells. The swift LTX-401-induced cancer cell death is likely attributed to a membranolytic mechanism of action resulting in necrosis, followed by the release of immune-stimulating 'danger signals' or DAMPs that may potentiate anti-tumor immunity.

The *in vivo* efficacy of LTX-401 has been evaluated in several animal models, including the B16 mouse melanoma, the MCA205 mouse fibrosarcoma and the JM1 rat hepatocellular carcinoma model, showing tumor regression and long-term protective effects.

Aim

This study was undertaken to investigate the anticancer effects and mode of action by LTX-401 *in vitro* and in rodent tumor models.

LTX-401

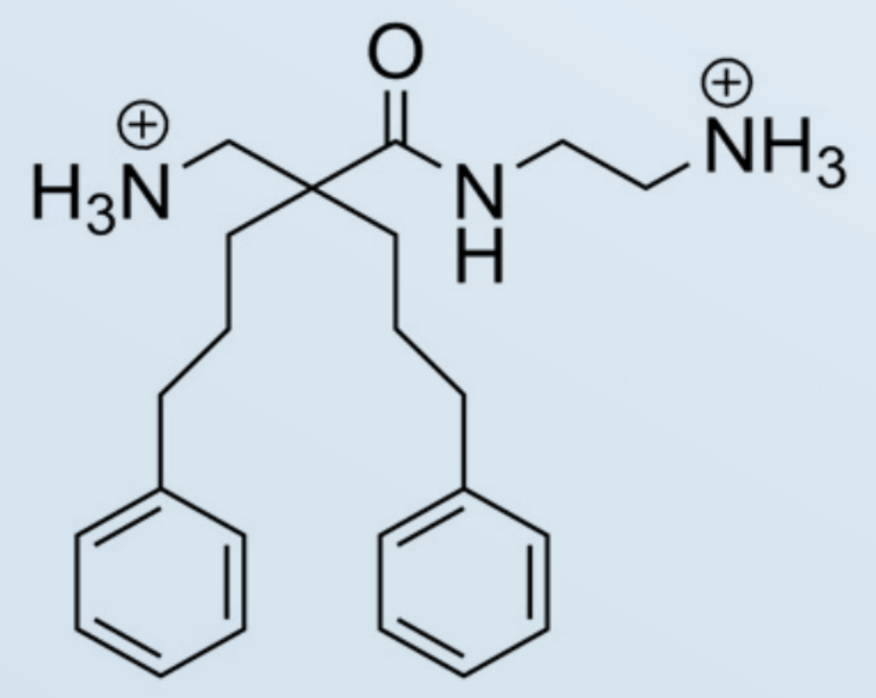

Chemical structure of LTX-401 (N-(2-aminoethyl)-2-(aminomethyl) -5-phenyl-2-(3-phenylpropyl) pentanamide), a $\beta^{2,2}$-amino acid derivative.

Results

Table 1 LTX-401 is highly active against a panel of malignant cells and displays rapid kill kinetics

Cell lines	LTX-401 IC_{50}[1] ± SD (μM)
JM1 (Rat hepatocellular carcinoma)	22.8 ± 3.3
HEPG2 (Human hepatocellular carcinoma)	35.4 ± 0.6
BEL7402 (Human hepatocellular carcinoma)	26.7 ± 0.4
B16F1 (Murine skin malignant melanoma)	23.3 ± 3.9
MDA-MB-435S (Human malignant melanoma)	13.5 ± 1.4
Malme-3M (Human malignant melanoma)	19.3 ± 0.4
HT-29 (Human colorectal adenocarcinoma)	31.7 ± 2.9
A375 (Human skin malignant carcinoma)	30 ± 0.9
SK-N-AS (Human neuroblastoma)	30.6 ± 0
MRC-5 (Human lung fibroblasts)	22.9 ± 1.4
HUV-EC-C (Human vascular endothelium)	18.4 ± 1.9
HaCat (Human keratinocytes)	22 ± 2.2
RBC (Human)	>1087[2]

[1] The peptide concentration killing 50 % of the cells
[2] Highest tested concentration before 50 % lysis of RBCs was 400 μg/ml (1087 μM)

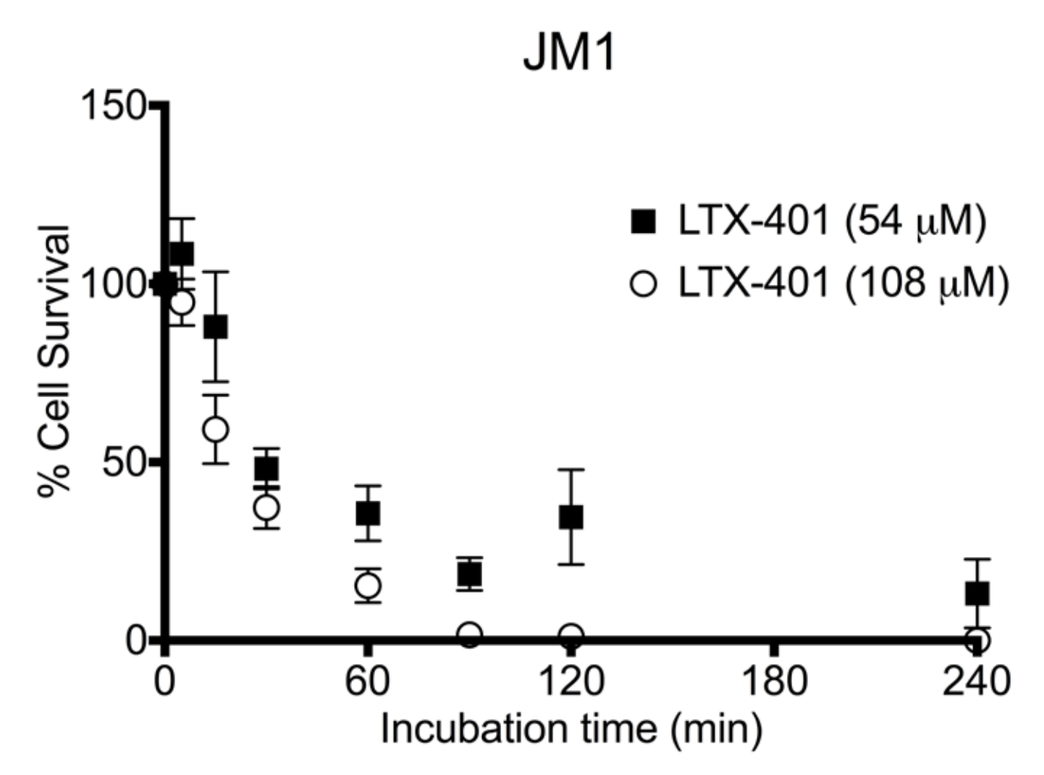
JM1
% Cell Survival
LTX-401 (54 μM)
LTX-401 (108 μM)
Incubation time (min)

