A2-030

LTX-109 is Active against CA-MRSA (USA300) and *S. pyogenes* in a Mouse Skin Infection Model

W.M. OLSEN[1], B. MORTENSEN[1], J.K. KHAN[2], M. RAHMAN2, J.S. SVENDSEN[1], A. FUGELLI[1]
[1]Lytix Biopharma, Norway, [2]Naeja Pharmaceutical Inc, Alberta, Canada

Lytix Biopharma

LTX-109

- A synthetic protein fragment - a peptidomimetic
- High stability against degradation
- Prooduced synthetically in large scale
- Low cost of synthesis

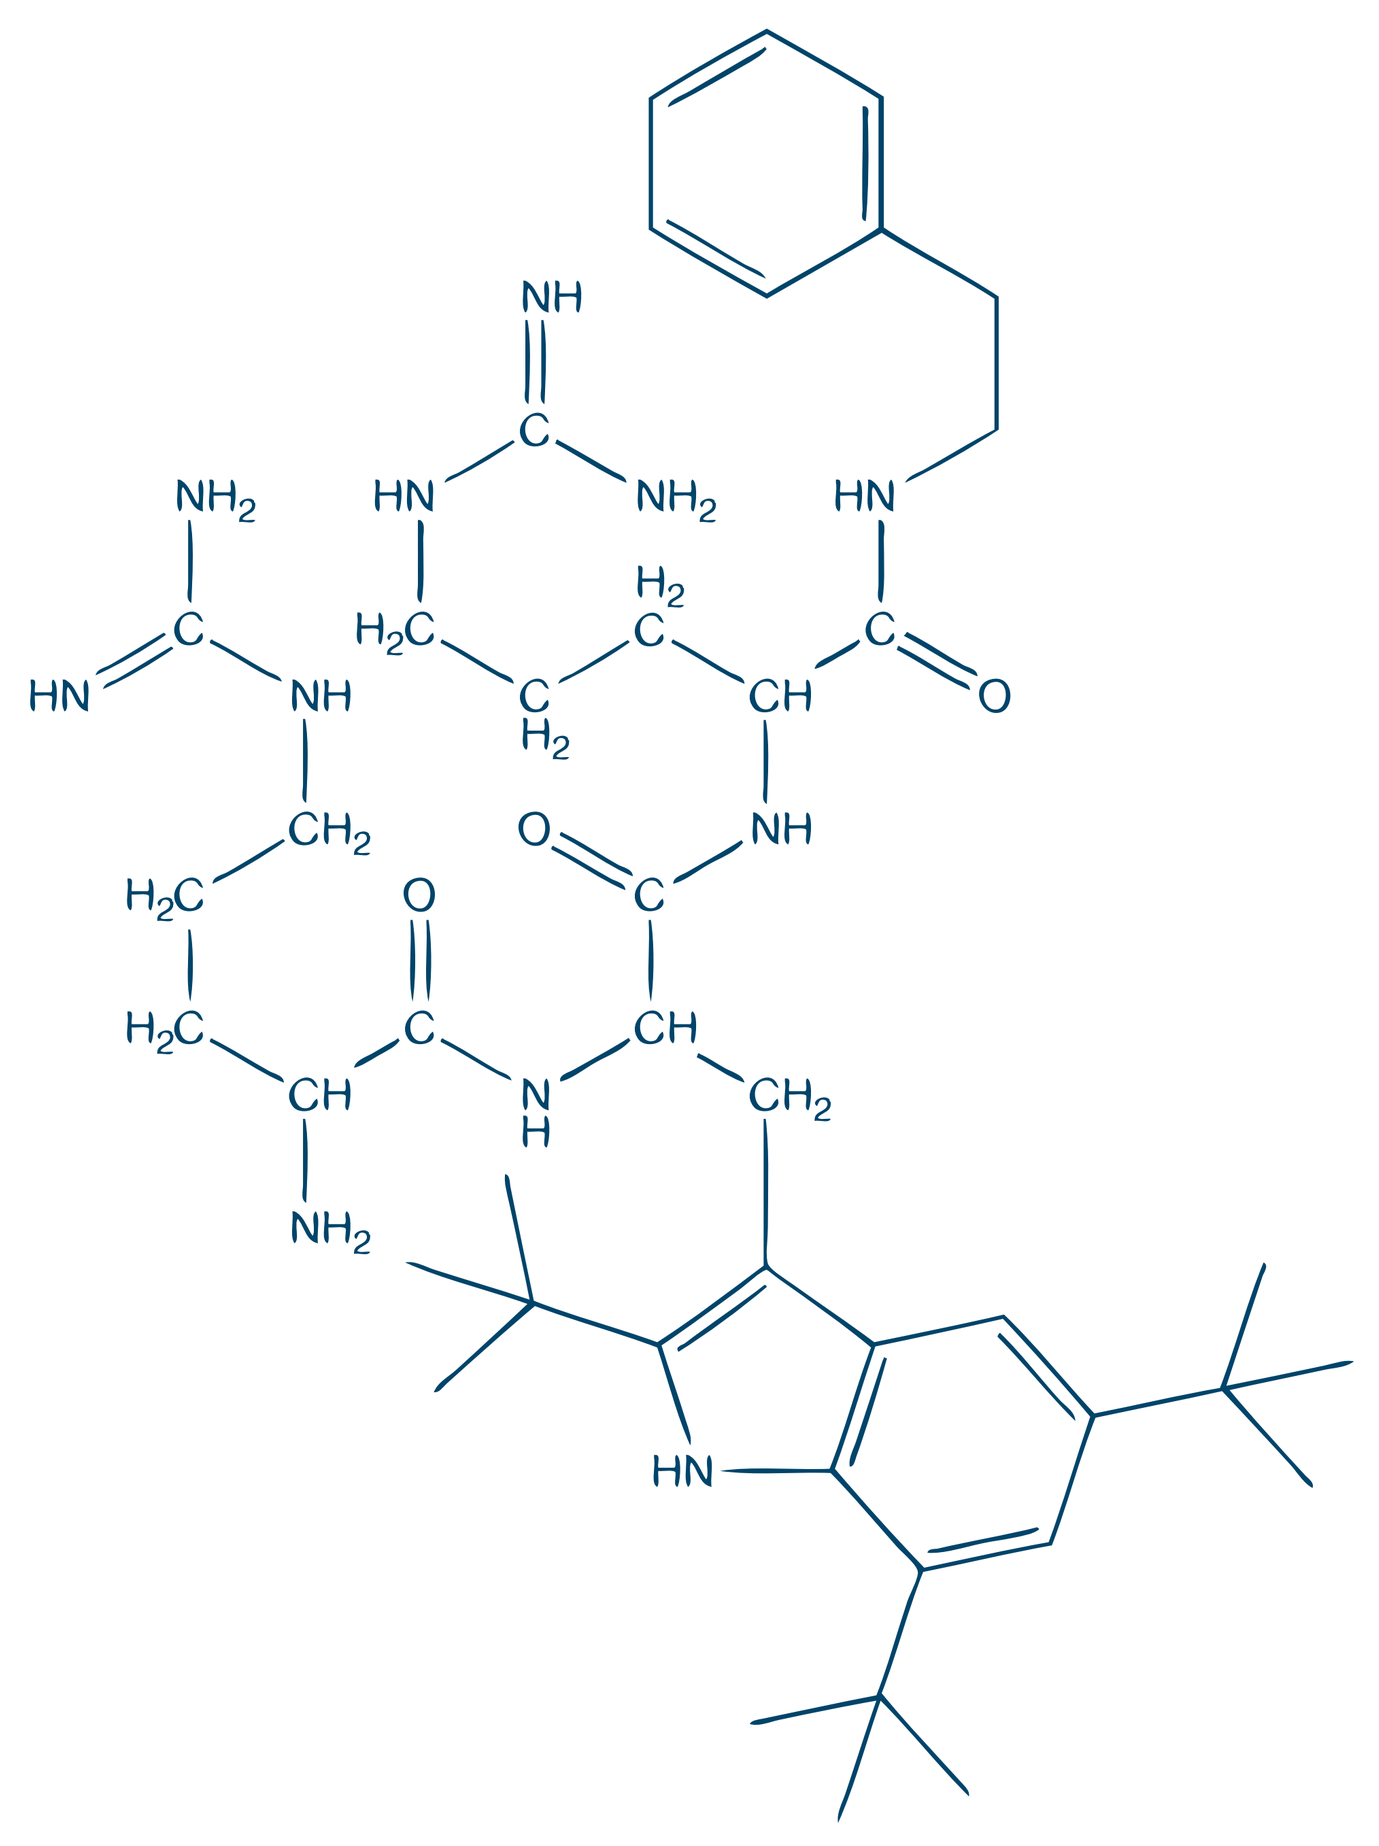

Chemical structure of LTX-109.

