LTX-315 AND ADOPTIVE CELL THERAPY USING TUMOR-INFILTRATING LYMPHOCYTES IN PATIENTS WITH METASTATIC SOFT TISSUE SARCOMA (ATLAS-IT-04)

11567

MORTEN NIELSEN[1], TINE MONBERG[1], BENEDETTE ALBIERI[1], VIBEKE SUNDVOLD[2], ØYSTEIN REKDAL[2], DORRIT HOVGAARD[3], MICHAEL MØRK PEDERSEN[3], NIELS JUNKER[4], INGE MARIE SVANE[1].

[1]NATIONAL CENTER FOR CANCER IMMUNE THERAPY; DEPARTMENT OF ONCOLOGY, COPENHAGEN UNIVERSITY HOSPITAL, HERLEV, DENMARK;
[2]LYTIX BIOPHARMA,
[3]IDEPARTMENT OF ORTHOPEDIC SURGERY;
[4]DEPARTMENT OF ONCOLOGY, COPENHAGEN UNIVERSITY HOSPITAL, HERLEV, DENMARK.

Background

Patients with advanced stages of soft tissue sarcomas (STS) respond poorly to current treatment and the prognosis is poor. Median survival for patients with metastatic STS at time of diagnosis was estimated to 10 months with a 5-year survival of 10% (1).

In general, STS responds poorly to immunotherapy due to lack of tumor infiltrating lymphocytes (TILs) (2,3).

LTX-315 is a first in class non-viral oncolytic peptide that in a recent Phase I/II study was shown to increase TILs in malignant solid tumors after intratumoral injection (4-6).

Adoptive Cell Therapy (ACT) with TILs is a potent treatment that can induce complete and durable tumor regression as documented in patients with melanoma. To our knowledge, ACT has not been utilized for patients with advanced STS.

Aim

This proof of concept study will evaluate the potential for LTX-315 to induce TILs prior to isolation and expansion of the TILs followed by infusion of the cultured TILs to patients with advanced STS.

LTX-315`s unique mode of action results in immunogenic cell death resulting in effective release of potent immunostimulants and antigens followed by a broad T-cell response (4-11)

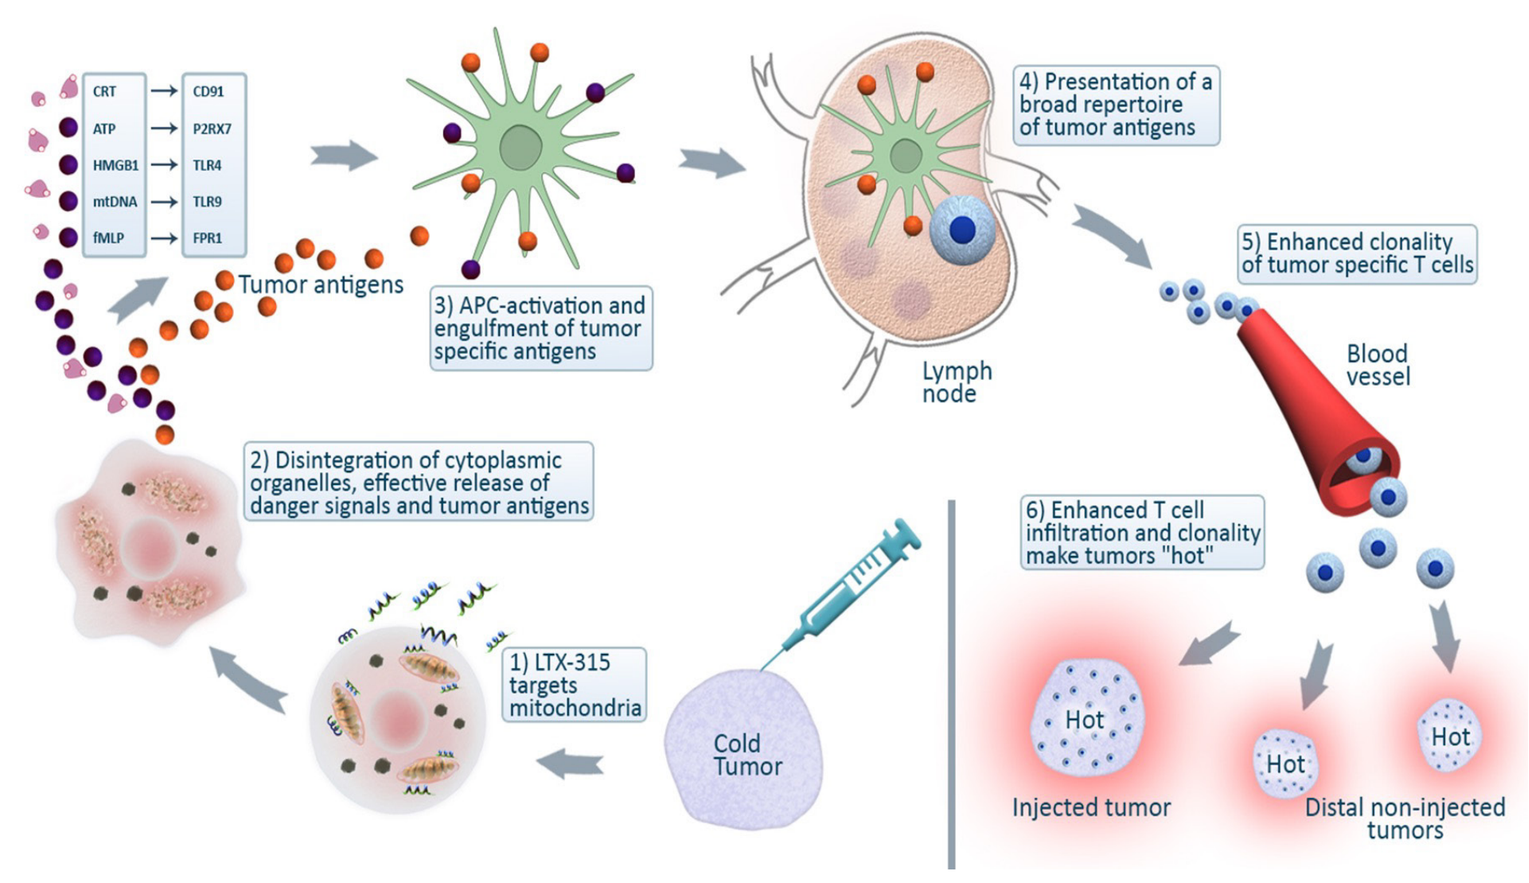
CRT
ATP
HMGB1
mtDNA
fMLP
CD91
P2RX7
TLR4
TLR9
FPR1
Tumor antigens
3) APC-activation and engulfment of tumor specific antigens
4) Presentation of a broad repertoire of tumor antigens
Lymph node
5) Enhanced clonality of tumor specific T cells
Blood vessel
2) Disintegration of cytoplasmic organelles, effective release of danger signals and tumor antigens
6) Enhanced T cell infiltration and clonality make tumors "hot"
1) LTX-315 targets mitochondria
Cold Tumor
Hot
Injected tumor
Distal non-injected tumors

