The oncolytic peptide LTX-315 enhances T cell clonality and induces synergy with chemotherapy

KETIL ANDRÉ CAMILIO[1,2], MENGYU WANG[1], JANNE NESTVOLD[1,2], GUNNAR KVALHEIM[1], GUNHILD MARI MÆLANDSMO[1], BALDUR SVEINBJØRNSSON[2,3] AND ØYSTEIN REKDAL[2,3]

1. Institute of Cancer Research, OUS, Oslo Norway
2. Lytix Biopharma AS, P.O. Box 6447, NO-9294 Tromsø, Norway
3. Department of Medical Biology, Faculty of Health Sciences, University of Tromsø, Tromsø, Norway

Lytix Biopharma

Background

LTX-315 is a novel **oncolytic peptide** derived from the naturally occurring host defense peptide, bovine lactoferricin [1]. LTX-315 interacts electrostatically with anionic components of negatively charged cancer cell membranes as well as intracellular targets such as mitochondria. This causes cellular lysis and a subsequent release of endogenous cellular content including danger signals and tumor antigens, leading to long lasting tumor-specific immune responses [2-9].

Low-dose chemotherapy often exerts a dual mode of action. In addition to direct tumor cell killing several chemotherapeutic drugs, e.g. cyclophosphamide and doxorubicin, have been shown to display immune modulating properties. Low-dose cyclophosphamide has been shown to selectively downregulate immunosuppressive regulatory T cells and doxorubicin immunosuppressive myeloid-derived suppressor cells. Treatment with LTX-315 modulates the tumor microenvironment, changing "cold" or non-inflamed tumors into "hot" or inflamed tumors through the induction of a unique type of immunogenic cell death. Thus, we hypothesized that an enhanced antitumor effect and augmented tumor-specific immune responses could be achieved when LTX-315 was combined with low-dose chemotherapy.

Aim

Investigate the antitumor efficacy and potential synergy of LTX-315 in combination with low-dose chemotherapy in experimental mouse models.