Fig. 1 Ultrastructural characteristics of LTX-401-induced cell death

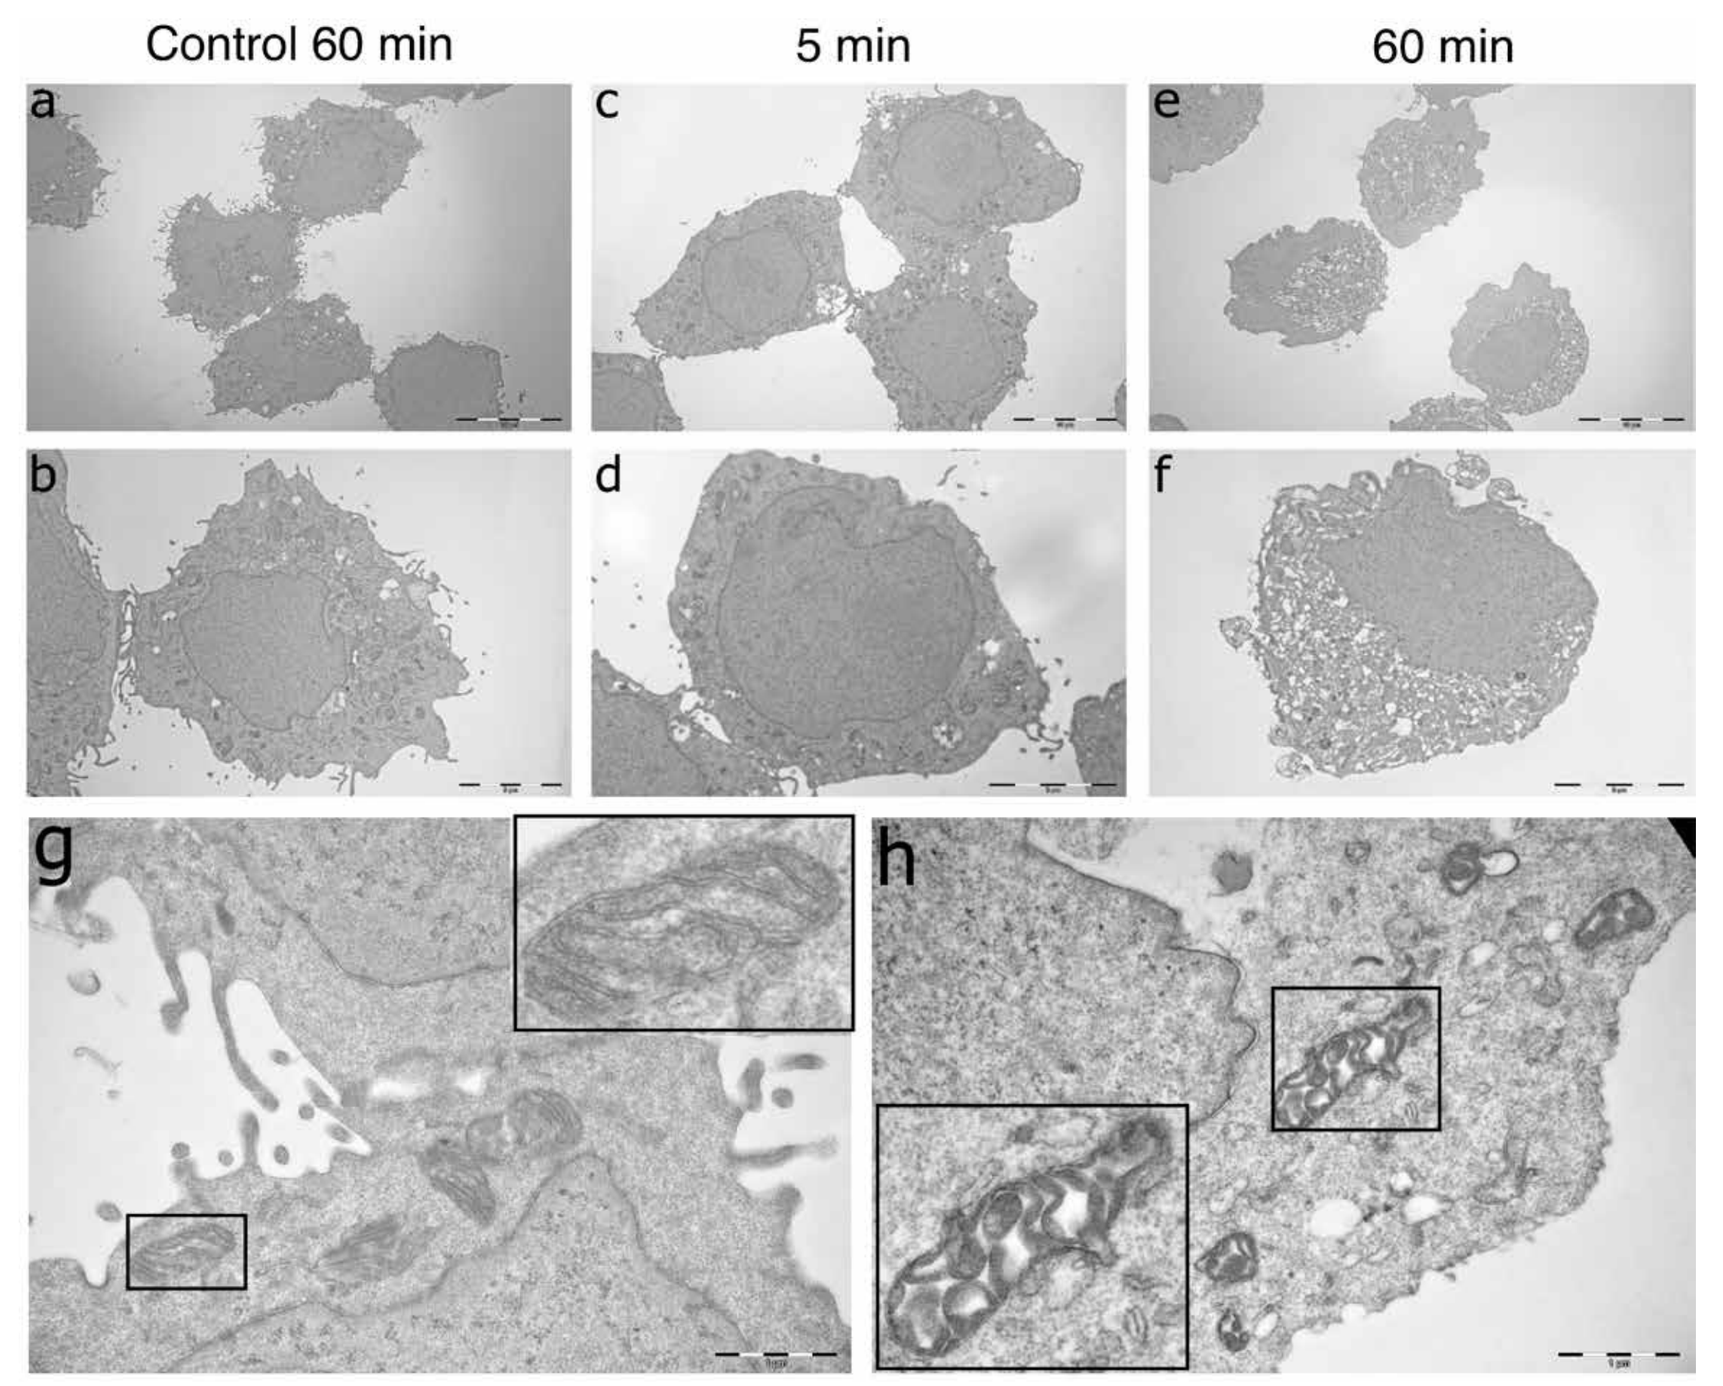
Control 60 min
5 min
60 min

Representative TEM micrographs of JM1 cells treated with 108 μM (4 x IC_{50}^{4h} value) of LTX-401 for various time points. Untreated cells (**a, b**) were kept in serum-free RPMI 1640 and compared with cell treated with LTX-401 for 5 min (**c, d**) and 60 min (**e, f**). Bottom panels; mitochondrial morphology in untreated (**g**) compared to LTX-401-treated cells (**h**).

