INTRATUMORAL INJECTION OF LTX-315 IN COMBINATION WITH PEMBROLIZUMAB IN PATIENTS WITH ADVANCED MELANOMA REFRACTORY TO PRIOR PD-1/PD-L1 THERAPY: INTERIM RESULTS FROM THE ATLAS-IT-05 TRIAL

1051P

STÉPHANE DALLE[1], MIGUEL F SANMAMED[2], JOHN M KIRKWOOD[3], CAROLINE ROBERT[4], MARTA NYAKAS[5], THOMAS MARRON[6], LAURENT MORTIER[7], GRAEME CURRIE[8], VIBEKE SUNDVOLD[8], ØYSTEIN REKDAL[8], BALDUR SVEINBJØRNSSON[8], JACKIE EARABINO[8], KARSTEN BRUINS SLOT[8], ADI DIAB[9]

[1] CHU HÔPITAL LYON SUD, FRANCE; [2] CLINICA UNIVERSIDAD DE NAVARRA, SPAIN; [3] UNIVERSITY OF PITTSBURGH MEDICAL CENTRE – HILMAN CANCER CENTRE, USA; [4] GUSTAVE ROUSSY/PARIS SACLAY UNIVERSITY, FRANCE; [5] OSLO UNIVERSITY HOSPITAL-RADIUMHOSPITAL, NORWAY; [6] ICAHN SCHOOL OF MEDICINE AT MOUNT SINAI, USA;[7] CHU LILLE, FRANCE; [8] LYTIX BIOPHARMA AS, NORWAY; [9] MD ANDERSON CANCER CENTRE, USA

Lytix Biopharma

BACKGROUND

LTX-315 is a first-in-class oncolytic peptide of non-viral origin that is in development for intratumoral treatment of solid tumors (1,2)

Pre-clinical studies of LTX-315 demonstrate:

- Unique immunogenic cell death mode of action by causing mitochondrial lysis and disintegration of cytoplasmic organelles resulting in effective release of danger signals and a broad repertoire of tumor antigens (3-6)
- Reduced number of immunosuppressive cells (T reg and myeloid derived suppressor cells) (7)
- Enhanced infiltration of T cells and T cell clonality (8)
- Complete regression of injected and non-injected tumors (i.e. systemic immune response) (8-10)

Clinical studies of LTX-315 demonstrate:

- Enhanced infiltration of T cells and T cell clonality (11, 12)
- Regression of injected and non-injected tumors (i.e. systemic immune response) (12)
- Generation of tumor-specific T cells (13)