LTX-315

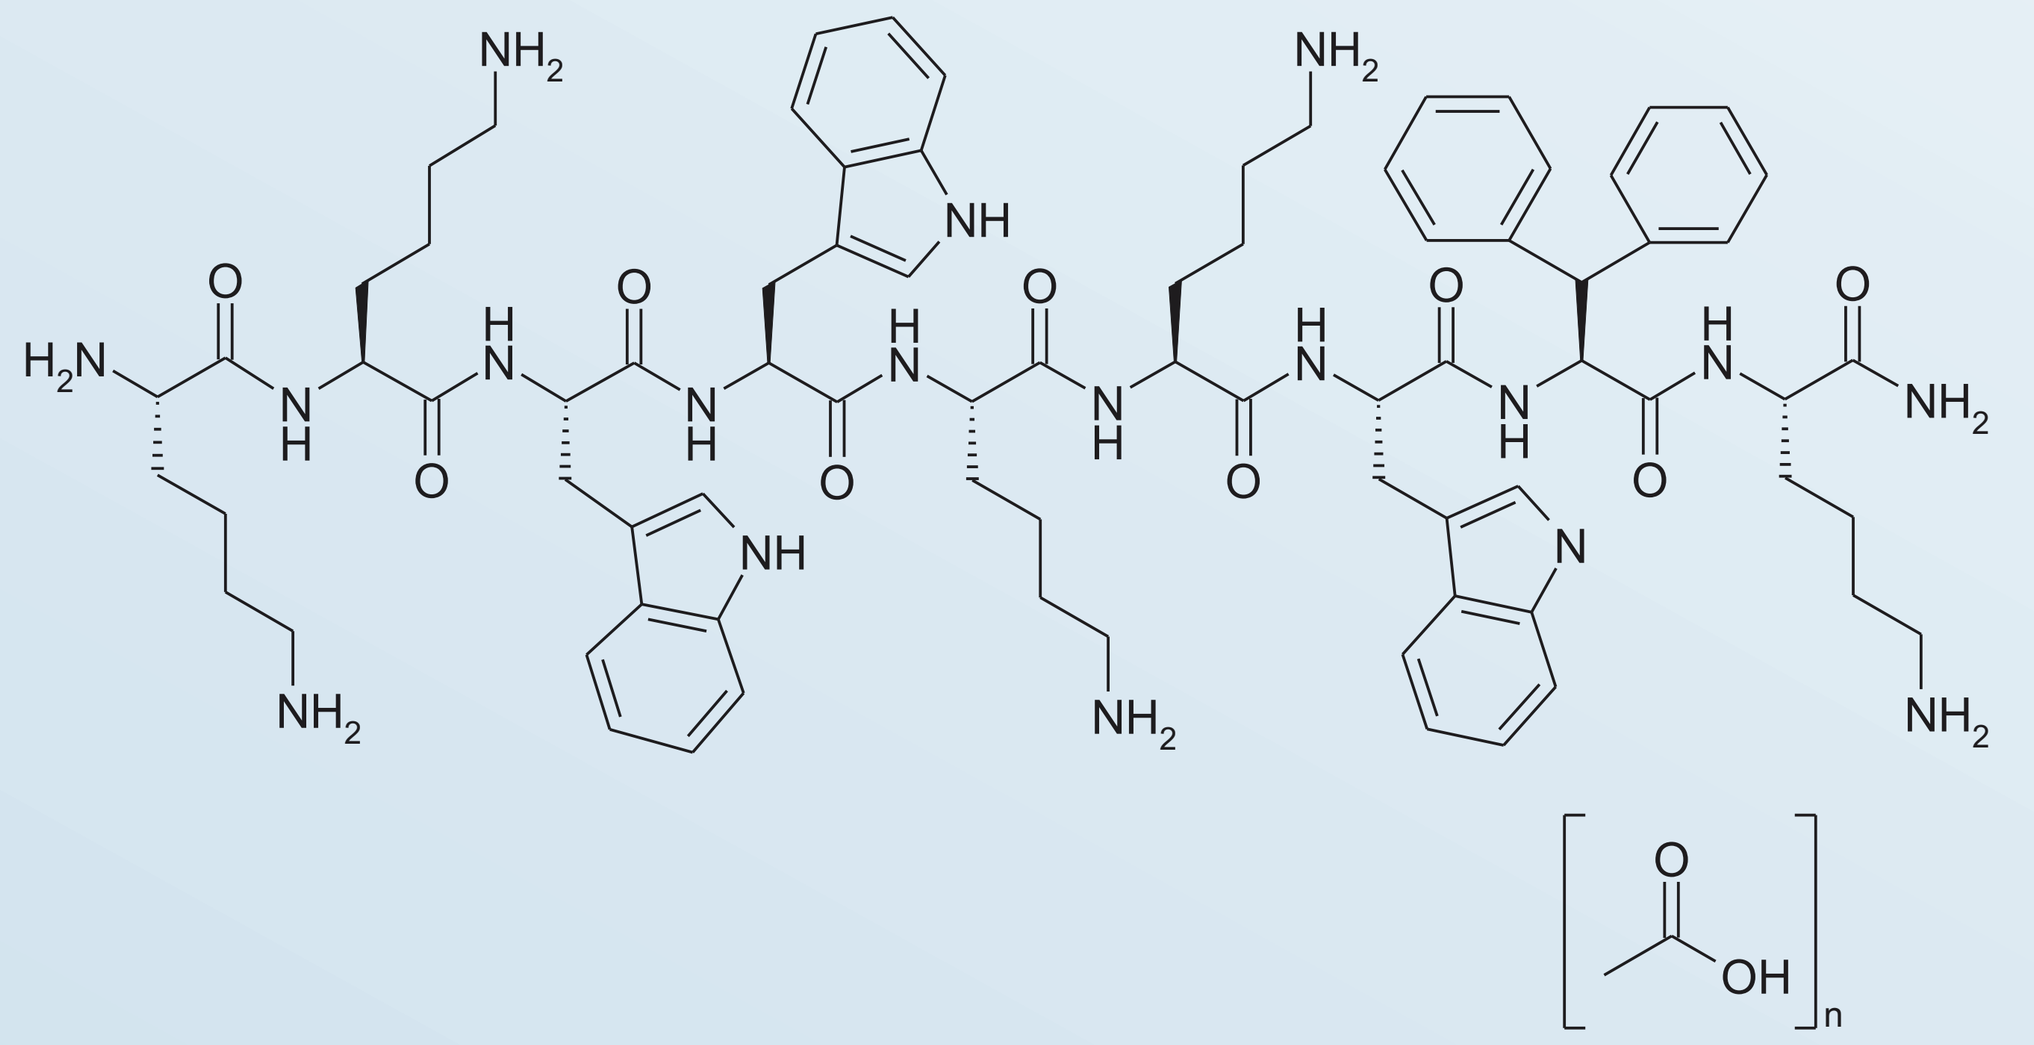
NH2
NH
O
OH
n

Mode of action

LTX-315 induces a unique type of immunogenic cell death

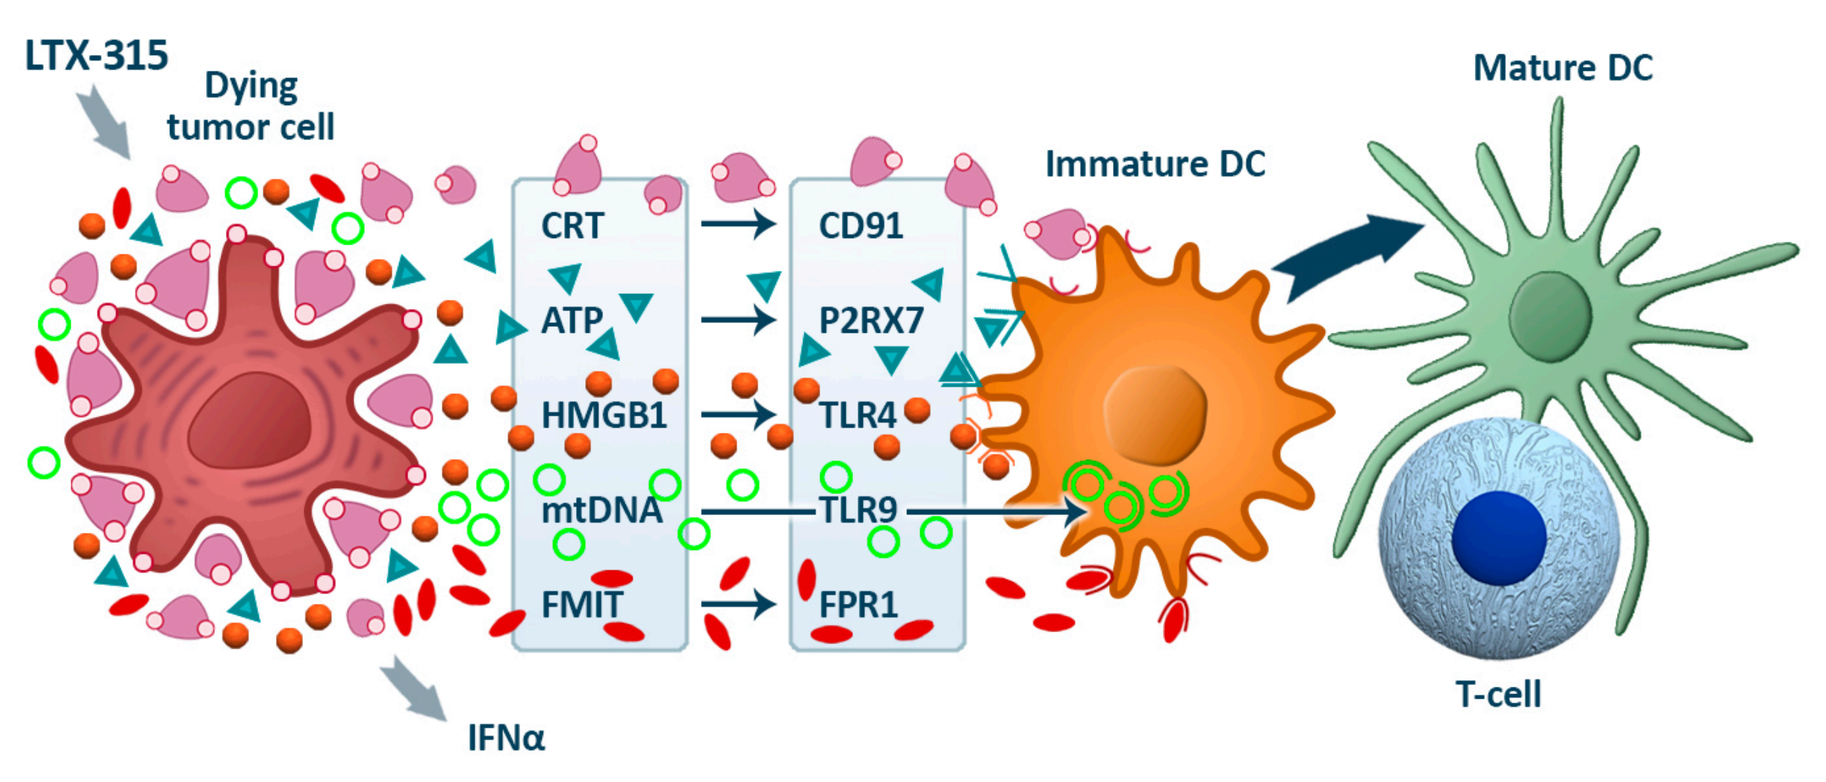
LTX-315
Dying tumor cell
CRT
ATP
HMGB1
mtDNA
FMIT
CD91
P2RX7
TLR4
TLR9
FPR1
Immature DC
Mature DC
T-cell
IFNα