Fig. 2 Brefeldin A partially inhibits the cytotoxic potential of LTX-401-treatment

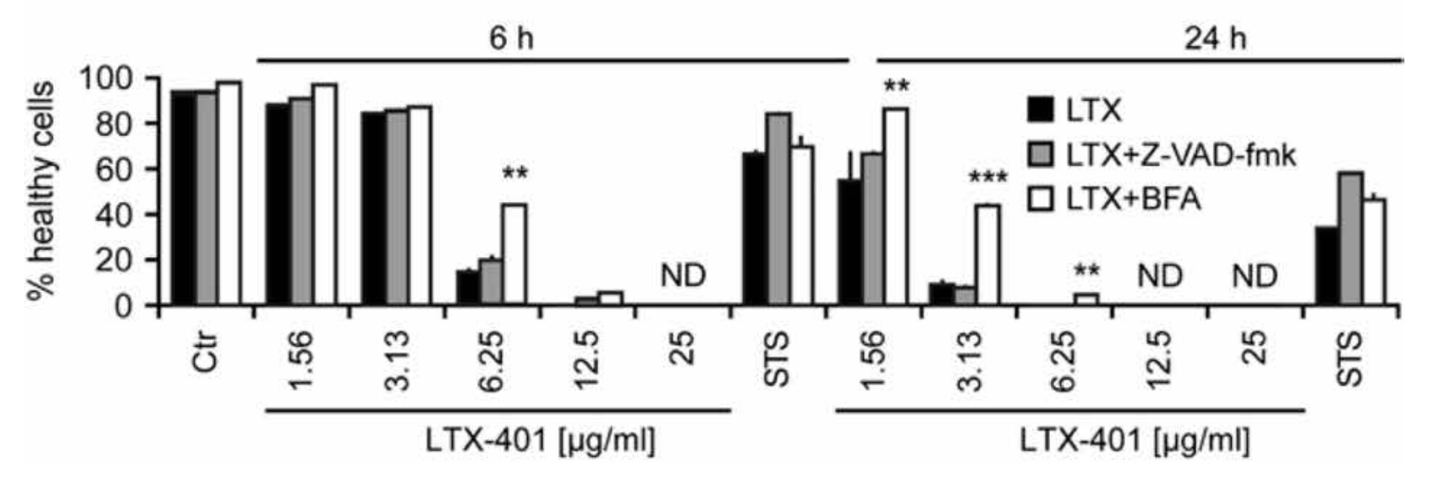
6 h
24 h
% healthy cells
LTX
LTX+Z-VAD-fmk
LTX+BFA
ND
LTX-401 [μg/ml]

U2OS cells were treated with the indicated concentrations of LTX-401, in the absence or presence of 10 μg/ml BFA, or the pan-caspase inhibitor Z-VAD-fmk (50 μM) and measured for cell survival. **P<0.01; ***P<0.001