Study Design (NCT03725605)

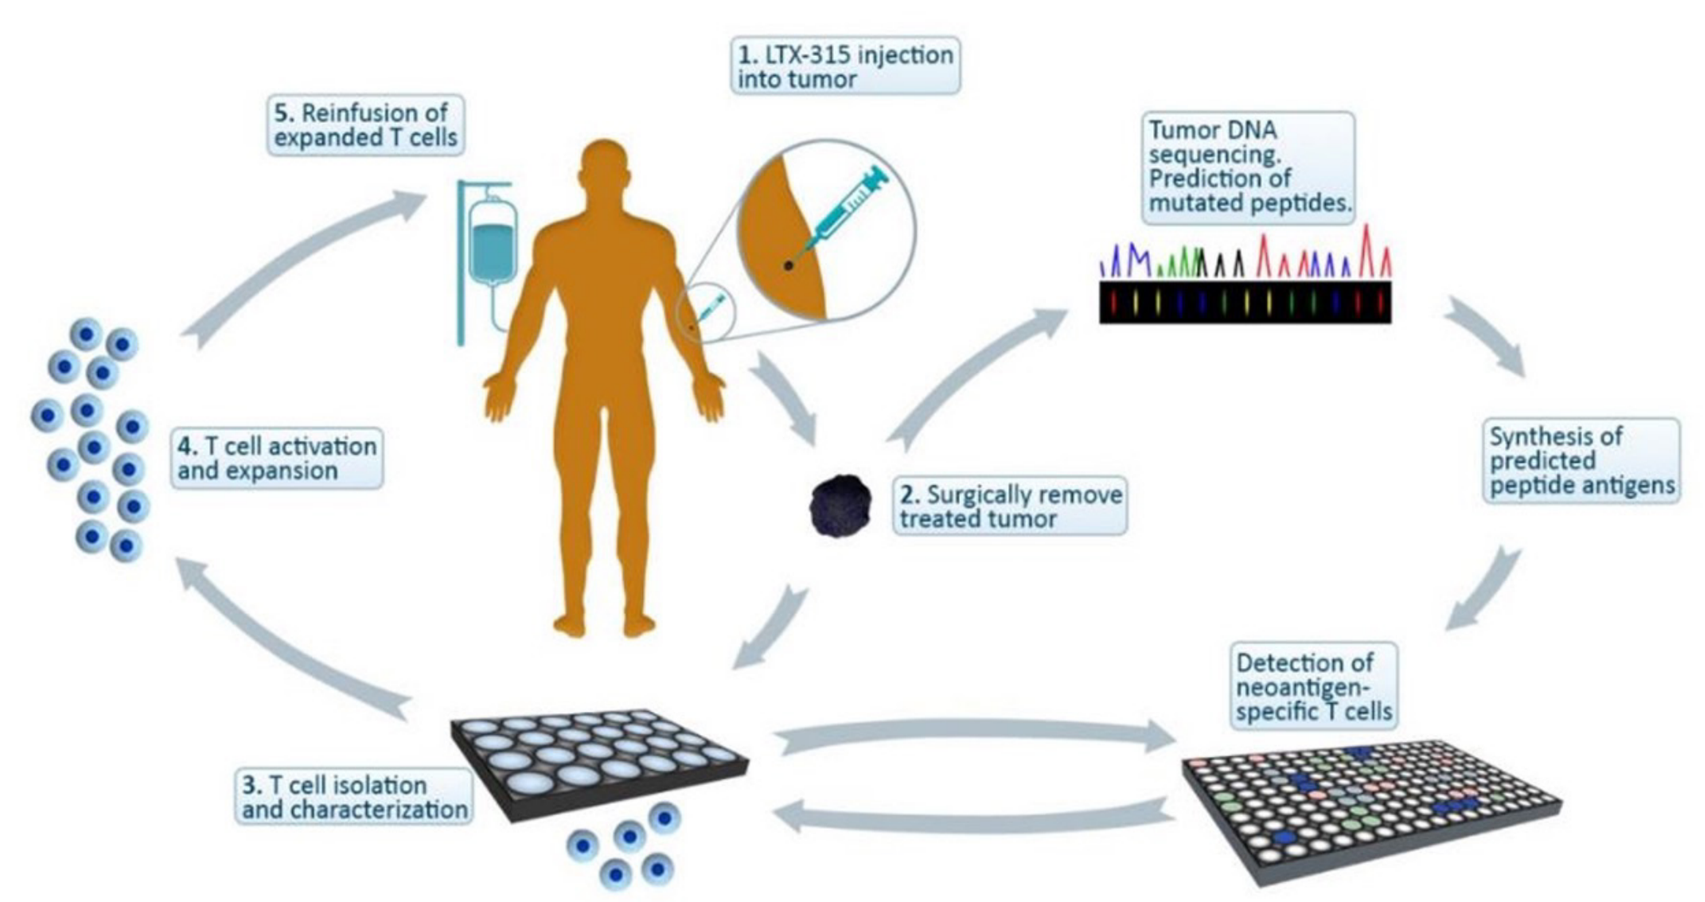
1. LTX-315 injection into tumor
2. Surgically remove treated tumor
3. T cell isolation and characterization
4. T cell activation and expansion
5. Reinfusion of expanded T cells
Tumor DNA sequencing. Prediction of mutated peptides.
Synthesis of predicted peptide antigens
Detection of neoantigen-specific T cells