Background

LTX-109 is a novel antimicrobial drug in clinical development for skin infections and nasal decolonisation of MRSA. The drug mimics the effects of natural antimicrobial peptides in a synthetic small molecule. LTX-109 has demonstrated a broad activity against several Gram (+) and Gram (-) bacteria *in vitro*, as well activity against a range of yeast and fungal species. The compound is equally effective against antibiotic-resistant species such as methicillin resistant *Staphylococcus aureus* (MRSA), vancomycin resistant *Enterococci* (VRE) and multi-resistant *Pseudomonas* isolates. The ultra-rapid membrane lysing mode of action may result in a lower propensity to resistant development and a rapid bactericidal mechanism of action, as shown in *in vitro* studies. To date LTX-109 demonstrates no *in vitro* cross-resistance with other classes of antibiotics.

The aim of the present studies was to evaluate the effect of LTX-109 against community-acquired MRSA (CA-MRSA) and *Streptococcus pyogenes* in a mouse skin infection model.

Methods

Superficial skin lesions were made by a tape-stripping and scalpel blade-cut injury method. Infection was established by adding bacterial suspensions of CA-MRSA (USA300) or *S. pyogenes*. Groups of mice were treated with LTX-109 22 hrs from the start of the infection. Skin biopsies were sampled and colony forming units (CFU/ml) were determined.

Results

LTX-109 demonstrates a significant reduction of the CA-MRSA USA300 tissue load

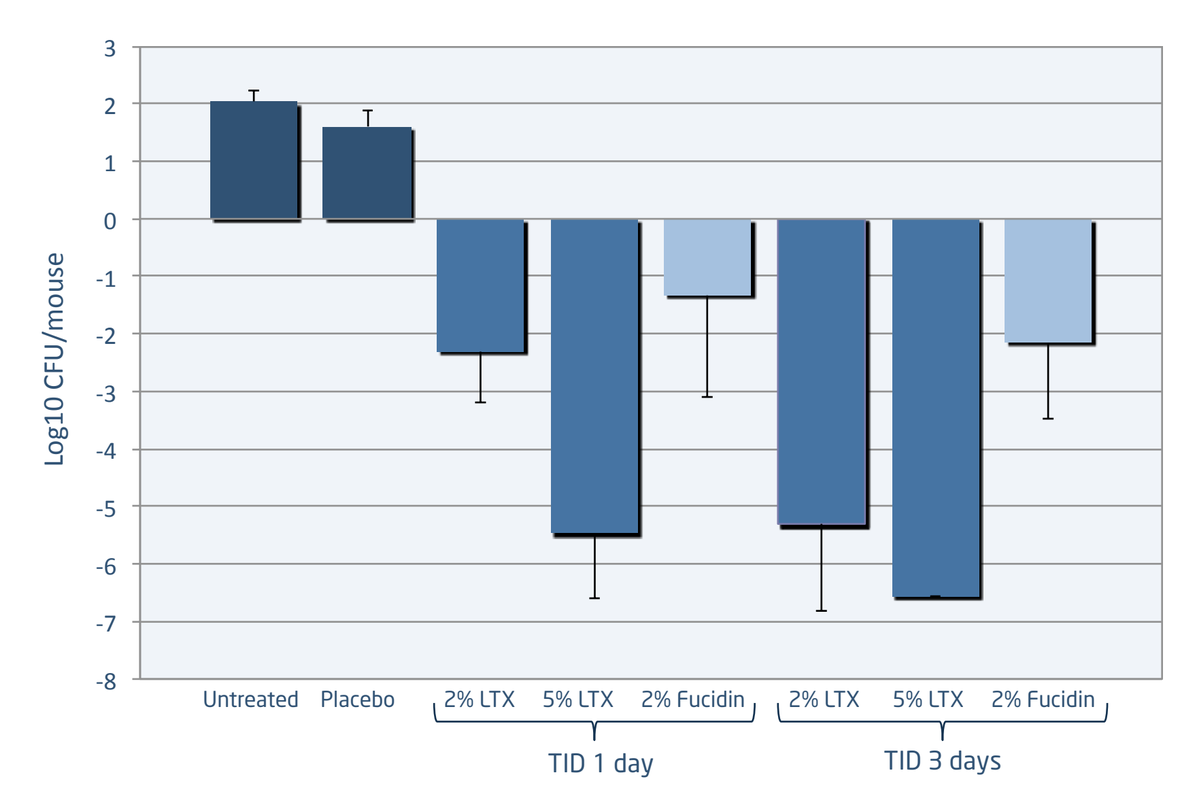
Log10 CFU/mouse
3
2
1
0
-1
-2
-3
-4
-5
-6
-7
-8
Untreated
Placebo
2% LTX
5% LTX
2% Fucidin
2% LTX
5% LTX
2% Fucidin
TID 1 day
TID 3 days