LTX-315 increases the number and diversity of T cell clones

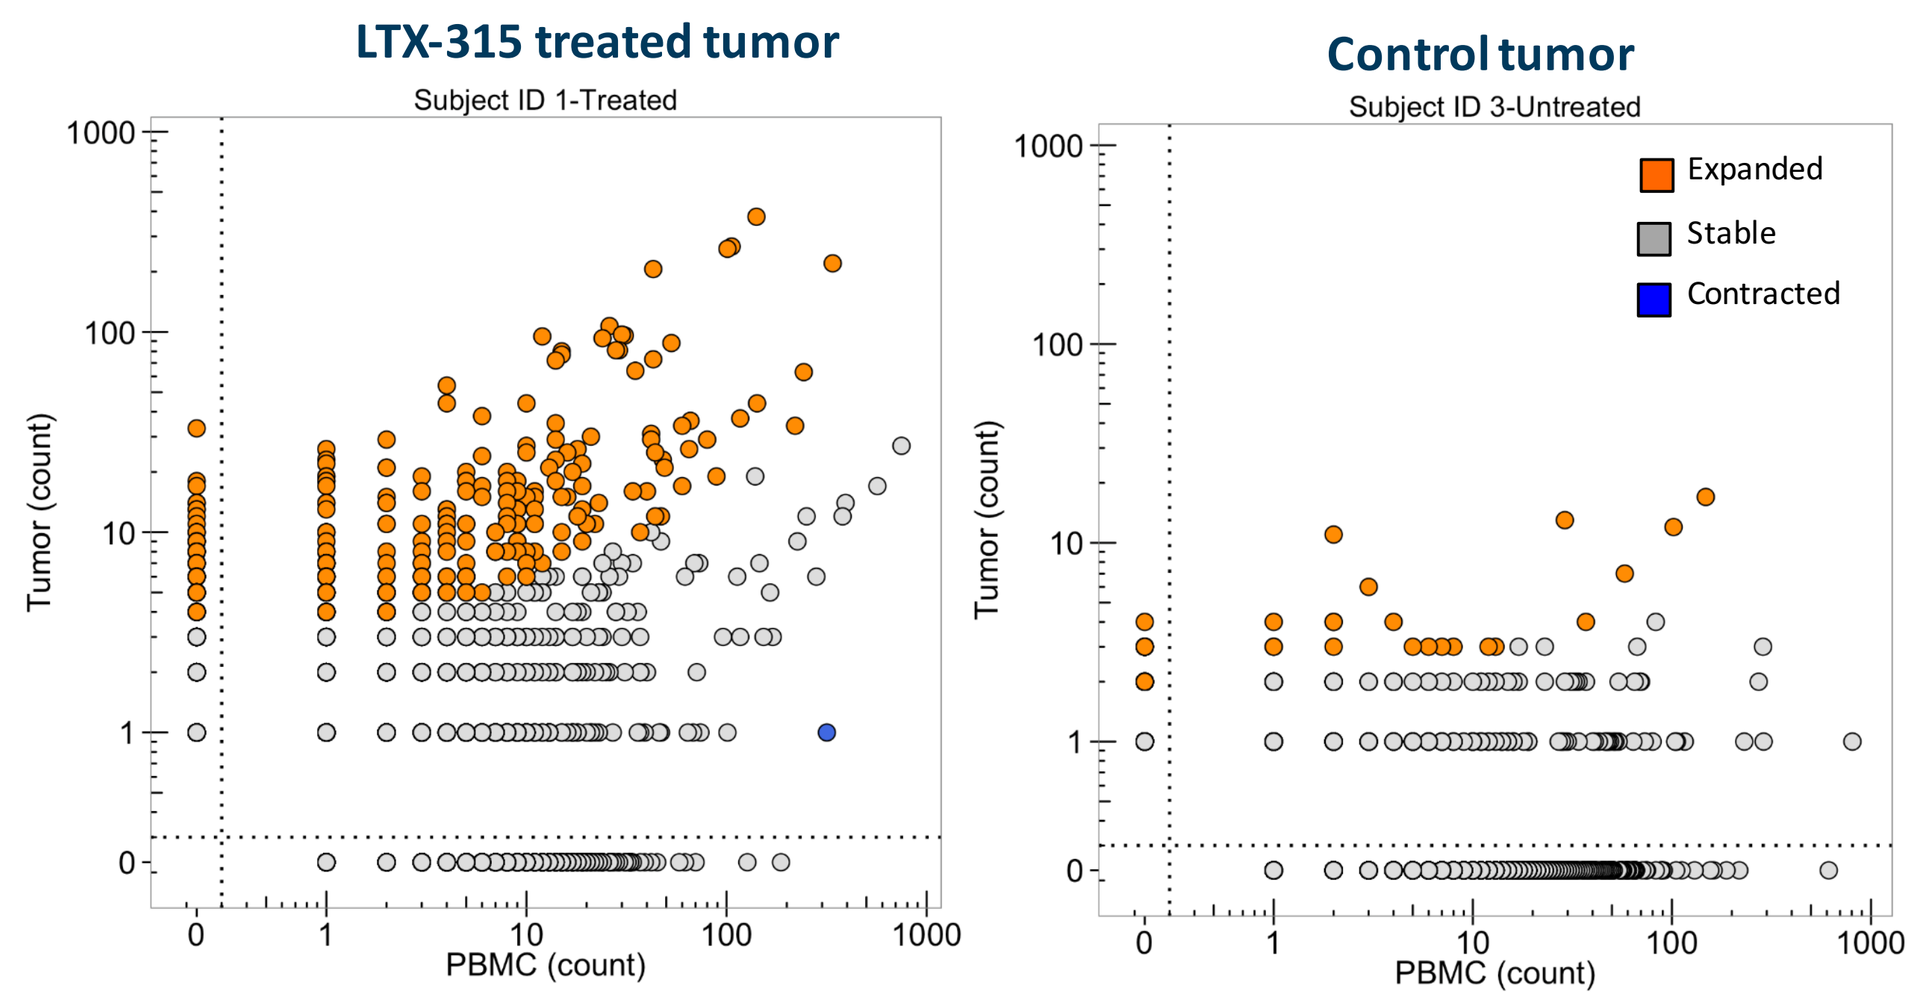
LTX-315 treated tumor
Subject ID 1-Treated
Control tumor
Subject ID 3-Untreated
Expanded
Stable
Contracted
Tumor (count)
PBMC (count)

T cell clones in LTX-315-treated and control tumors were amplified and sequenced using the ImmunoSeq platform by Adaptive Biotech. Multiplex PCR was used to amplify the rearranged TCR3b sequences from sample DNA (VDJ region).

Results

LTX-315 in combination with cyclophosphamide

Study design

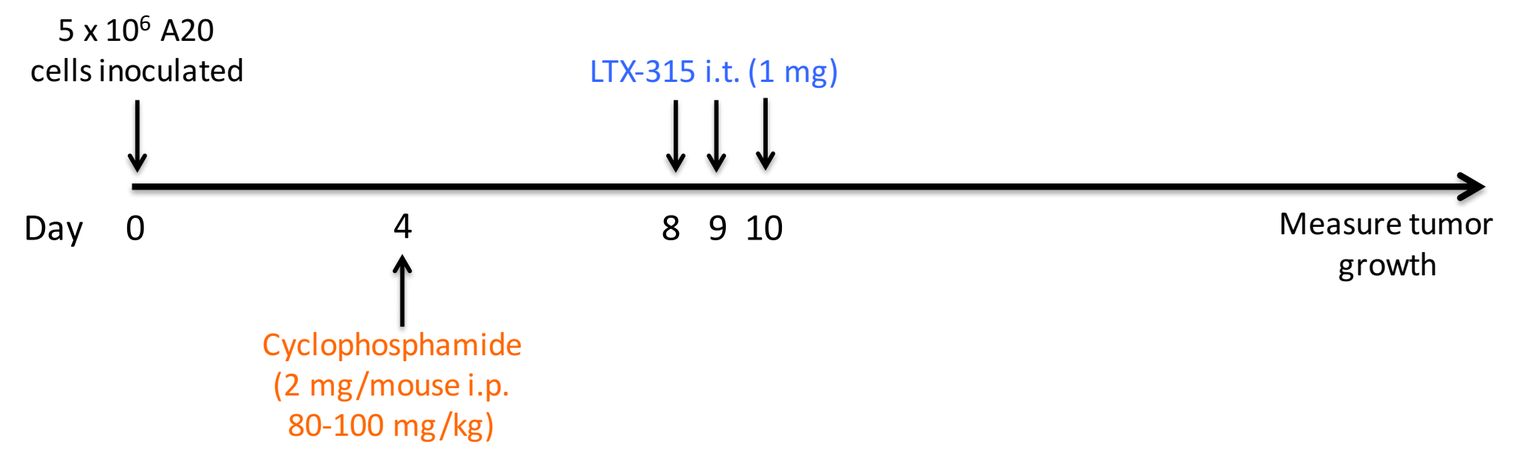
5 x 10⁶ A20 cells inoculated
LTX-315 i.t. (1 mg)
Day 0 4 8 9 10
Measure tumor growth
Cyclophosphamide (2 mg/mouse i.p. 80-100 mg/kg)

LTX-315 in combination with cyclophosphamide induced complete regression of A20 B-lymphomas (s.c.)