LTX-315 - UNIQUE MODE OF ACTION RESULTS IN EFFECTIVE RELEASE OF POTENT IMMUNOSTIMULANTS AND ANTIGENS

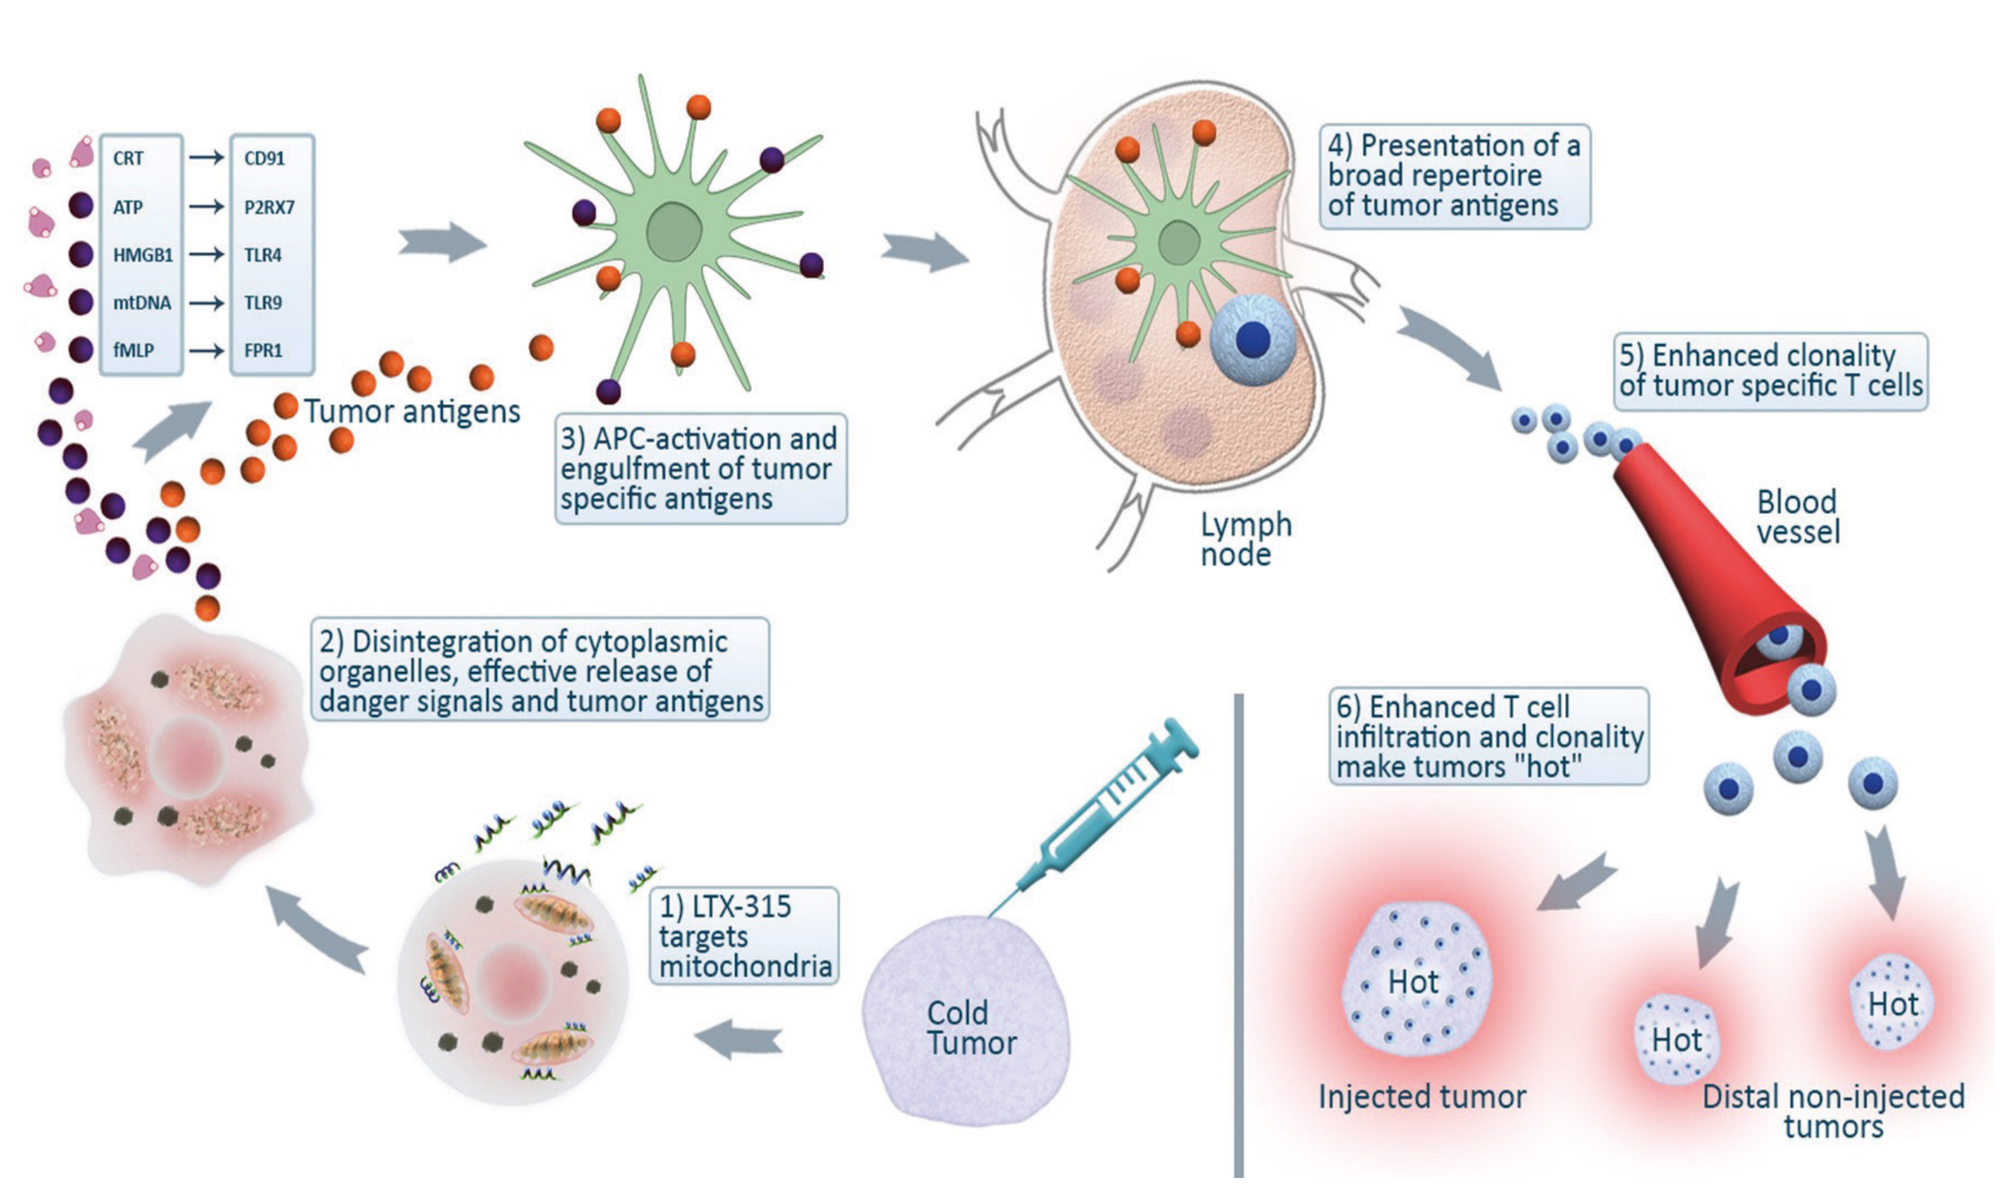
CRT → CD91
ATP → P2RX7
HMGB1 → TLR4
mtDNA → TLR9
fMLP → FPR1
Tumor antigens
3) APC-activation and engulfment of tumor specific antigens
4) Presentation of a broad repertoire of tumor antigens
Lymph node
5) Enhanced clonality of tumor specific T cells
Blood vessel
2) Disintegration of cytoplasmic organelles, effective release of danger signals and tumor antigens
1) LTX-315 targets mitochondria
Cold Tumor
6) Enhanced T cell infiltration and clonality make tumors "hot"
Hot
Injected tumor
Distal non-injected tumors