Fig. 3 Treatment with LTX-401 induces loss of lysosomal integrity

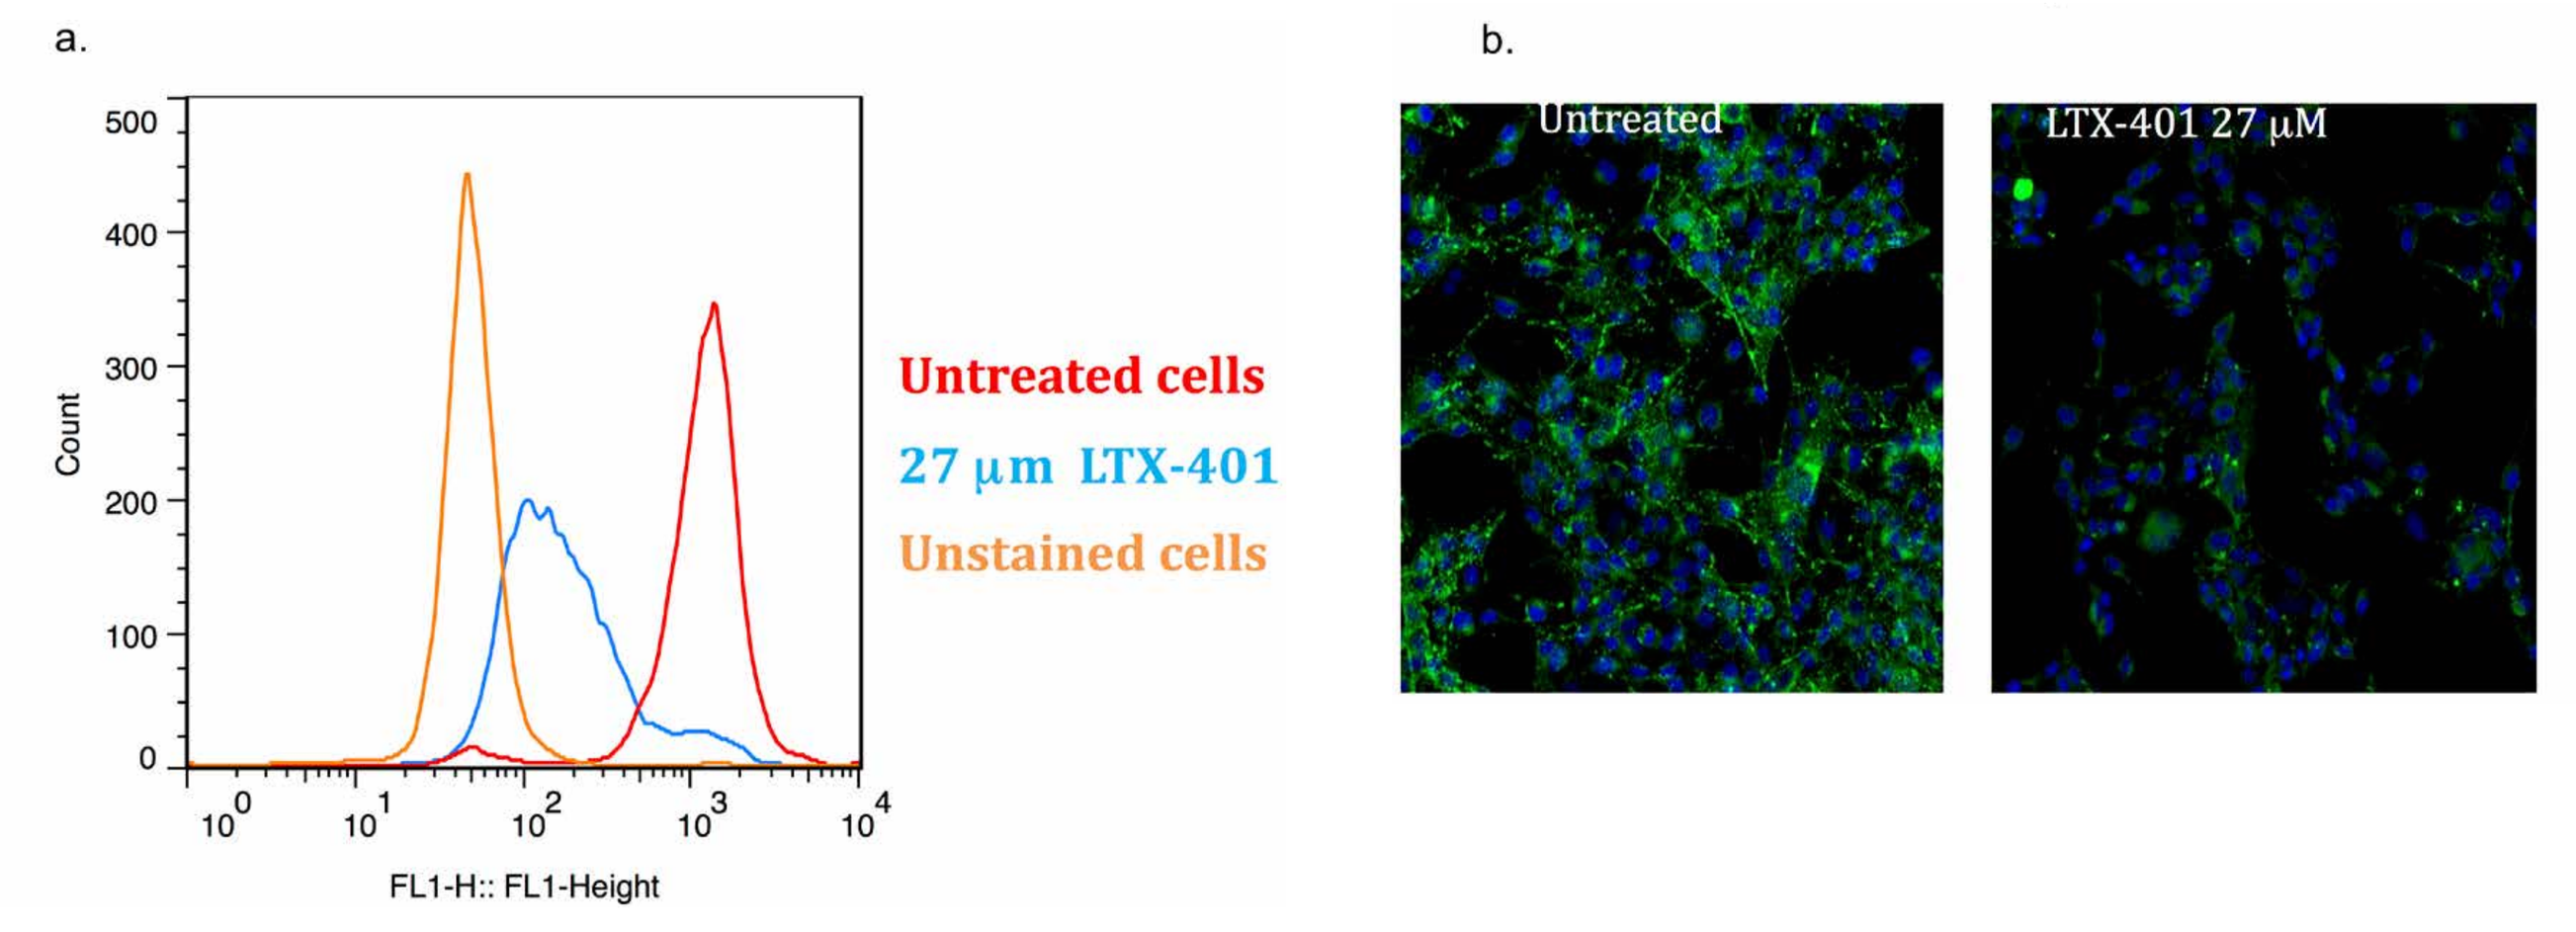
a.
b.
Count
Untreated cells
27 μm LTX-401
Unstained cells
FL1-H:: FL1-Height
Untreated
LTX-401 27 μM