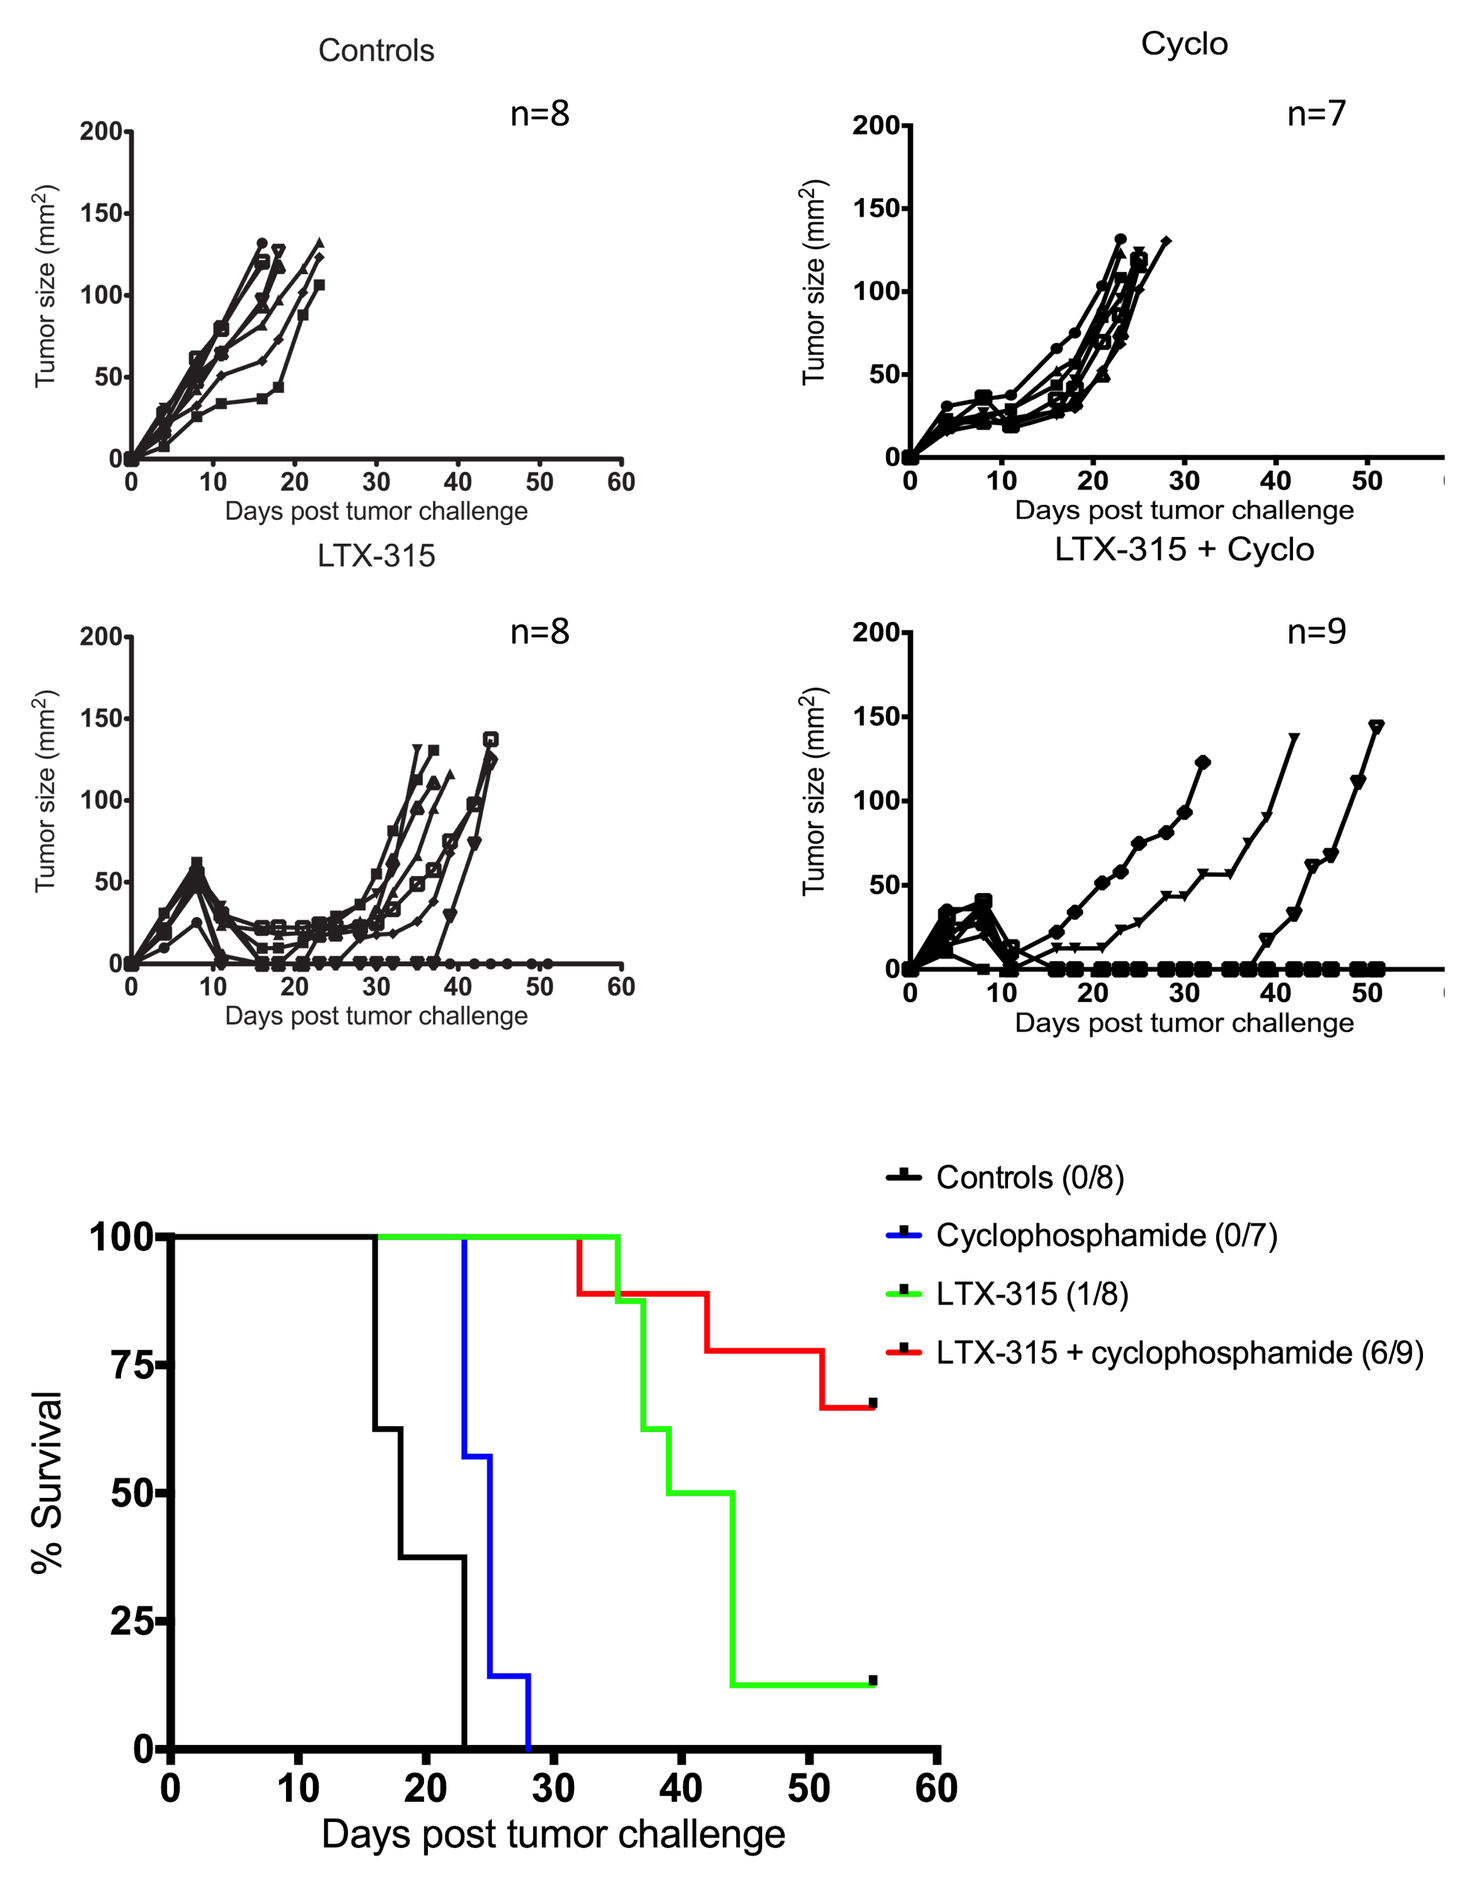
Controls n=8
Cyclo n=7
LTX-315 n=8
LTX-315 + Cyclo n=9
Tumor size (mm²)
Days post tumor challenge
Controls (0/8)
Cyclophosphamide (0/7)
LTX-315 (1/8)
LTX-315 + cyclophosphamide (6/9)
% Survival