Treatment Schedule

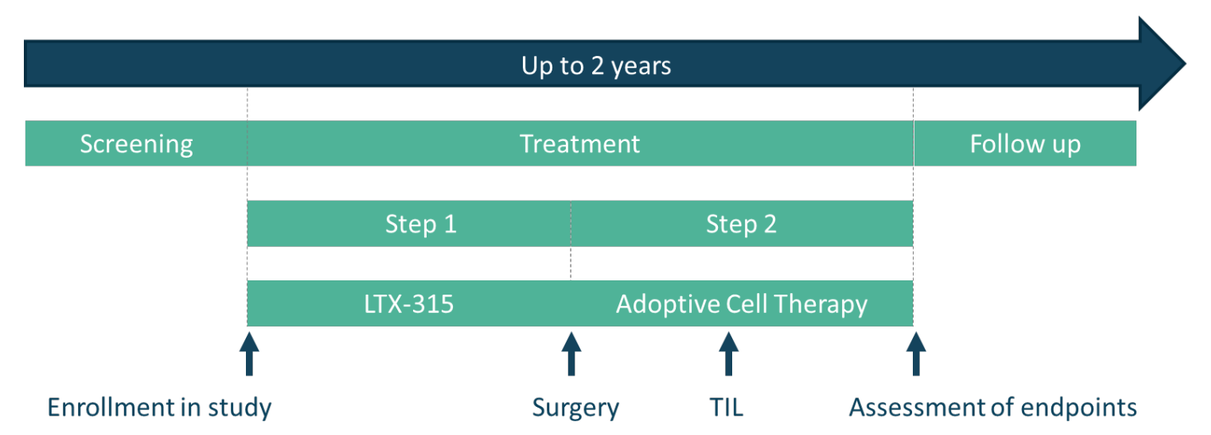
Up to 2 years
Screening
Treatment
Follow up
Step 1
Step 2
LTX-315
Adoptive Cell Therapy
Enrollment in study
Surgery
TIL
Assessment of endpoints

Mean time (days)	22 (15-35)	45 (41-49)	42

Study course	Step 1		Step 2		Follow-up		
	Baseline	Surgery	Before ACT	After ACT	6 w	12 w	Every 3rd m
Biopsy of injected tumour	•	•					
Blood samples	•	•	•	•	•	•	•
Tumor assessment	•		•		•	•	•

Objectives

Primary

- Ability of LTX-315 to induce T-cell infiltration prior to TIL expansion in advanced STS
- Safety of LTX-315 as part of adoptive T-cell therapy in advanced STS

Secondary

- Ability to expand CD8+ T-cells from tumor tissues
- Anti-tumor activity of LTX-315 as a part of ATC therapy in advanced STS

Exploratory

- Assess tumor antigen specificity
- Investigate and characterize immune status and nature of anti-tumor immune responses

Endpoints

Primary

- Change in T-cell level in tumour tissue from Baseline to end of Step 1
- Adverse Events (AE) related to LTX-315 or the combination of LTX-315 and ACT from Baseline to end of treament (EoT) (Step 2)

Secondary

- Total number of CD8+ T cells in the final infusion product
- Anti-tumor effect assed by ORR, CBR and PFS

Exploratory

- Identification of tumor-antigen specific T cells in tumor tissue and peripheral blood
- Change in immunological parameters from Baseline to 15 m after EoT.

Key Inclusion Criteria

- Histologically confirmed advanced/metastatic STS that is stable or has progressed or after minimum 1 line of systemic treatment
- At least one lesion accessible for injection
- At least one measurable non-injected lesion used for RECIST 1.1 response assessment
- ECOG Performance status (PS): 0 - 1

Key Exclusion Criteria

- A history of clinically significant active systemic autoimmune disease requiring anti-inflammatory or immunosuppressive therapy within the last 3 months.
- Received an investigational drug therapy within 4 weeks prior to study
- External radiotherapy or cytotoxic chemotherapy within the last 4 weeks prior to study
- Currently taking any agent with a known effect on the immune system. Patients are allowed to be on a stable dose of corticosteroids (up to 10 mg daily predniso-lone or equivalent) for at least 2 weeks prior to LTX-315 administration
- Clinically active or unstable metastases in the central nervous system as assessed by the treating physician

Patient characteristics

ID	Histology	Sites of disease	Prior systemic treatment	Status at inclusion	ECOG PS at inclusion
01-1001	DSRCT	Breast, lymph nodes, pleura, liver	Epirubicin/Cyclophosphamide, Atezolizumab, Vincristine/Ifosfamide/ Doxorubicin/Etoposide, Evincristine/ Actinomycin D/Ifosfamide	PD	0
01-1002*	-	-	-	-	
01-1003	Leiomyosarcoma	Muscle, bone, lung	Doxorubicin/Olaratumab, Olaratumab	PD	0
01-1004	DSRCT	Subcutis, lymph nodes	Doxorubicin/Cisplatin/Etoposide, Doxorubicin/Vincristine/Actinomycin D/ Etoposide/Ifosfamide/ Cyclophosphamide, Pazopanib	PD	1
01-1005	Solitary fibrous tumour	Abdomen	Doxorubicin/Dexrazoxane	SD	1
01-1006	Sclerosing epithelial fibrosarcoma	Eye, bone	Vincristine/Ifosfamide/Doxorubicin/ Etoposide Vincristine/Actinomycin D/Ifosfamide Gemcitabin/Docetaxel, Gemcitabin/Paclitaxel, Gemcitabine	PD	1
01-1007	Solitary fibrous tumour	Parotid gland, lung, liver, kidney	Doxorubicin/Dexrazoxane	PD	0