B16F1 cells were treated with the 1 x IC_{50}^{4h} value of LTX-401 (27 μM) for 60 minutes before determining the signal strength from LysoTracker using flow cytometry (**a**) and confocal microscopy (**b**).

Fig. 4 LTX-401-induced cancer cell death promotes the release of endogenous 'danger signals' such as High-Mobility Group Box 1 (HMGB1) protein, ATP and cytochrome C *in vitro*

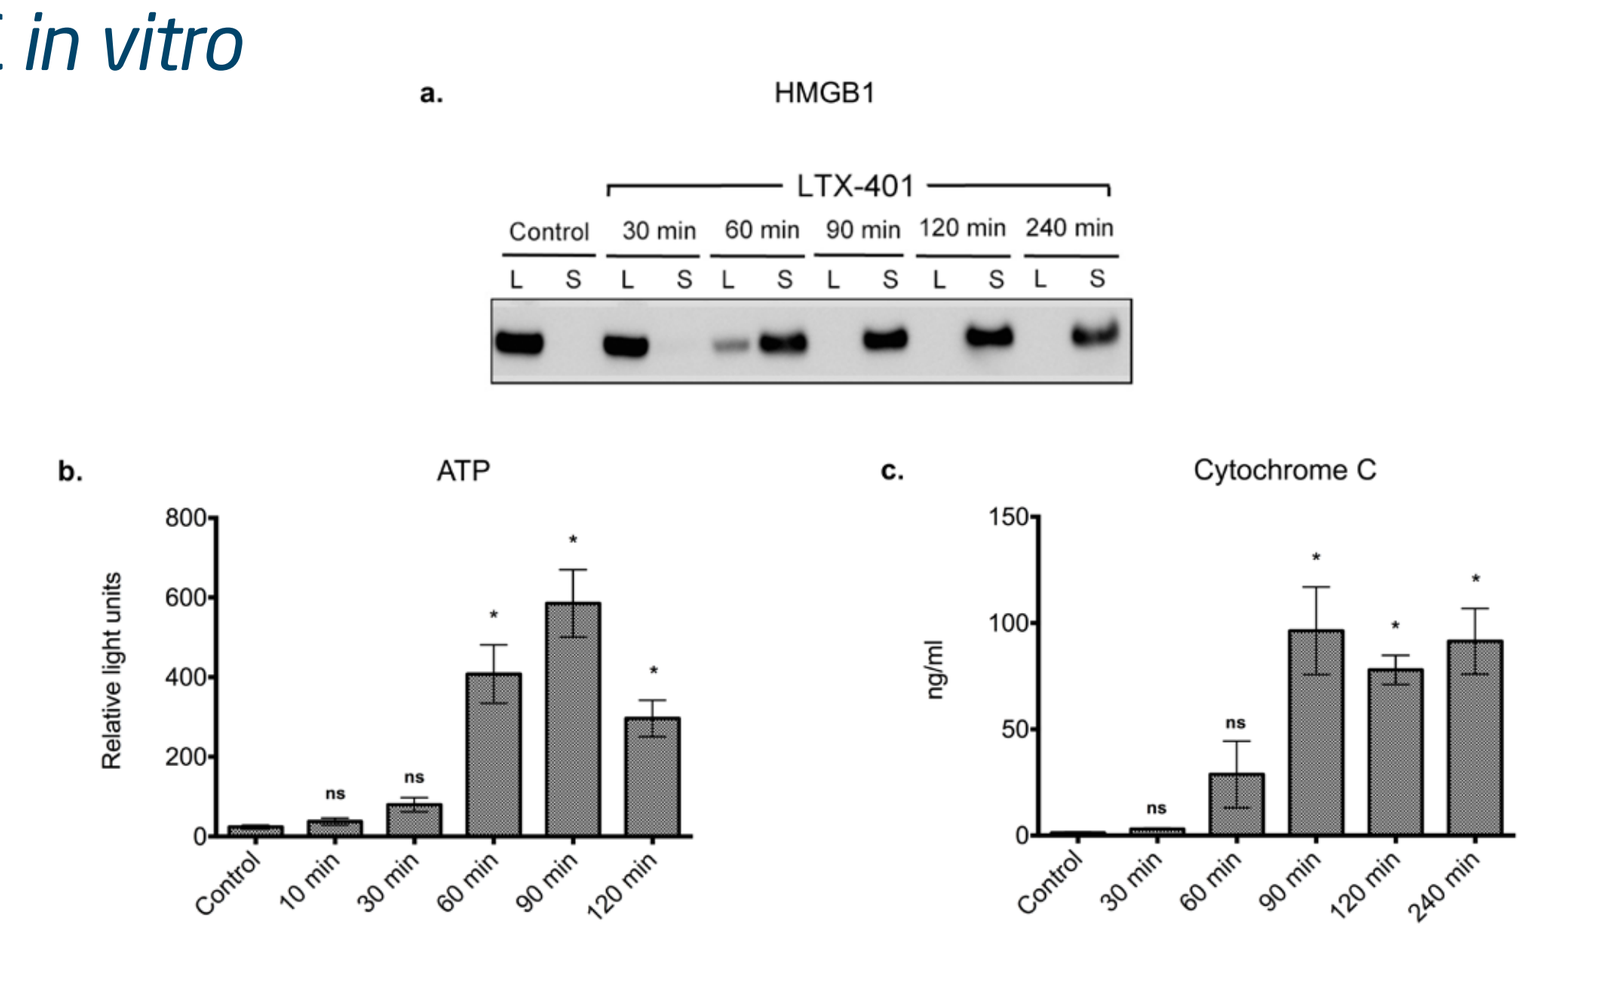
HMGB1
LTX-401
Control
ATP
Relative light units
Cytochrome C
ng/ml