Tumor growth of subcutaneously established A20 tumors in animals injected intratumorally with LTX-315 alone (1 mg/50 µl), intraperitoneally with cyclophosphamide alone (2mg/mouse), or with LTX-315 in combination with cyclophosphamide.

LTX-315 in combination with doxorubicin

Study design

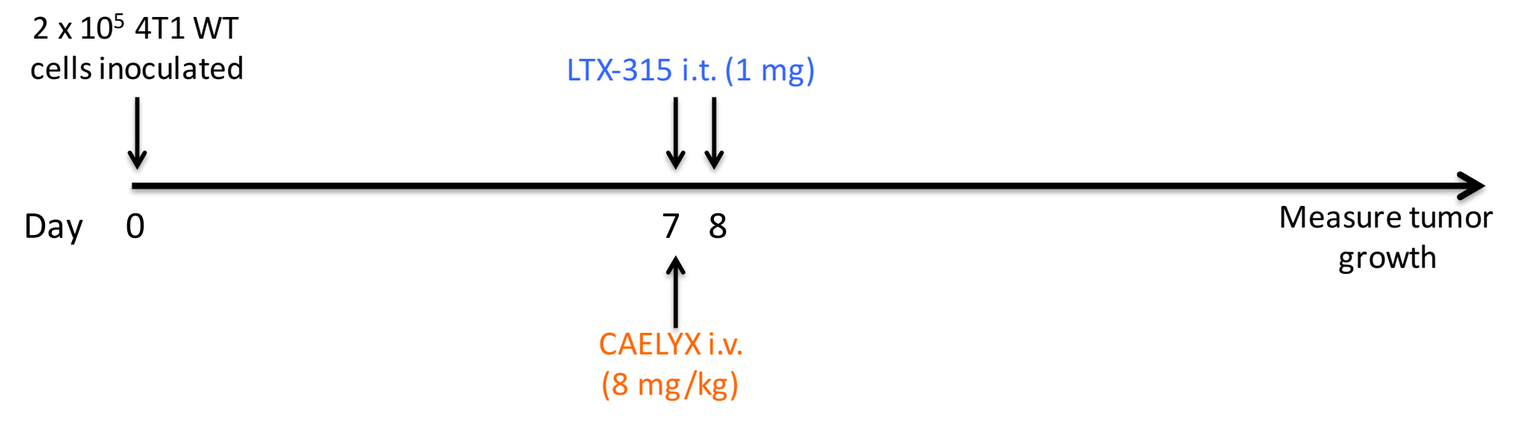
2 x 10⁵ 4T1 WT cells inoculated
LTX-315 i.t. (1 mg)
Day 0 7 8
Measure tumor growth
CAELYX i.v. (8 mg/kg)

LTX-315 in combination with doxorubicin induced complete regression of orthotopic 4T1 mammary carcinomas