Adverse Events during therapy

Adverse events reported by the investigator as related to LTX-315 and Adoptive Cell Transfer therapy

Therapy	AE	Grade 1-2 (n)	Grade 3-4 (n)
LTX-315	Diarrhoea	1	0
	Fever	1	0
	Itching	1	0
	Pain at injection site	4	0
	Redness at injections site	1	0
	Stomach pain	1	0
Chemotherapy	Anaemia	3	0
	Constipation	1	0
	Diarrhoea	1	0
	Dry mouth	1	0
	Fatigue	1	1
	Headache	1	0
	Haematuria	1	0
	Hot flashes	1	0
	Hypocalcaemia	1	0
	Hypomagnesaemia	1	0
	Hyponatraemia	1	0
	Hypophosphatemia	1	0
	Nausea	4	0
	Neutropenia	0	4
	Rash	2	0
	Stomach-ache	1	0
	Stomatitis	1	0
	Thrombocytopenia	0	4
	Vomiting	1	0
TIL	Chills	2	0
	Diarrhoea	1	0
	Dyspnoea	0	1
	Fatigue	0	1
	Fever	2	2
	Headache	1	0
	Rash	1	0
IL2	Anaemia	2	0
	Diarrhoea	3	0
	Elevated alkaline phosphate	1	0
	Elevated liver enzymes	1	0
	Fever	2	2
	Headache	1	0
	Rash	1	0
	Vomiting	1	0

Characteristics of infused cells

TILs were successfully expanded from 4 out of 6 patient. Cultivation failed from patient 01-1004 and 01-1006, hence they were withdrawn from the study after Step 1.

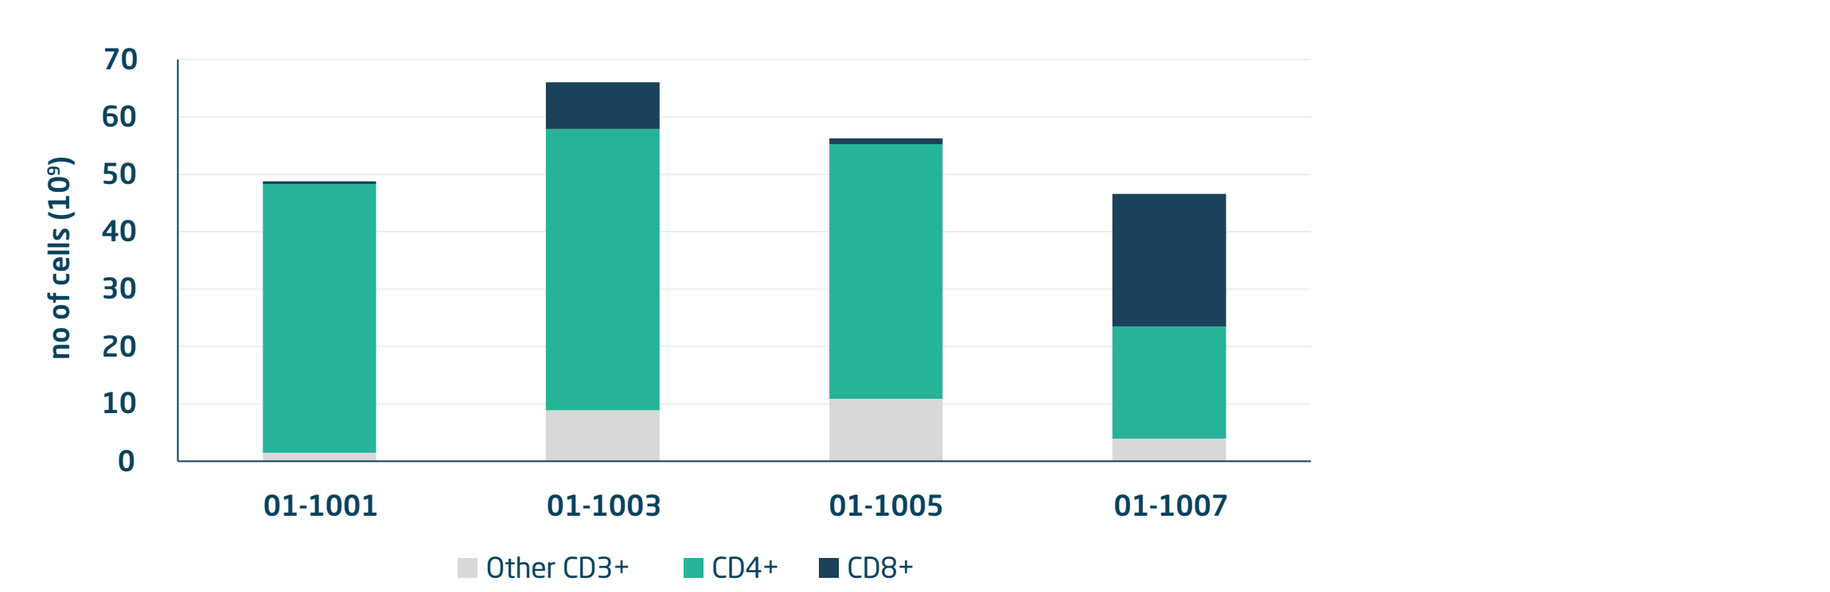
no of cells (10⁹)
01-1001
01-1003
01-1005
01-1007
Other CD3+
CD4+
CD8+

Bar plot shows the total number and phenotype of infused cells.
Each bar shows cumulated CD4+ (green), CD8+ (blue), and other CD3+ cells (grey).