(**a**) Translocation of HMGB1 from the lysate (L) to supernatant (S) of JM1 cells after being treated with the 4 x IC_{50}^{4h} of LTX-401 (108 μM). (**b**) JM1 cells release ATP following treatment with the 2 x IC_{50}^{4h} of LTX-401 (54 μM). (**c**) Cytochrome c is released from JM1 cells after treatment with the 4 x IC504h of LTX-401 (108 μM). *P<0.05, ns=not significant

Fig. 5 *In vivo* efficacy of LTX-401 against B16 melanomas and JM1 hepatomas

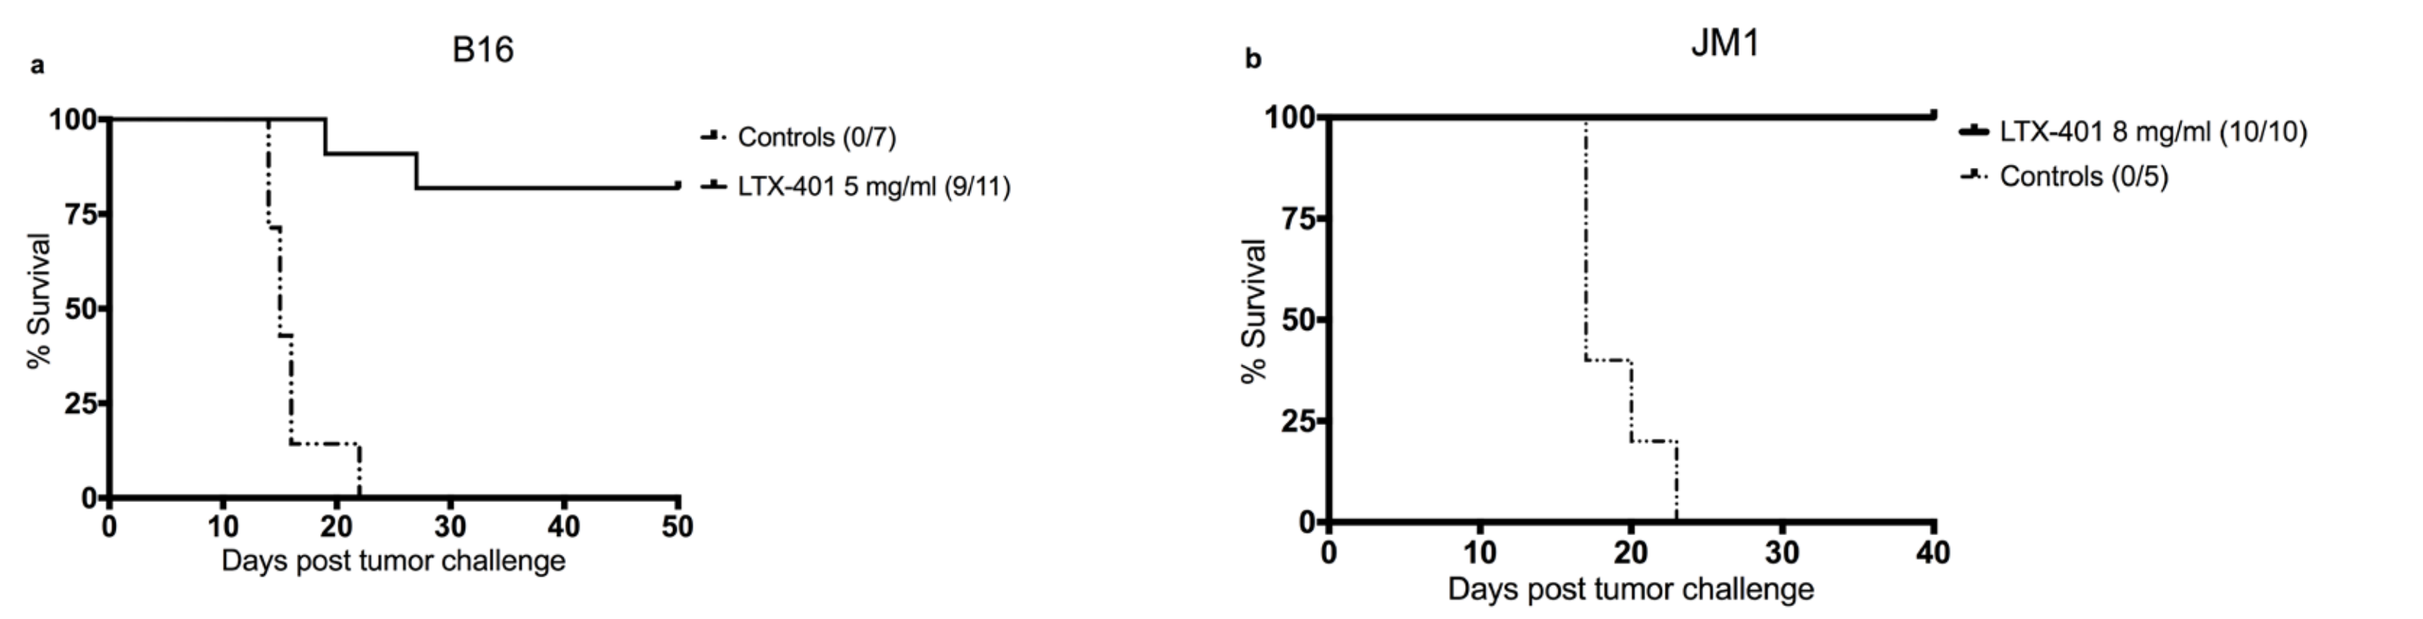
B16
Controls (0/7)
LTX-401 5 mg/ml (9/11)
% Survival
Days post tumor challenge
JM1
LTX-401 8 mg/ml (10/10)
Controls (0/5)