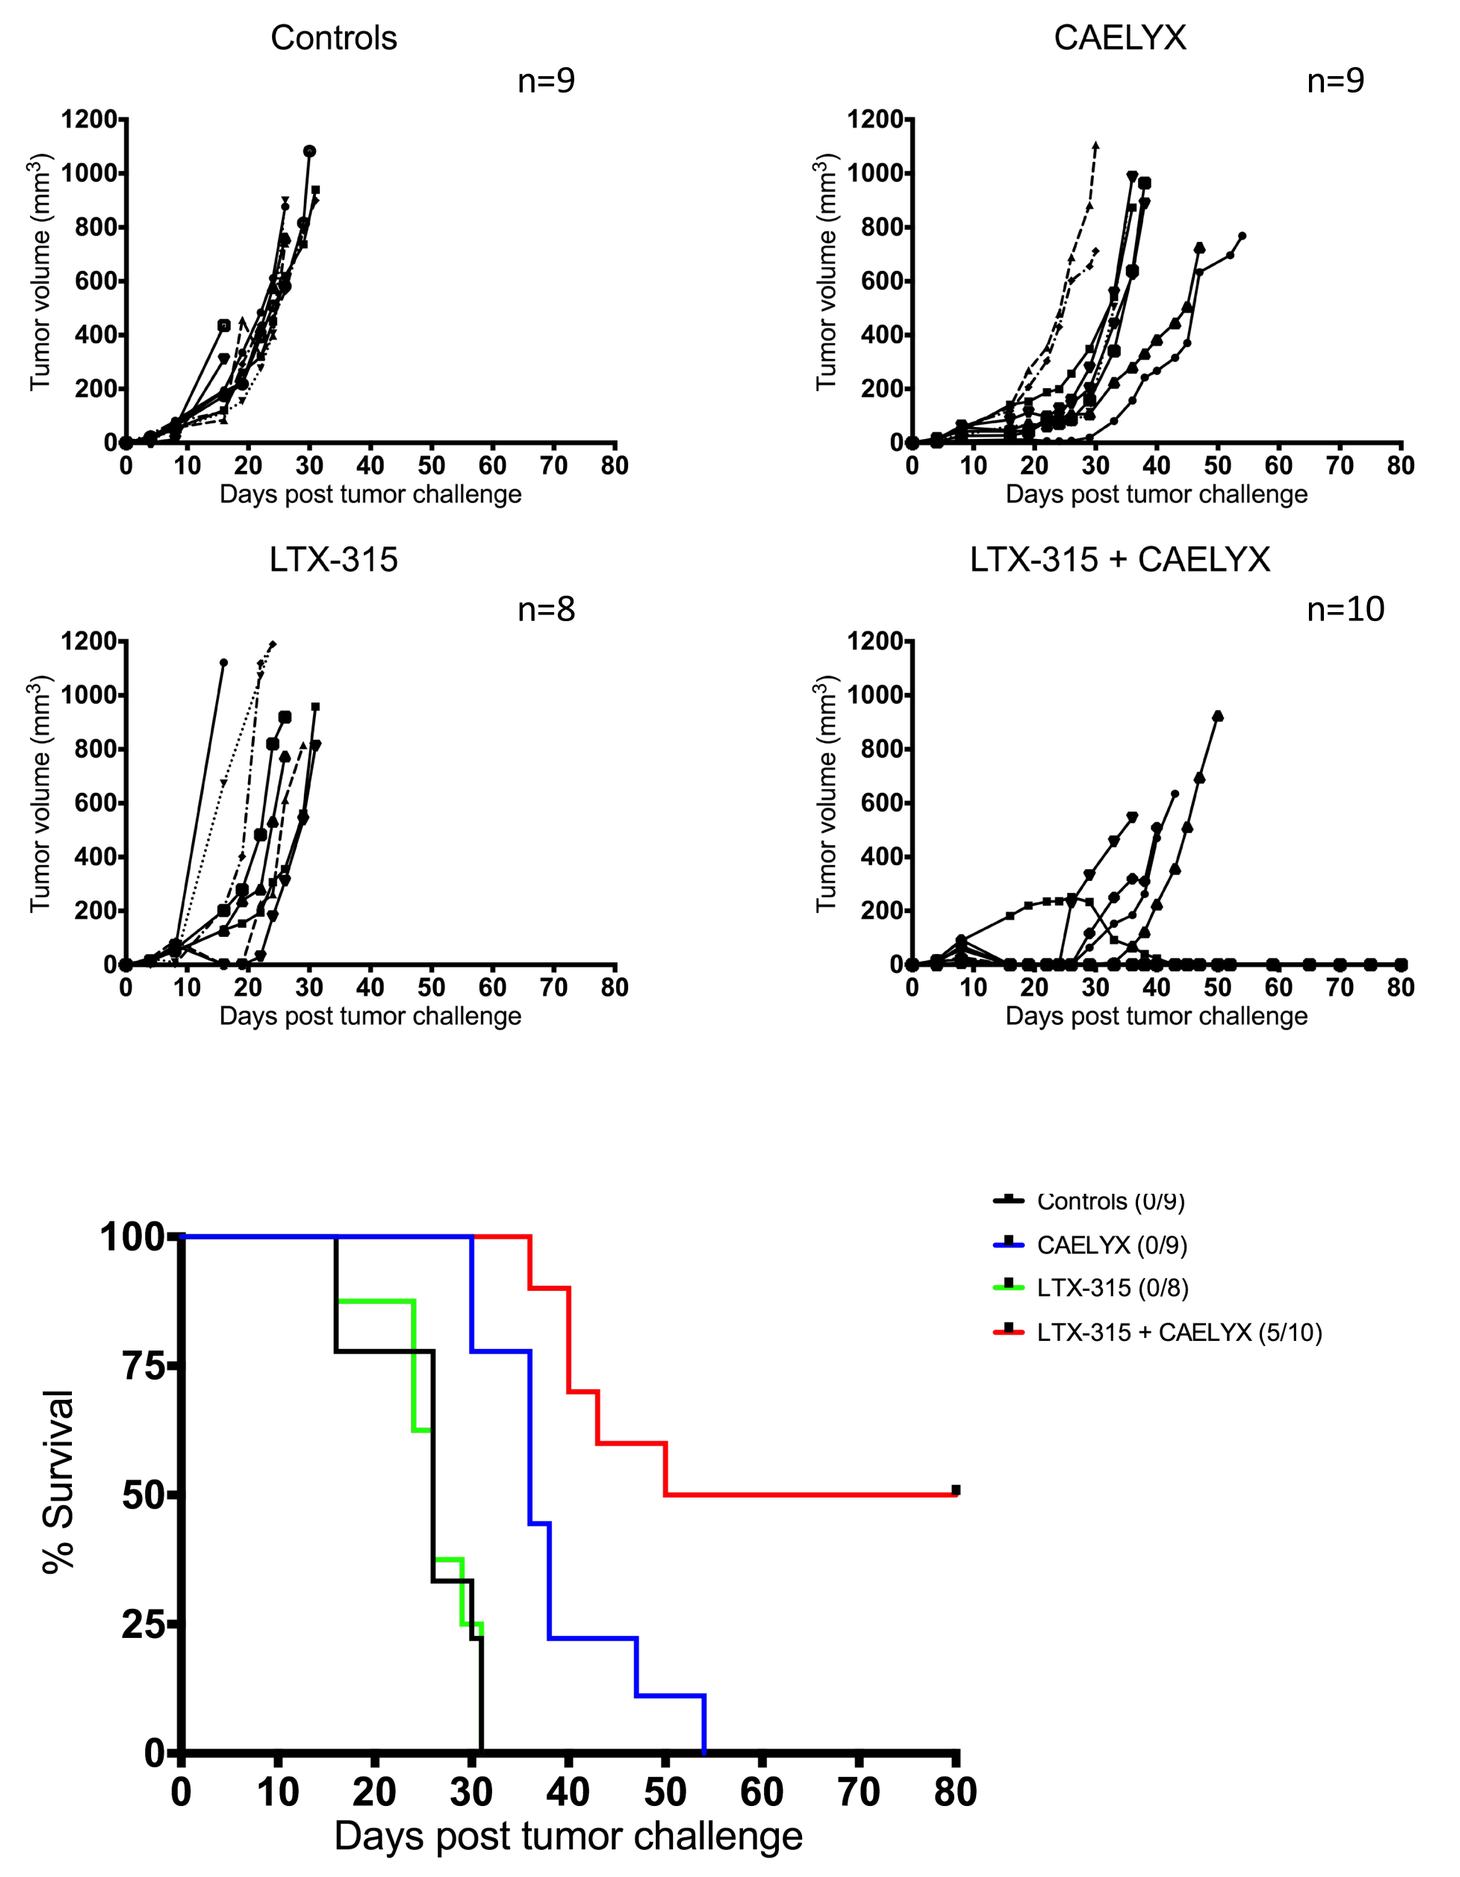
Controls n=9
CAELYX n=9
LTX-315 n=8
LTX-315 + CAELYX n=10
Tumor volume (mm³)
Days post tumor challenge
Controls (0/9)
CAELYX (0/9)
LTX-315 (0/8)
LTX-315 + CAELYX (5/10)
% Survival

Tumor growth of orthotopically established 4T1 tumors in animals injected intratumorally with LTX-315 alone (1 mg/50 µl), intravenously with CAELYX alone (8mg/kg), or with LTX-315 in combination with CAELYX. CAELYX is liposomal doxorubicin.