STUDY OBJECTIVES AND ENDPOINTS

Objectives

Evaluate the efficacy and safety of intratumoral LTX-315 in combination with pembrolizumab in patients with Stage IIIB-IVm1b melanoma, who have progressed on or after prior treatment with a PD-1/PD-L1 inhibitor

Primary Efficacy Endpoint

- Objective Response Rate (ORR) using RECIST v1.1 criteria assessed by investigators
- Disease Control Rate (DCR) using RECIST v1.1 criteria assessed by investigators

Secondary Efficacy Endpoint

- Regression of injected lesions assessed by CT/MRI or ultrasound measurements by investigators
- Incidence and severity of treatment emergent adverse events related to LTX-315

STUDY DESIGN

Open label, single-arm, phase II study with 10 study sites in Europe and the US (NCT04796194)

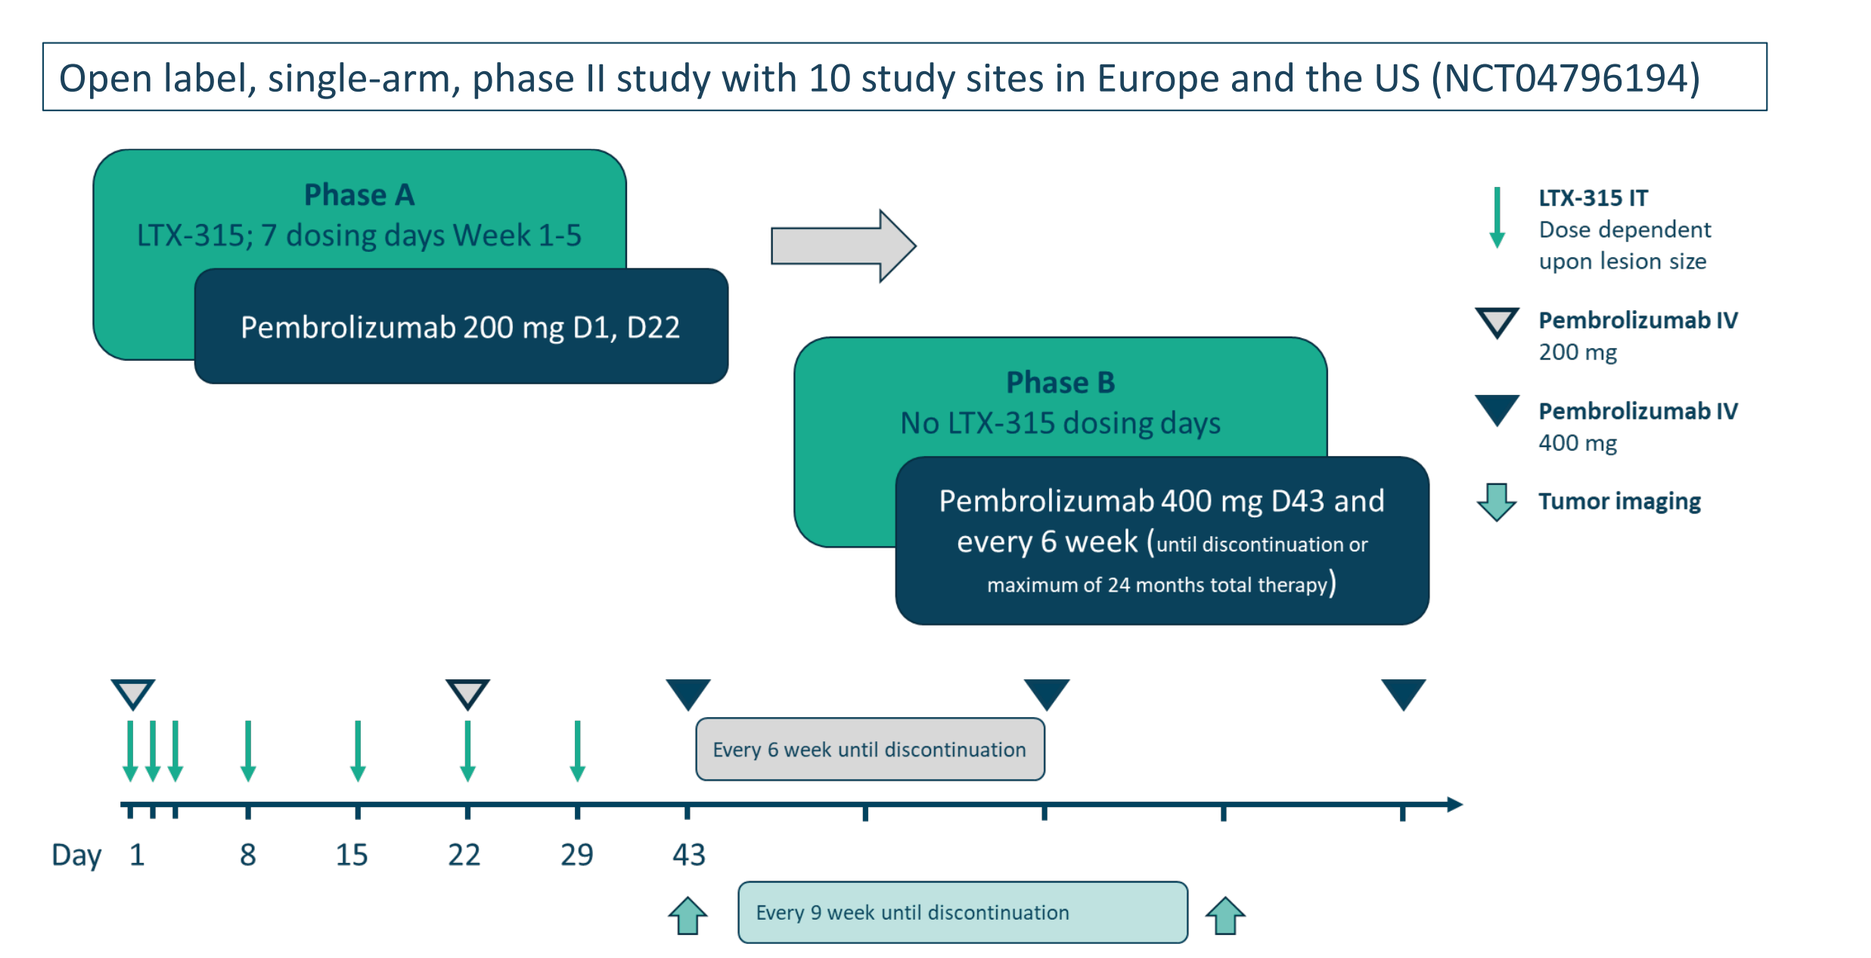
Open label, single-arm, phase II study with 10 study sites in Europe and the US (NCT04796194)
Phase A
LTX-315; 7 dosing days Week 1-5
Pembrolizumab 200 mg D1, D22
Phase B
No LTX-315 dosing days
Pembrolizumab 400 mg D43 and every 6 week (until discontinuation or maximum of 24 months total therapy)
LTX-315 IT
Dose dependent upon lesion size
Pembrolizumab IV 200 mg
Pembrolizumab IV 400 mg
Tumor imaging
Every 6 week until discontinuation
Day 1 8 15 22 29 43
Every 9 week until discontinuation