Survival curves of (**a**) B16 melanomas and (**b**) JM1 hepatomas (subcutaneous) injected with either sterile 0.9% NaCl (vehicle control) or 0.25 mg LTX-401 (B16)/0.4 mg LTX-401 (JM1) once per day for three consecutive days.

Fig. 6 LTX-401 treatment induces infiltration of immune cells

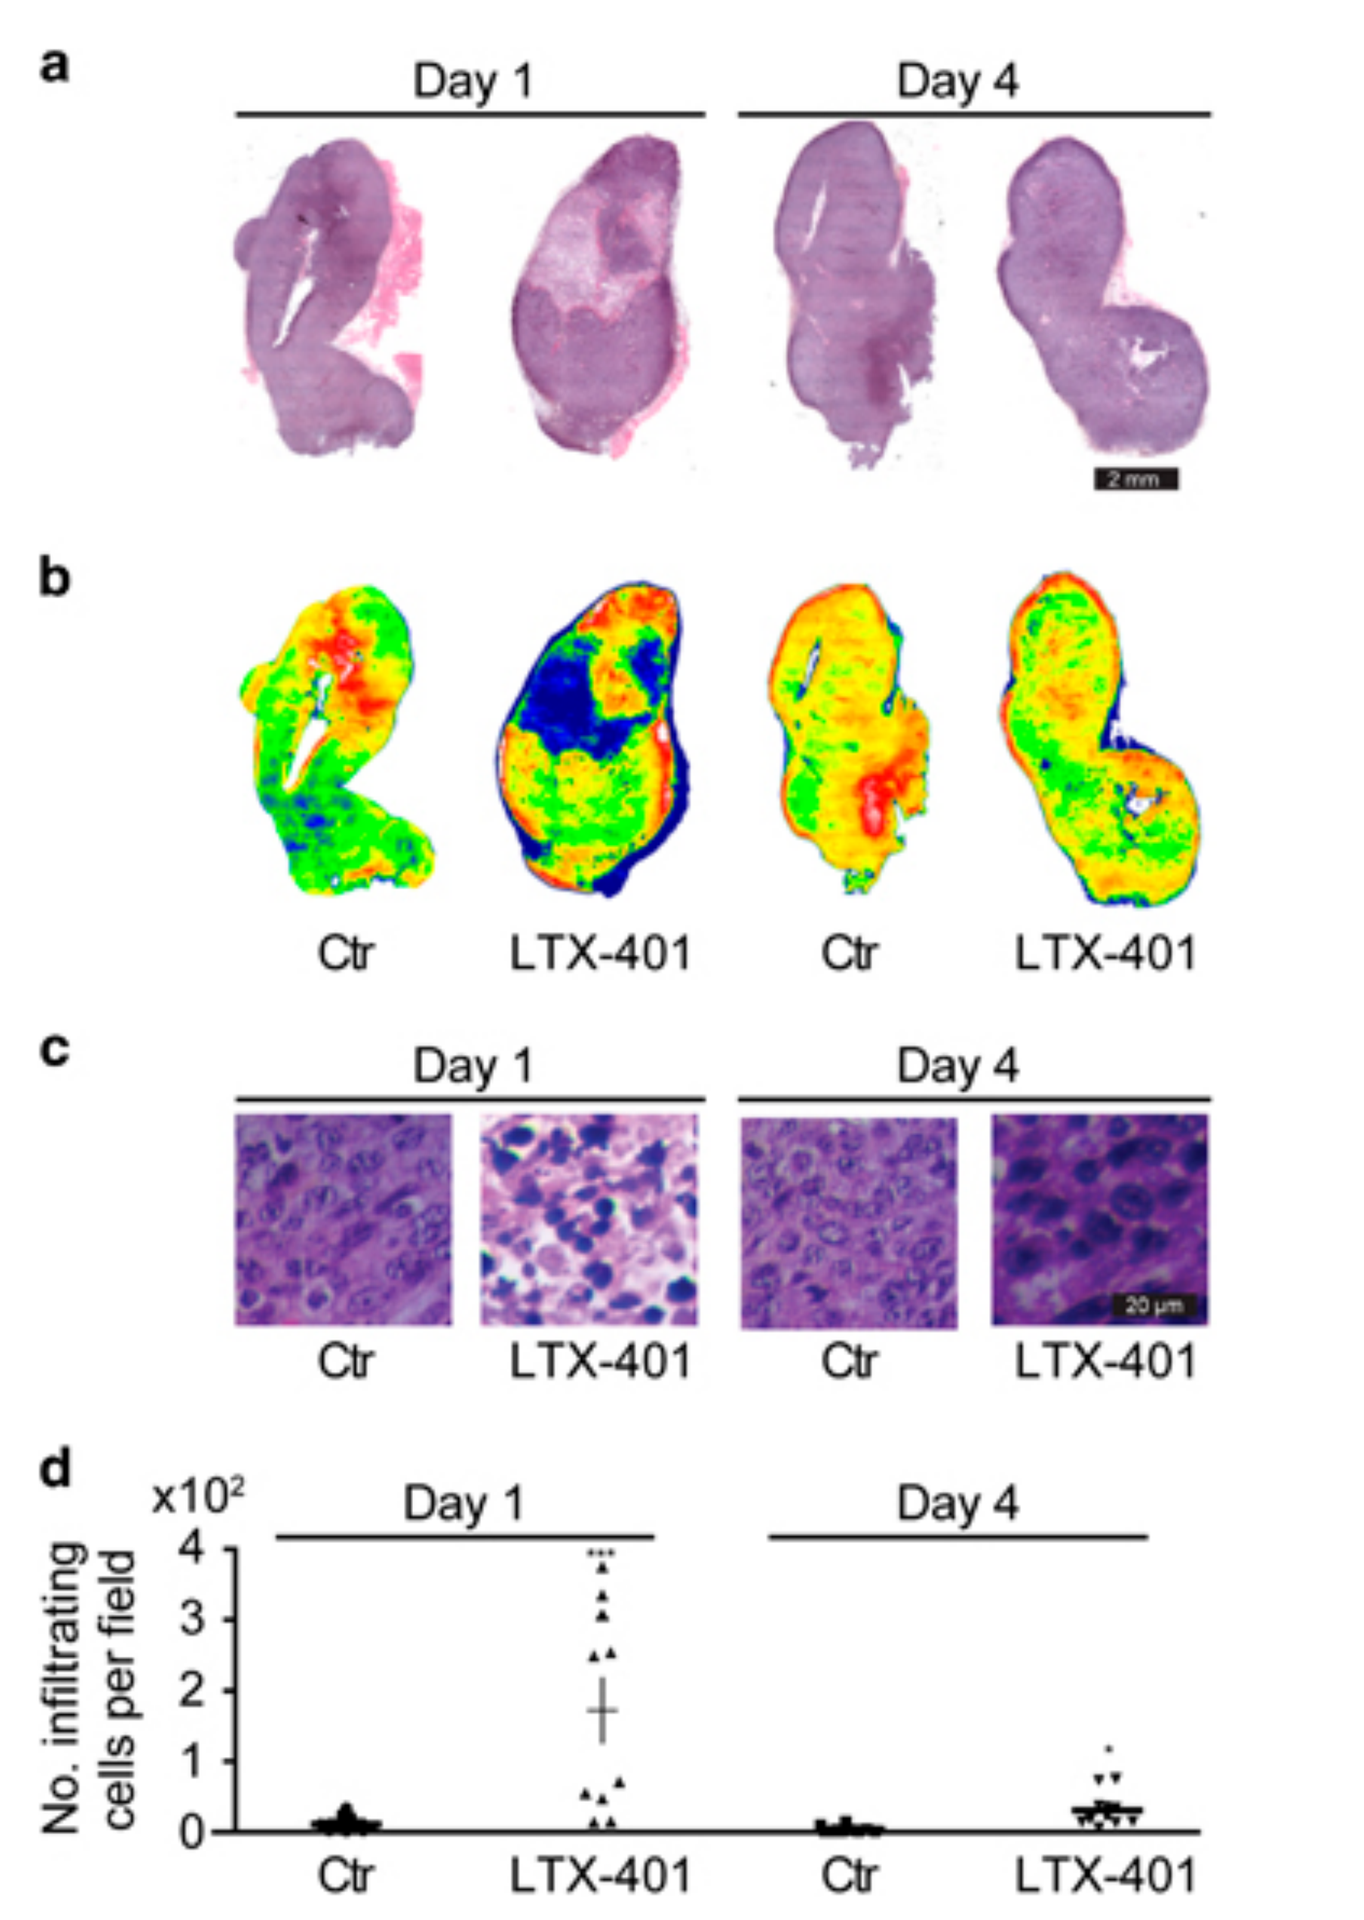
Day 1
Day 4
Ctr
LTX-401
No. infiltrating cells per field