Anti-tumor response assessed by RECIST 1.1

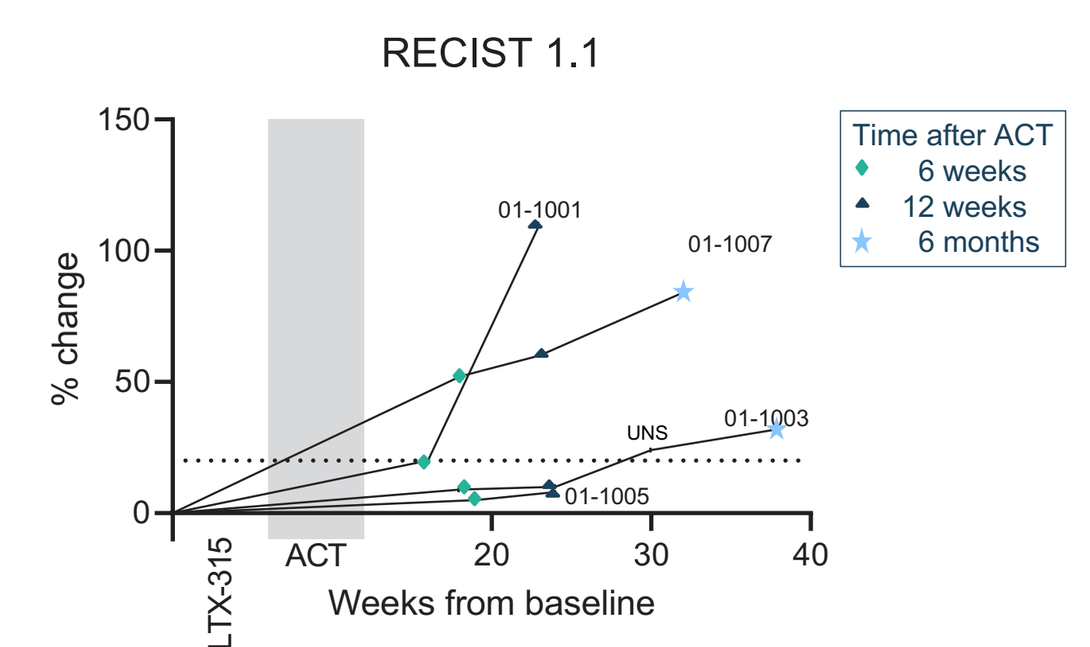
RECIST 1.1
Time after ACT
6 weeks
12 weeks
6 months
% change
01-1001
01-1007
01-1003
01-1005
UNS
ACT
LTX-315
Weeks from baseline

LTX-315 modulates the tumor environment (Step 1)

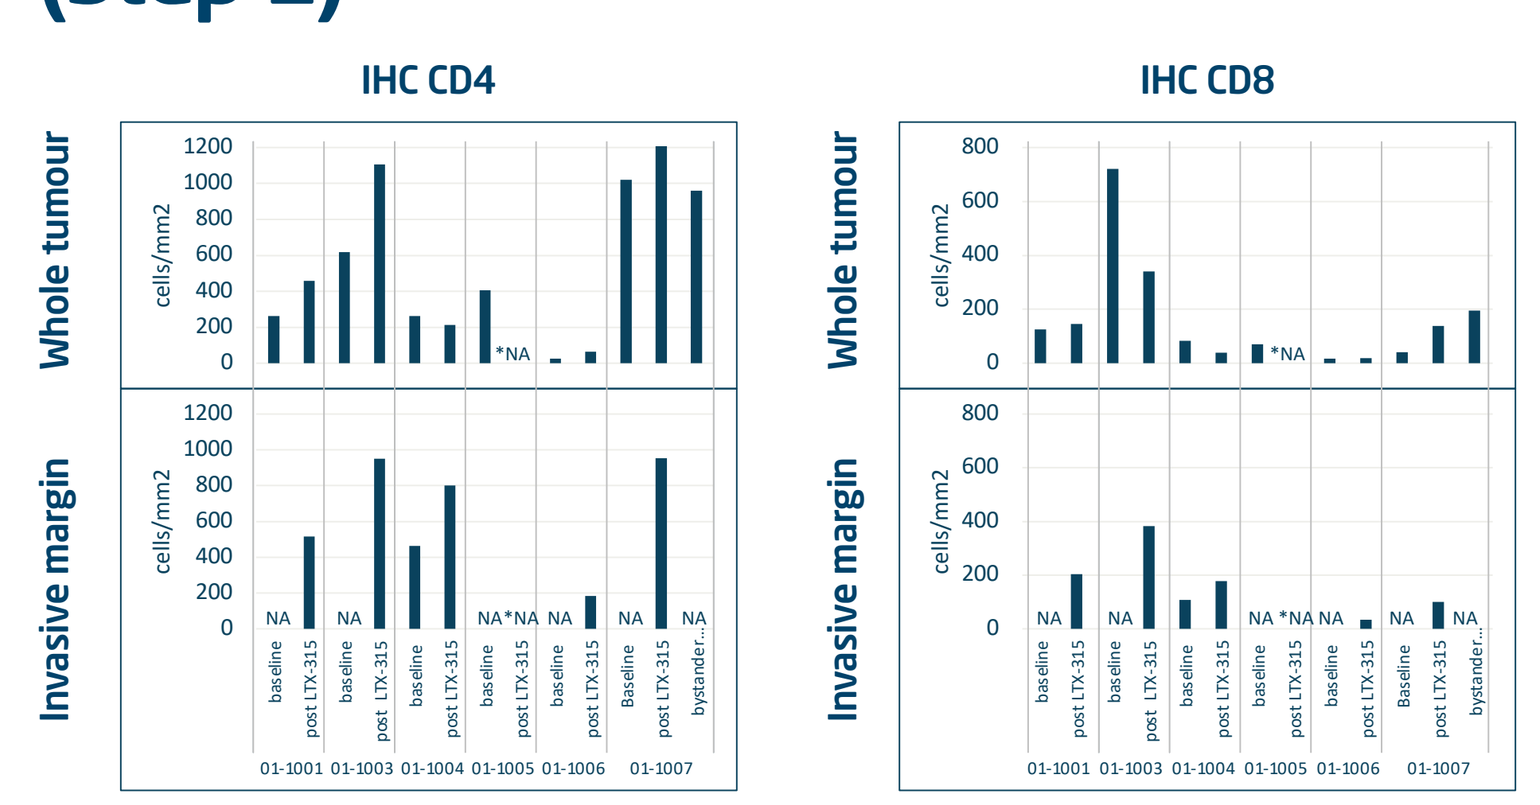
IHC CD4
IHC CD8
Whole tumour
Invasive margin
cells/mm2
01-1001
01-1003
01-1004
01-1005
01-1006
01-1007