KEY INCLUSION AND EXCLUSION CRITERIA

- Histologically confirmed, Stage IIIB-IVm1b unresectable melanoma
- Confirmed disease progression on or after prior treatment with PD-1/PD-L1 inhibitor
- ≤3 prior lines of systemic treatment for metastatic disease
- ECOG performance status of 0-1
- At least 1 superficial, non-visceral tumor lesion accessible for injection – superficial lymph nodes with metastatic disease can also be injected
- LDH ≤ 2 x ULN
- No ocular or mucosal melanoma diagnosis

PATIENT DISPOSITION

	Number of patients
Patients with available data at cutoff date (13 September 2023)	20
Patients with melanoma diagnosis	19
Patients included in Safety Analysis Set (SAS)	20
Patients included in Efficacy Analysis Set (EAS)	14
Reasons for exclusion from EAS	
• No available post-baseline scan at cutoff date yet	3
• Patient withdrew consent prior to first post-baseline scan	1
• Only 1 available post-baseline scan and insufficient follow-up	1
• No melanoma diagnosis*	1

*Patient with acinic cell carcinoma enrolled under earlier protocol version

Median duration on study was 15 weeks at cutoff date

BASELINE CHARACTERISTICS

Baseline characteristic	All patients (N=20)
Mean age (range)	68 years (42-91)
Sex	
Female	9 (45%)
Male	11 (55%)
ECOG	
0	16 (80%)
1	4 (20%)
Melanoma stage	
Stage IIIB	1 (5%)
Stage IIIC	4 (20%)
Stage IIID	2 (10%)
Stage IVm1a	7 (35%)
Stage IVm1b	5 (25%)
Prior systemic treatment for metastatic disease*	
BRAF/MEK	4 (20%)
PD-(L)1 monotherapy	10 (50%)
PD-(L)1+ CTLA-4	8 (40%)
PD-(L)1 + other	6 (30%)
CTLA-4+ other	1 (5%)

Baseline characteristic	All patients (N=20)
Prior systemic treatment lines in metastatic setting	
0	1 (5%)**
1	11 (55%)
2	6 (30%)
3	2 (10%)
Prior lines of treatment with checkpoint inhibitor	
0	1 (5%)**
1	6 (30%)
≥2	13 (65%)
BRAF status	
Positive	5 (25%)
Negative	15 (75%)
LDH	
Normal	9 (45%)
>ULN	11 (55%)

*More than 100% as some patients received >1 systemic metastatic treatment line
**Patient with acinic cell carcinoma
All melanoma patients had prior surgery

BEST OVERALL RESPONSE - RECIST V1.1

Best overall response (RECIST v1.1)	N (%)
Complete response	0 (0%)
Partial Response*	1 (7%)
Stable Disease	5 (36%)
Progressive Disease	8 (57%)

Objective Response Rate (ORR) = 7% (95% CI 1-30%)
Disease Control Rate (DCR)= 43% (95% CI 20-70%)