MRI

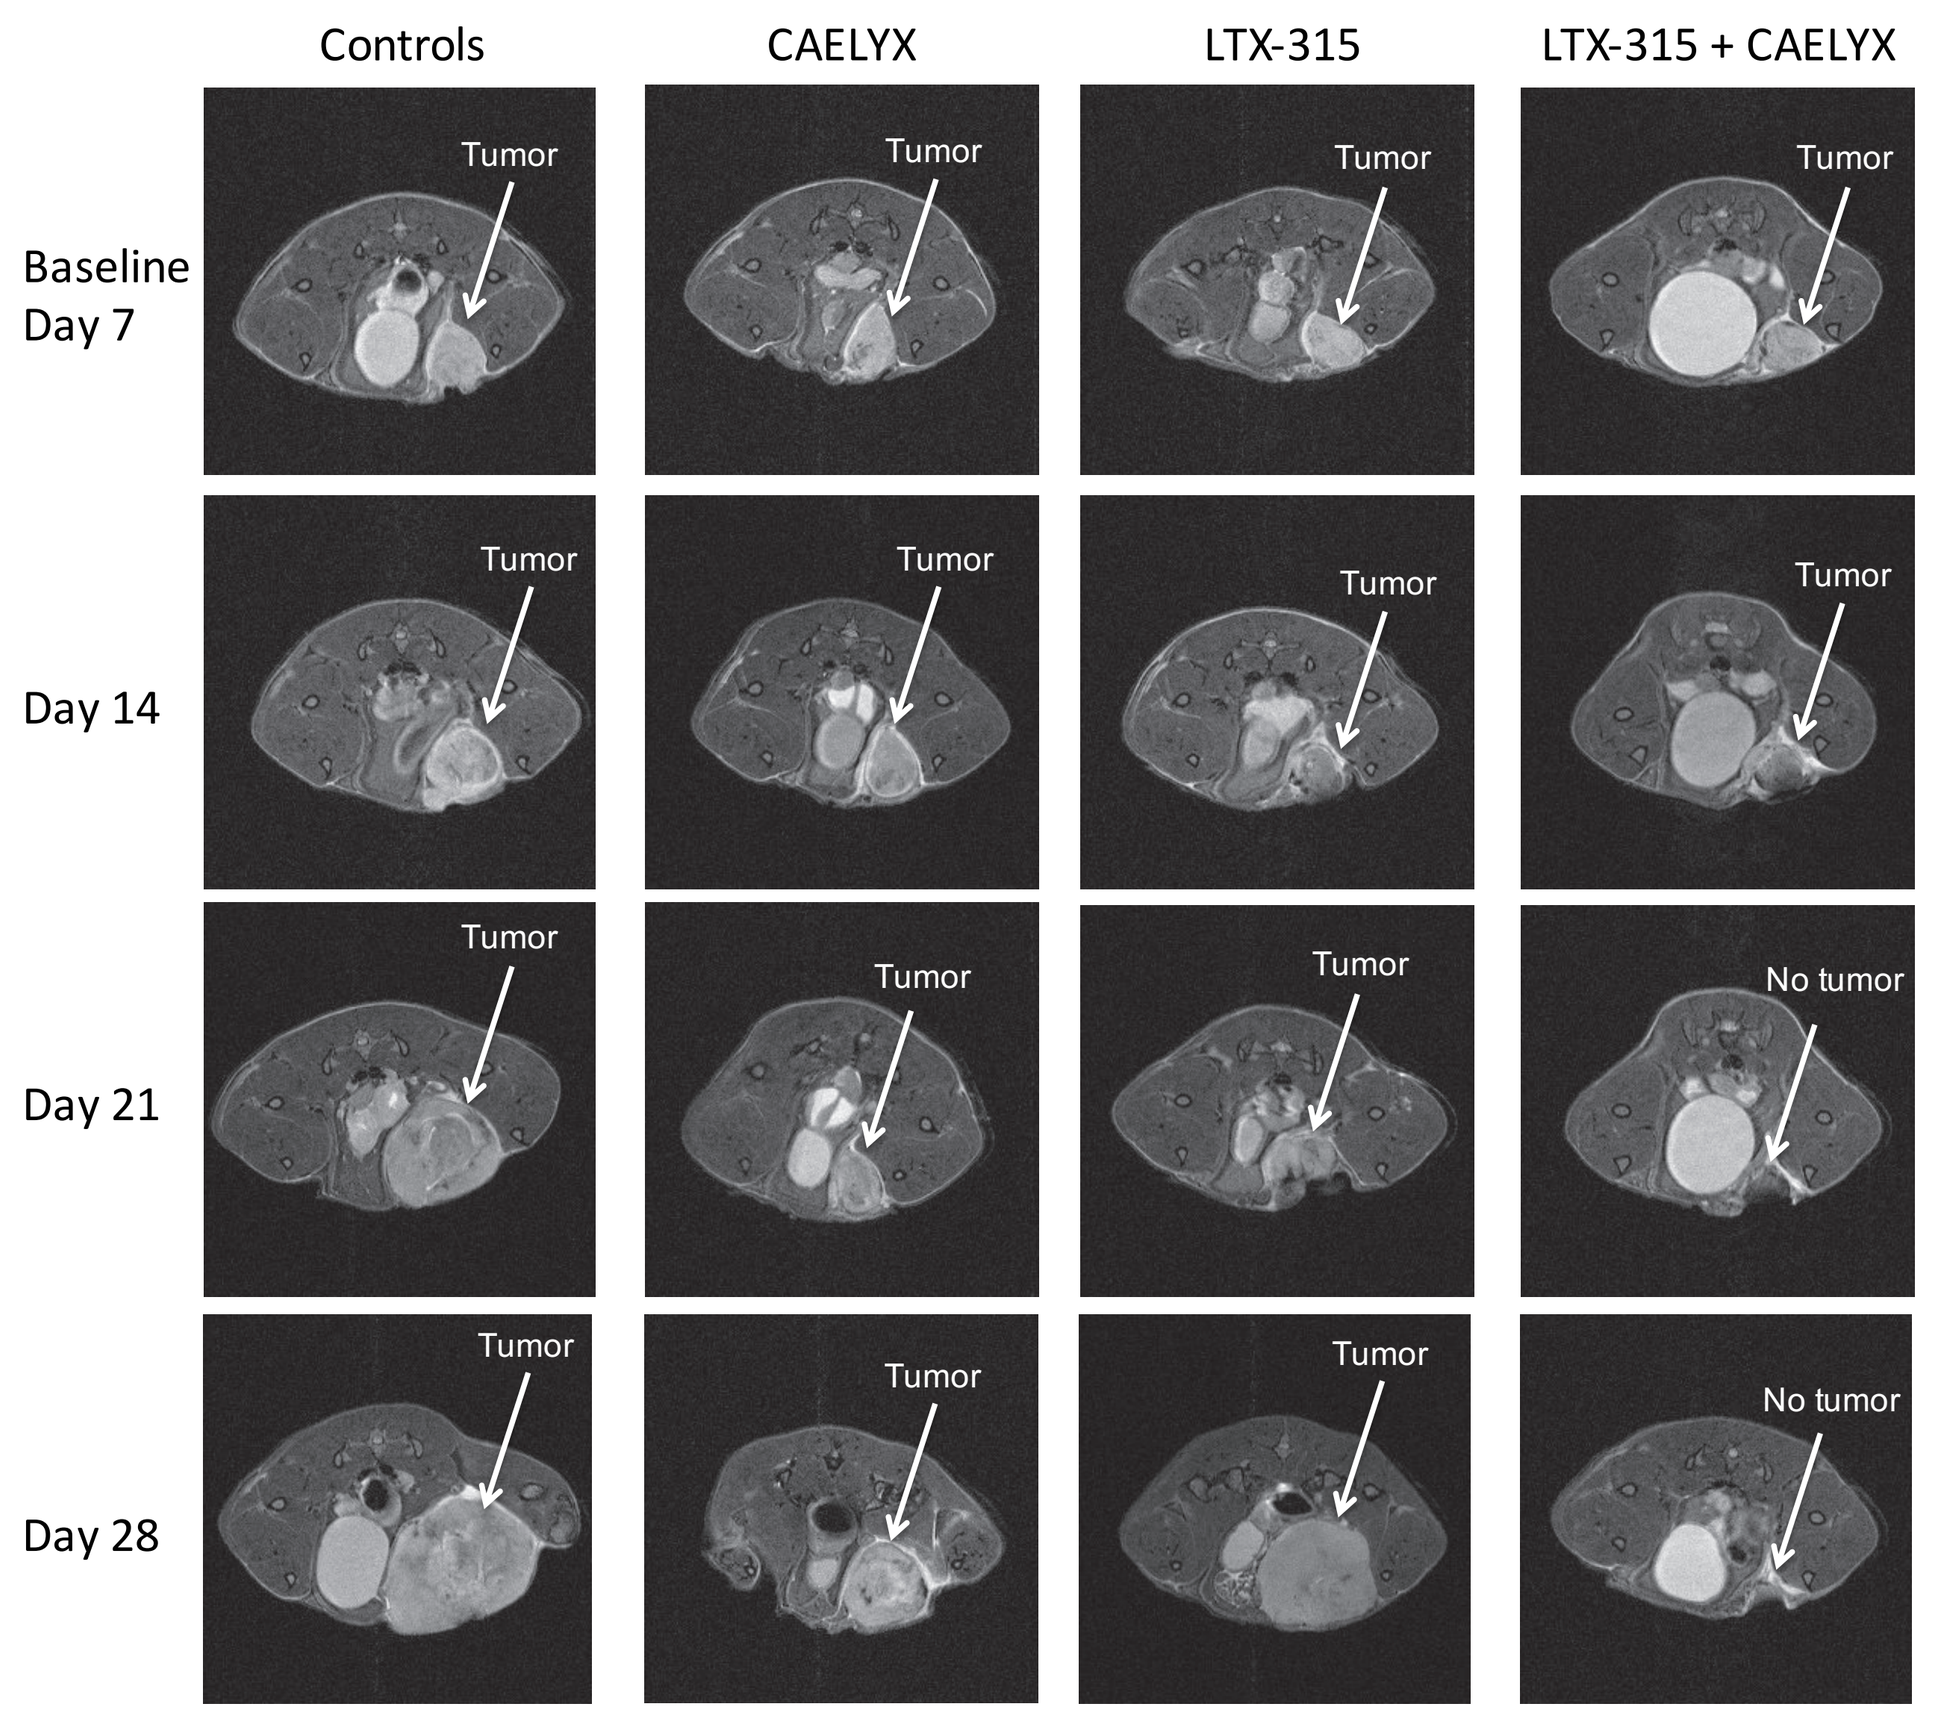
Controls
CAELYX
LTX-315
LTX-315 + CAELYX
Baseline Day 7
Day 14
Day 21
Day 28
Tumor
No tumor