*H&E staining of the tumor from pts 01-1005 showed complete necrosis post treatment and IHC was therefore not assessed

Modulation of the systemic immune response

T-cell response induced against tumor antigens

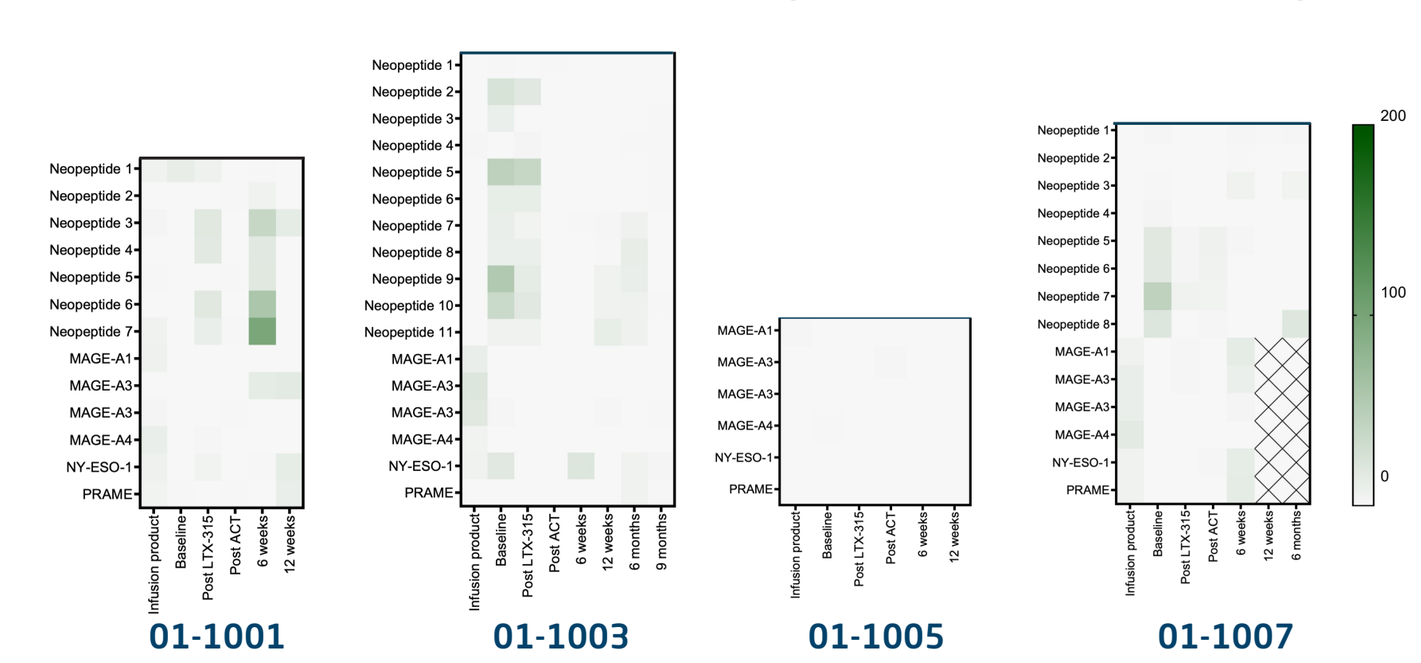
01-1001
01-1003
01-1005
01-1007

Heatmaps showing reactivity of PBMC and infused TIL towards predicted neo-peptides and selected Cancer Testis Antigens analysed with IFNγ ELIspot. Colour intensity shows the difference between spot count of the test samples (mean of triplicates) and the negative control.

T-cell response induced against Tumor Cell Line (TCL)

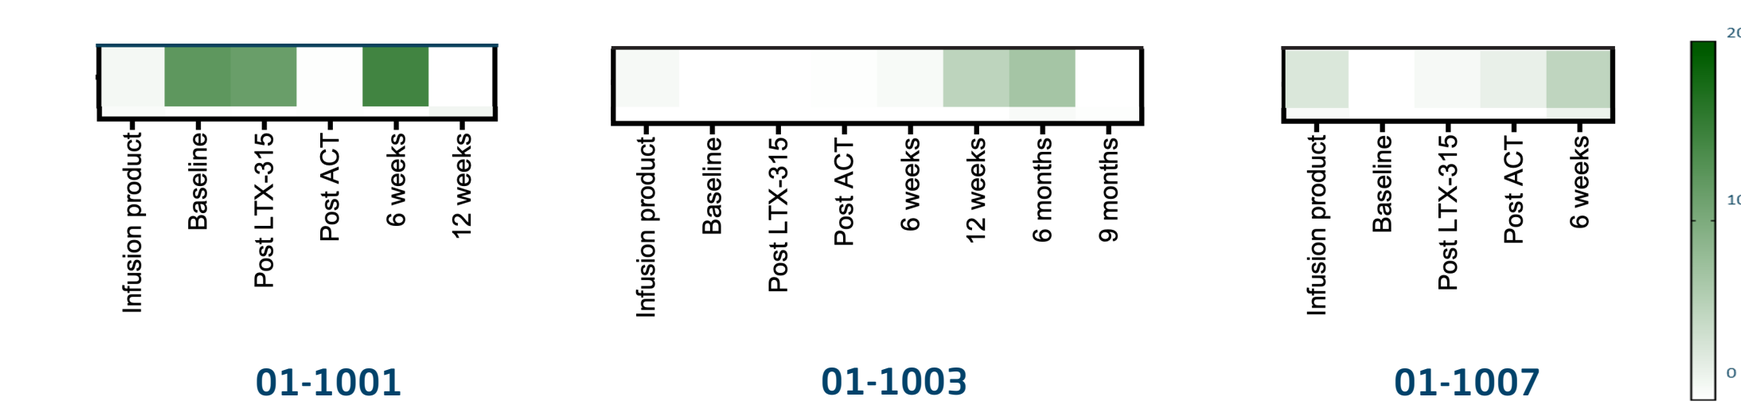
Infusion product
Baseline
Post LTX-315
Post ACT
6 weeks
12 weeks
6 months
9 months
01-1001
01-1003
01-1007