Infection:
Community acquired-MRSA
ATCC BA-1717 (USA300)

Treatment:
TID – one day vs three days

Measure:
Bacterial growth was assessed +9 and +57 hours, respectivly, after first treatment – change in CFU from inoculation load

Result:
Treatment with 1% and 2% LTX-109 was highly effective in reducing bacterial tissue load of CA-MRSA (USA300). A dose-dependent reduction in CFU-counts was observed after one day of treatment. LTX-109 doses of 1% and 2% were significantly better than the comparator Fucidin®

LTX-109 demonstrates a superior effect on CA-MRSA USA300 compared to marketed products

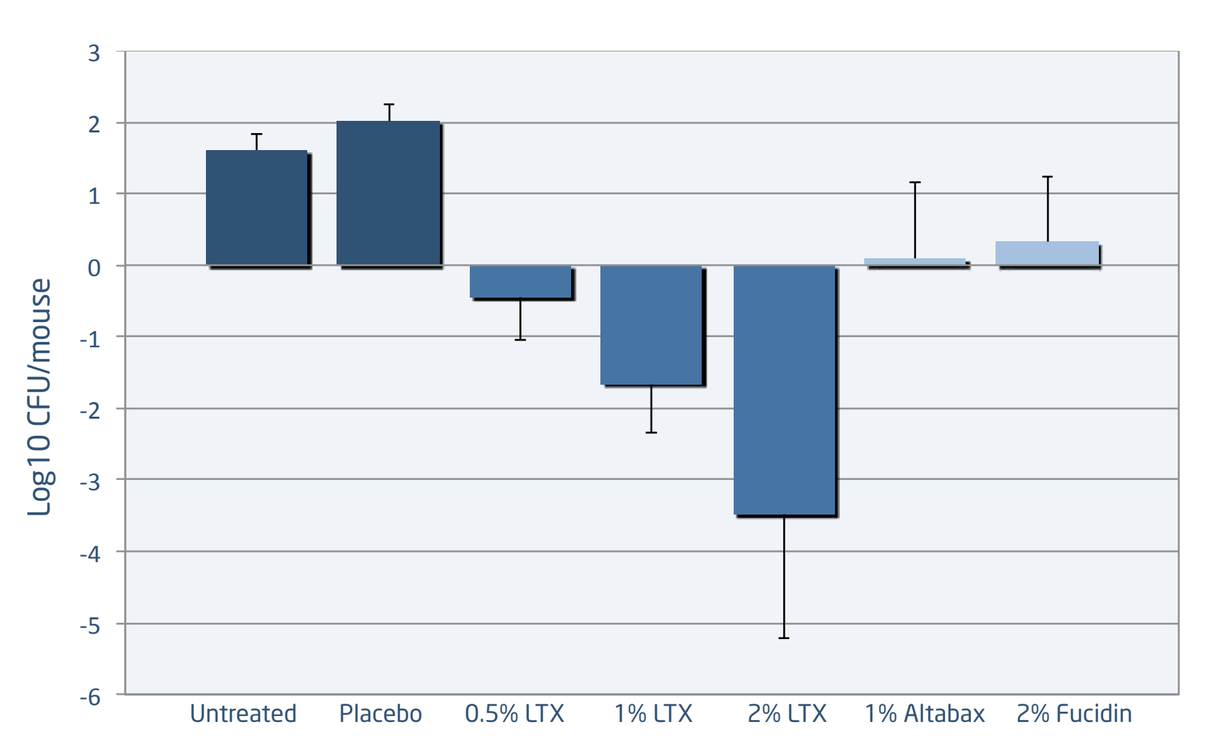
Log10 CFU/mouse
3
2
1
0
-1
-2
-3
-4
-5
-6
Untreated
Placebo
0.5% LTX
1% LTX
2% LTX
1% Altabax
2% Fucidin

Infection:
Community acquired-MRSA
ATCC BA-1717 (USA300)

Treatment:
TID – one day

Measure:
Bacterial growth was assessed +9 hours after first treatment – change in CFU from inoculation load