MCA205 fibrosarcoma was established on C57/Bl6 mice and injected with PBS (control, Ctr) or LTX-401. Tumors were harvested at day 1 or 4 later and were photographed after excision to document their macroscopic appearance (**a**) and subjected to hematoxilin and eosin (HE) staining (raw appearance in (**a**), ratio of eosin over hematoxilin in (**b**)). Representative HE staining patterns of necrotic areas are shown in (**c**) and the number of infiltrating leukocytes per view field was determined in (**d**). Asterisks indicate significant differences (unpaired Student's t-test) with respect to PBS-treated controls. *P<0.05; **P<0.01; ***P<0.001

Conclusions

- LTX-401 induced membranolytic cancer cell death, accompanied by massive vacuolization of the cytoplasm and release of intracellular 'danger signals'.
- LTX-401 acts on and selectively destroys intracellular organelles such as mitochondria, lysosomes and Golgi apparatus
- Intratumoral injection with LTX-401 induced infiltration of immune cells followed by complete regression of syngeneic B16 melanomas and JM1 hepatomas, thus supporting the rationale for further evaluation of the compound as an immunotherapeutic agent against solid tumors.

References

Eike LM, Mauseth B, Camilio KA, Rekdal O, Sveinbjornsson B. The Cytolytic Amphipathic beta(2,2)-Amino Acid LTX-401 Induces DAMP Release in Melanoma Cells and Causes Complete Regression of B16 Melanoma. Plos One. 2016;11(2):e0148980.

Zhou H, Sauvat A, Gomes-da-Silva LC, Durand S, Forveille S, Iribarren K, et al. The oncolytic compound LTX-401 targets the Golgi apparatus. Cell Death Differ. 2016;23(12):2031-41.

UNIVERSITY OF TROMSØ

GUSTAVE ROUSSY CANCER CAMPUS GRAND PARIS

Oslo University Hospital

UiO : University of Oslo

CHILI. © WWW.CHILIGROUP.NO | 2017

Lytix Biopharma AS | P.O. Box 6447 | NO-9294 Tromsø, Norway | E-mail: post@lytixbiopharma.com | Phone: +47 77 67 55 00 | Fax: +47 77 67 55 01