Heatmaps showing reactivity of PBMC and infused TIL towards autologous Tumor Cell Line analysed with IFNγ ELIspot. The tumor cell line derived from tumor lesion after LTX-315 injections were cultivated with the indicated PBMC/TIL. Colour intensity shows the difference between spot count of the test samples (mean of triplicates) and the negative control. Generation of TCL from pts 01-1005 failed.

LTX-315 treatment induces peripheral expansion of T cell clones

	Infusion product	PBMC in periphery						Tissue biopsy		
		baseline	Post LTX-315	Post ACT	6 w post ACT	12 w post ACT	6 m post ACT	Baseline	Post LTX-315	Post ACT bystander
01-1003	✓	✓	✓	✓	✓	✓	✓	✓		
01-1007	✓	✓	✓	✓	✓	✓	✓	✓ *	✓	✓

* T cells <100

Overview of samples for immunosequencing of the TCRβ locus

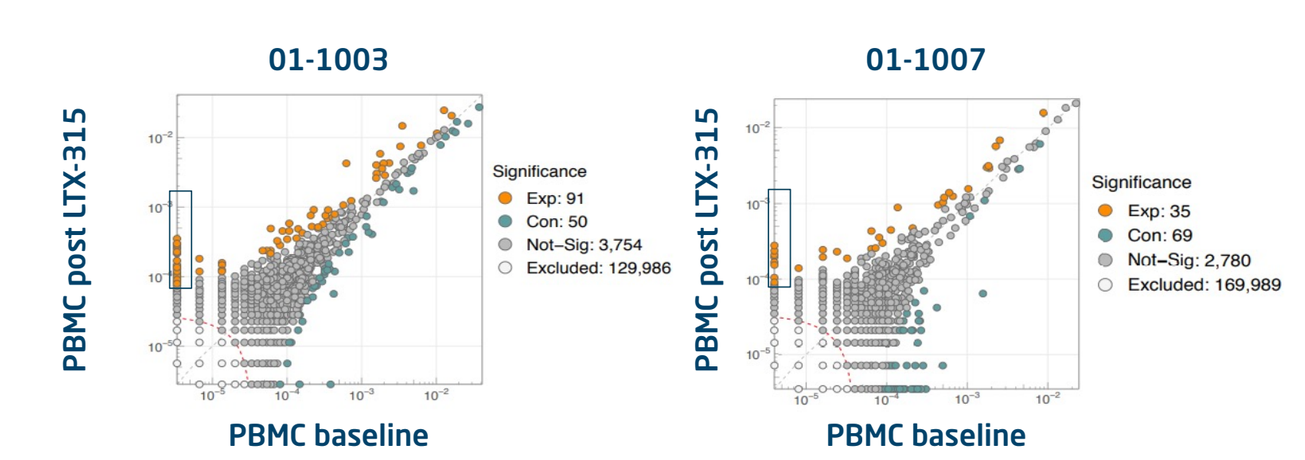
01-1003
01-1007
PBMC post LTX-315
PBMC baseline
Significance
Exp: 91
Con: 50
Not-Sig: 3,754
Excluded: 129,986
Exp: 35
Con: 69
Not-Sig: 2,780
Excluded: 169,989

- Comparing the TCR frequency between T-cell clones present at baseline and post LTX-315 show evidence of clonal expansion beyond that found in healthy controls over a two-week period (~10 clones).
- Both patients show evidence of newly detected T-cell clones post LTX-315 treatment (along the y-axis)

Treatment-associated peripheral expansion is maintained 6 months post treatment

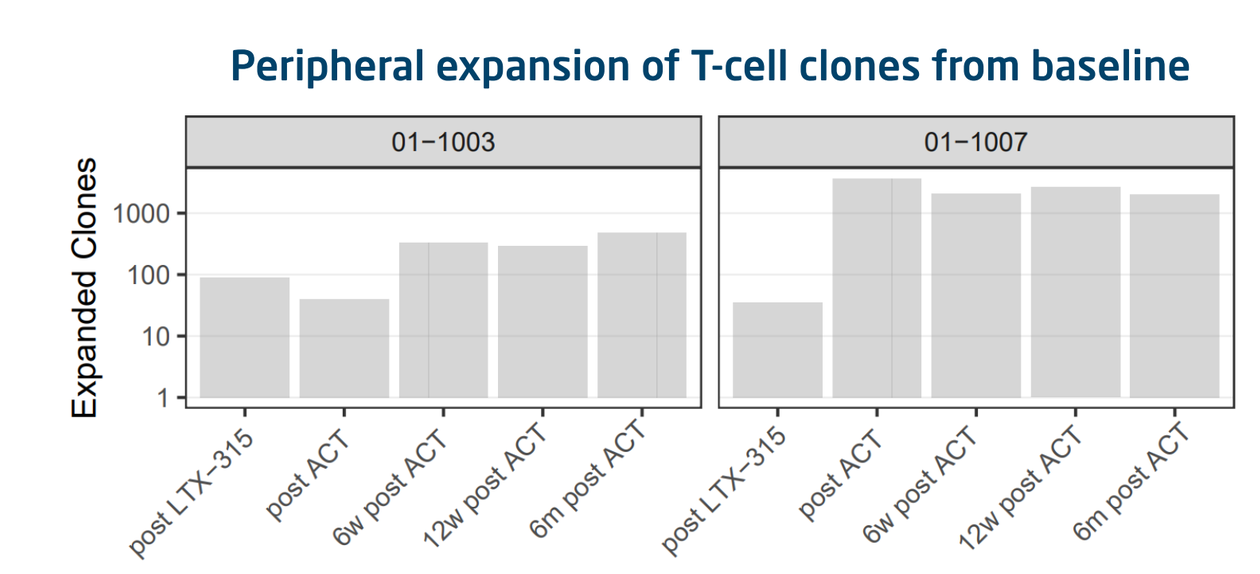
Peripheral expansion of T-cell clones from baseline
01−1003
01−1007
Expanded Clones
post LTX−315
post ACT
6w post ACT
12w post ACT
6m post ACT