Result:
Treatment with 0.5%, 1% and 2% LTX-109 was highly effective in reducing bacterial tissue load of CA-MRSA (USA300). A dose-dependent reduction in CFU-counts was observed after one day of treatment. LTX-109 doses of 1% and 2% were significantly better than the comparators Altabax® and Fucidin®

LTX-109 demonstrates a superior effect upon *S. pyogenes* compared to marketed products

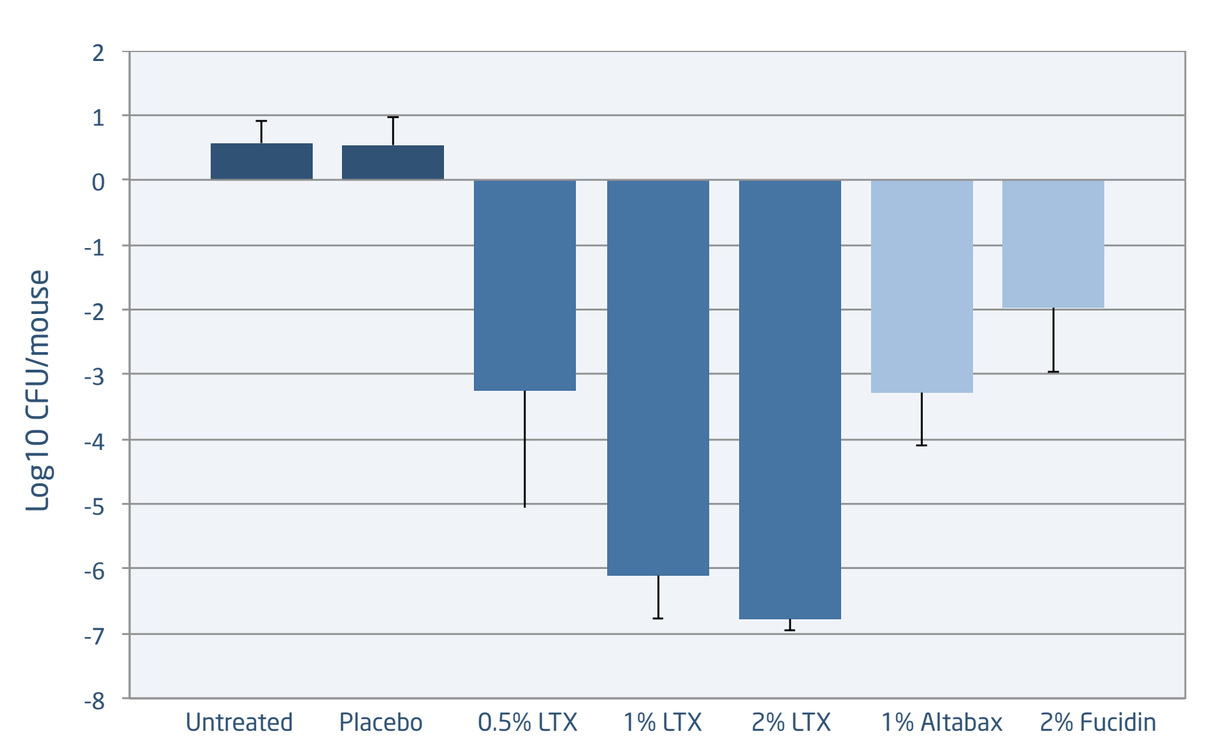
Log10 CFU/mouse
2
1
0
-1
-2
-3
-4
-5
-6
-7
-8
Untreated
Placebo
0.5% LTX
1% LTX
2% LTX
1% Altabax
2% Fucidin

Infection:
Streptococcus pyogenes (SR186)

Treatment:
TID – one day

Measure:
Bacterial growth was assessed +9 hours after first treatment – change in CFU from inoculation load

Result:
Treatment with 0.5%, 1% and 2% LTX-109 was highly effective in reducing bacterial tissue load of *S. pyogenes*. A dose-dependent reduction in CFU-counts was observed after one day of treatment. LTX-109 doses of 1% and 2% were significantly better than the comparators, Altabax® and Fucidin®