Representative magnetic resonance images are shown using a BioSpec 7T animal MR scanner from Bruker Corporation.

Histology

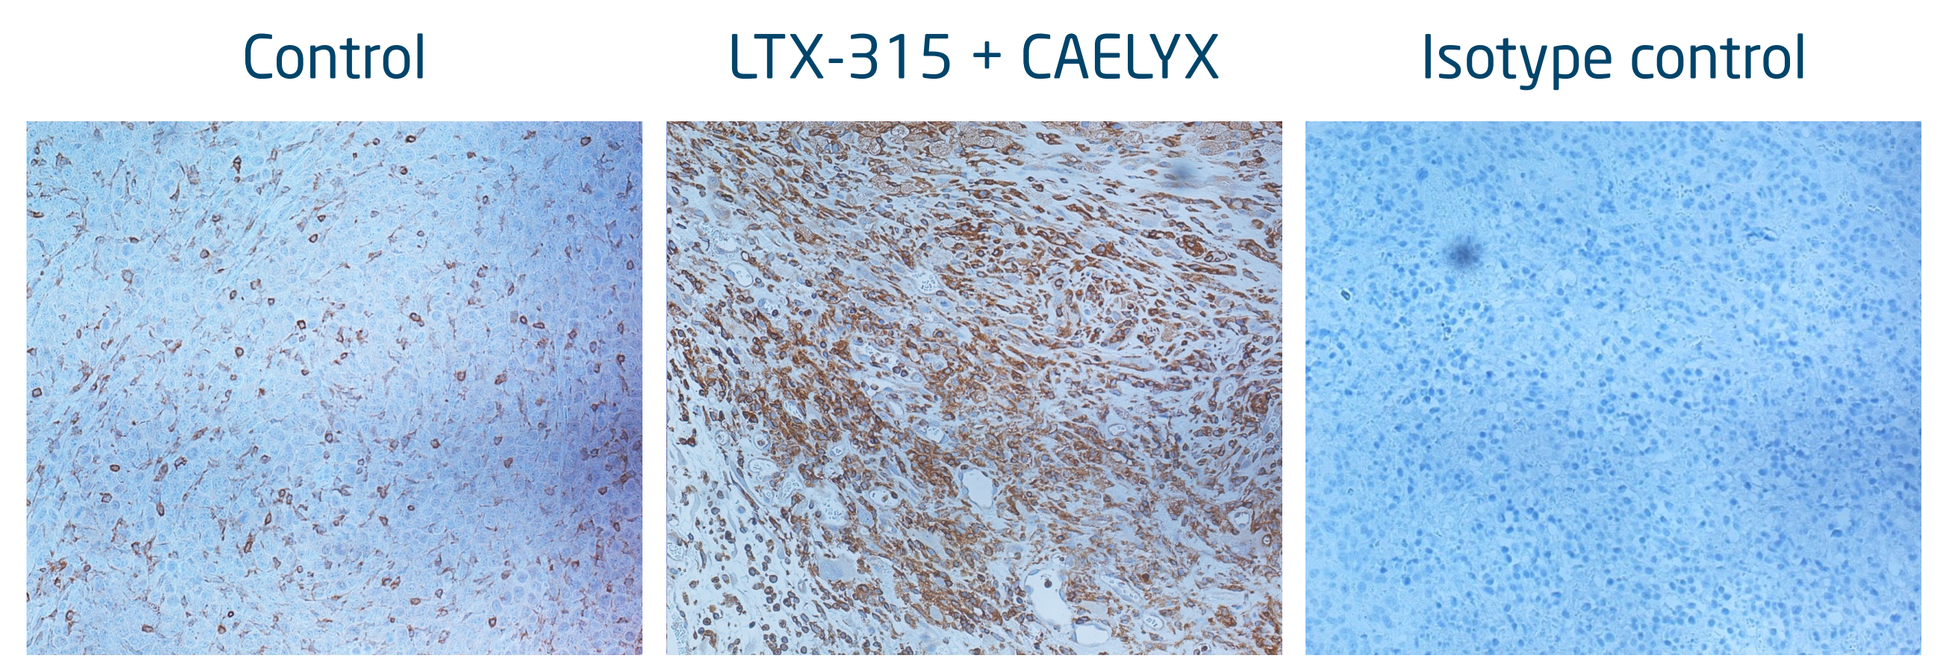
Control
LTX-315 + CAELYX
Isotype control

Combination treatment with LTX-315 and CAELYX induces infiltration of CD3+ T cells into the tumor parenchyma.

Conclusion

- LTX-315 showed an enhanced anticancer efficacy against A20 lymphomas and 4T1 breast carcinomas when combined with cyclophosphamide and doxorubicin, respectively.
- The LTX-315 unique "release and reshape" properties make it a promising candidate for combination with several types of immunotherapies.
- LTX-315 is currently in clinical phase 1/2a studies.

References

1. Haug et al. J Med Chem. 2016
2. Camilio et al. Cancer Immunol Immunother. 2014
3. Camilio et al. Oncoimmunology. 2014
4. Zhou et al. Oncotarget. 2015
5. Eike et al. Oncotarget. 2015
6. Forveille et al. Cell Cycle. 2015
7. Zhou et al. Cell Death Dis. 2016
8. Sistigu et al. Cell Cycle. 2016
9. Yamazaki et al. Cell Death Differ. 2016

Oslo University Hospital

CHILI. © WWW.CHILIGROUP.NO | 2017

Lytix Biopharma AS | P.O. Box 6447 | NO-9294 Tromsø, Norway | E-mail: post@lytixbiopharma.com | Phone: +47 77 67 55 00 | Fax: +47 77 67 55 01