- Both LTX-315 and ACT result in peripheral expansion
- High levels of clonal expansion are observed 6 months post ACT
- Peripheral expansion post LTX-315 was more pronounced than post ACT in patient 01-1003
- Patient 01-1007 showed very high levels of expansion after ACT therapy

T-cell clones that expand post baseline are present in injected lesion and in the bystander lesion

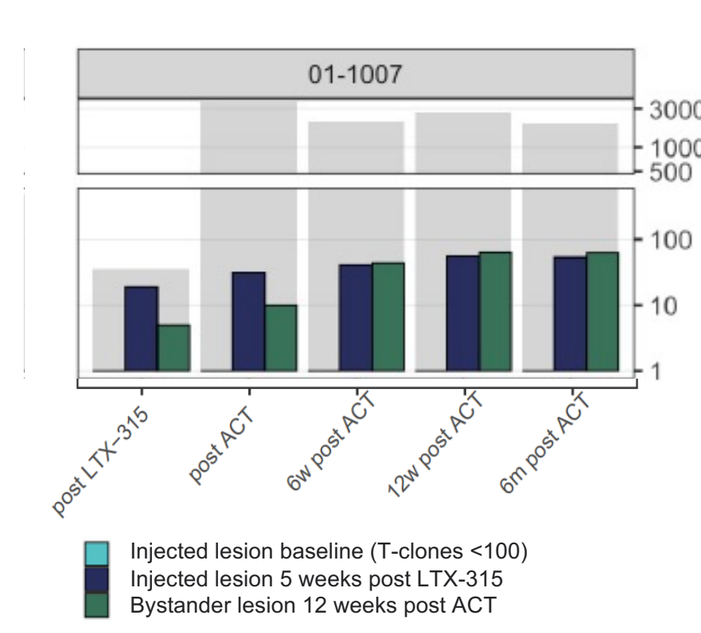
01-1007
post LTX-315
post ACT
6w post ACT
12w post ACT
6m post ACT
Injected lesion baseline (T-clones <100)
Injected lesion 5 weeks post LTX-315
Bystander lesion 12 weeks post ACT

- Many of the expanded T-cell clones in patient 01-1007 (grey bar) were also present in the injected lesion (blue bar) and in the bystander lesion (green bar) post treatment.
- The number of expanded T-cell clones that were present in both injected lesion and bystander increased over time.
- Patient 01-1003; tissue biopsy post LTX-315 was not available and similar analysis is therefore not included.

Key findings of immune response data

- TILs were successfully cultivated from the LTX-315 treated tumor in four out of six patients. Two patients did not receive full treatment (patient 01-1004, 01-1006). Common characteristics for the excluded patients were low abundance of TILs in the resected tumor, both at baseline and after LTX-315 treatment. Moreover, both patients had received 3 lines of prior treatment and had ECOG status 1 at screening.
- Exploratory analysis of immune responses were assessed for the patients that received ACT of cultivated TILs (01-1001, 01-1003, 01-1005, 01-1007);

→ Three out of four patients showed some level of reactivity against one or several Cancer Testis Antigens (CTA) in the infusion product and in PBMC collected after EoT
→ Two patients (01-1003, 01-1007) showed induced reactivity against an autologous Tumor Cell Line (TCL) post treatment
→ One patient (01-1001) showed induced reactivity against predicted neo-peptides

- T-cell receptor (TCR) repertoire analysis of infusion product, PBMC and tissue samples collected from patient 01-1003 and pts 01-1007 showed;

→ LTX-315 induced expansion of a significant number of T-cell clones in the periphery
→ Expanded peripheral T cell clones were present in tumor tissue post treatment
→ New T-cell clones were detected post LTX-315 treatment, they expanded significantly in the periphery, and were present in tumor tissue post treatment

Conclusion

- **In this hard-to-treat patient population, LTX-315 in combination with ACT therapy was able to stabilize the disease in 3 out of 4 patients that received the full treatment.**
- **LTX-315 was well tolerated with adverse events being mostly mild or moderate in severity.**
- **The treatment was shown to generate tumor-specific T cells, expand T-cell clones in the periphery, and generate *de novo* T-cell clones.**

REFERENCES

1. Sorbye, S.W. et al.; PlosOne (2011)
2. Smolle, M.A. et al.; Oncoimmunology (2021)
3. Sousa, L.M. et al.; Cancers (2021).
4. Haug, B.E. et al.; Journal of Medicinal Chemistry (2016)
5. Sveinbjørnsson, B. et al.; Future Medicinal Chemistry (2017)
6. Spicer, J. et al.; Cancer Research (2021)
7. Jebsen,N.L. et al.; Journal of Medical Case reports (2019)
8. Liao H-W. et al.; Cell Stress (2019)
9. Nestvold,J. et al.; Oncoimmunology (2017)
10. Eike, L-M. et al.; Oncotarget (2015)
11. Forveille, S. et al.; Cell Cycle (2015)

Collaborators

CCIT DENMARK, veracyte, Adaptive, Personalis, OncoImmunity

Lytix Biopharma AS | Sandakerveien 138, 0484 Oslo, Norway | E-mail: post@lytixbiopharma.com