LTX-109 demonstrates a superior effect upon *S. pyogenes* compared to Bactroban®

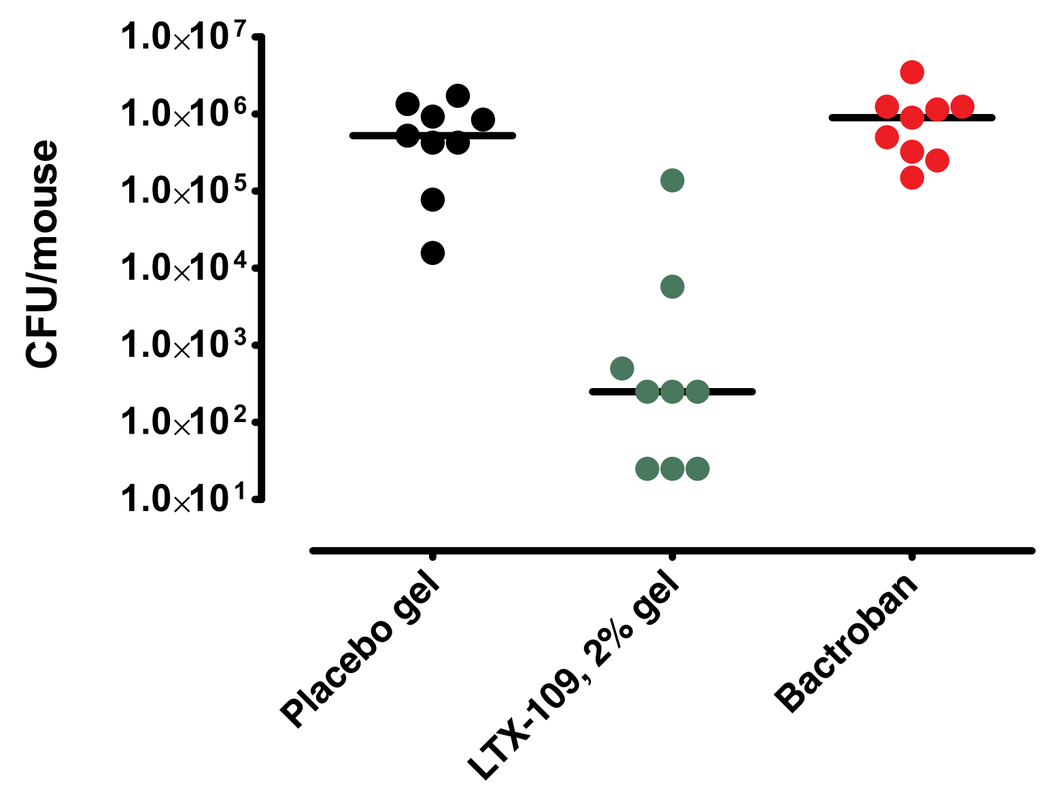
CFU/mouse
1.0×10⁷
1.0×10⁶
1.0×10⁵
1.0×10⁴
1.0×10³
1.0×10²
1.0×10¹
Placebo gel
LTX-109, 2% gel
Bactroban

Infection:
Streptococcus pyogenes (CS301)

Treatment:
TID – one day

Measure:
Bacterial growth was assessed +9 hours after first treatment

Result:
Treatment with 2% LTX-109 was highly effective in reducing bacterial tissue load of *S. pyogenes*. LTX-109 treatment was significantly better than the comparator Bactroban®.

(Figure from Svendsen et al, Bactericidal effect of LTX-109 against *Staphylococcus aureus* and *Streptococcus pyogenes* in a murine skin infection model. ICAAC abstract 2008 poster F1-3947)

Conclusions

- LTX-109 demonstrates a significant effect already after a single day of treatment probably due to the bactericidal mode of action of the drug.
- LTX-109 demonstrates a significant effect on communty- acquired MRSA (USA300) and streptococcal tissue loads in a superficial skin infection model after only one day of treatment.
- The effect was superior to Bactroban®, Altabax® and Fucidin®
- LTX-109 appears to be a valuable drug for treatment of Gram (+) skin infections including those caused by CA-MRSA and *S. pyogenes*.
- The drug has been tested in Phase I and two Phase I/IIa trials with good tolerance, minimal systemic bioavailability
- LTX-109 has demonstrated Proof-of-Concept in decolonisation of nasal MRSA / MSSA.
- Further Phase II studies are planned to demonstrate efficacy in larger patient populations.

LTX-109

- Novel mechanism of action
- Broad spectrum of activity
- Low propensity for resistance development and active against drug-resistant strains
- Effective against fungal and bacterial biofilms
- Superior efficacy compared to market leaders (Bactroban®, Fucidin®, Altabax®/Altargo®)

Lytix Biopharma AS | P.O. Box 6447 | NO-9294 Tromsø, Norway | E-mail: post@lytixbiopharma.com | Phone: +47 77 67 55 00 | Fax: +47 77 67 55 01