*Confirmed PR

BEST CHANGE IN RECIST TARGET LESIONS

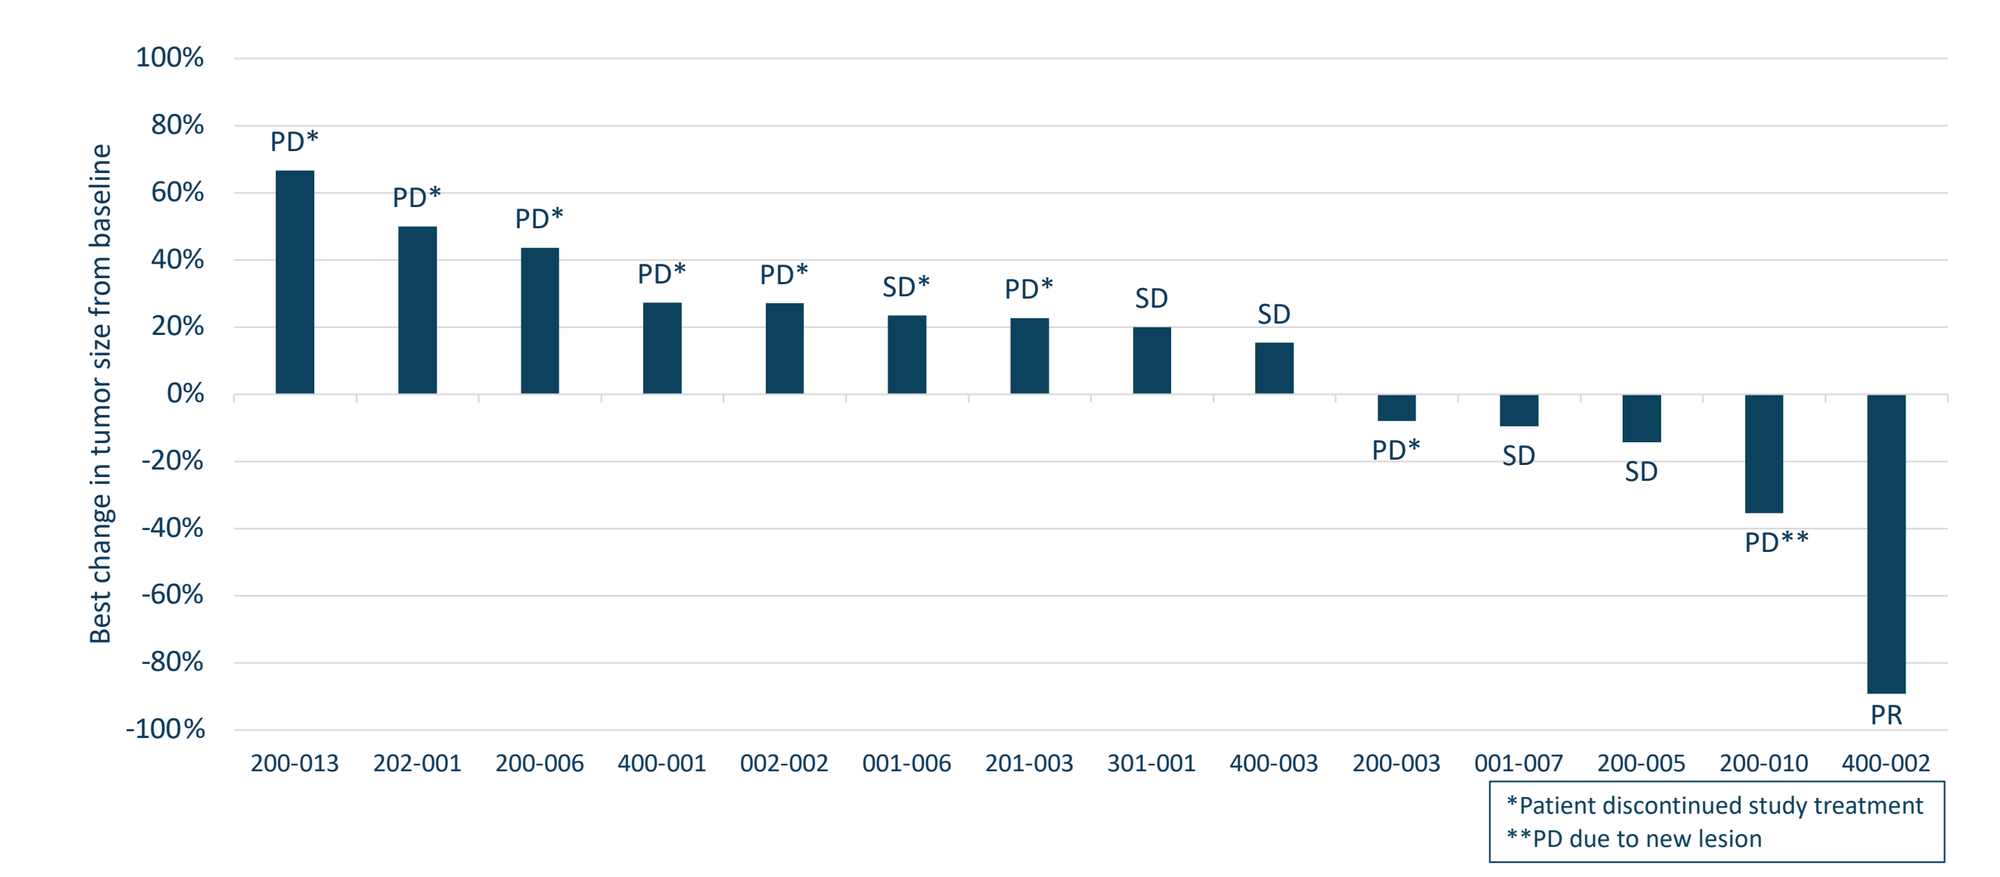
Best change in tumor size from baseline
100% 80% 60% 40% 20% 0% -20% -40% -60% -80% -100%
PD* PD* PD* PD* PD* SD* PD* SD SD PD* SD SD PD** PR
200-013 202-001 200-006 400-001 002-002 001-006 201-003 301-001 400-003 200-003 001-007 200-005 200-010 400-002
*Patient discontinued study treatment
**PD due to new lesion

CHANGE IN RECIST TARGET LESIONS

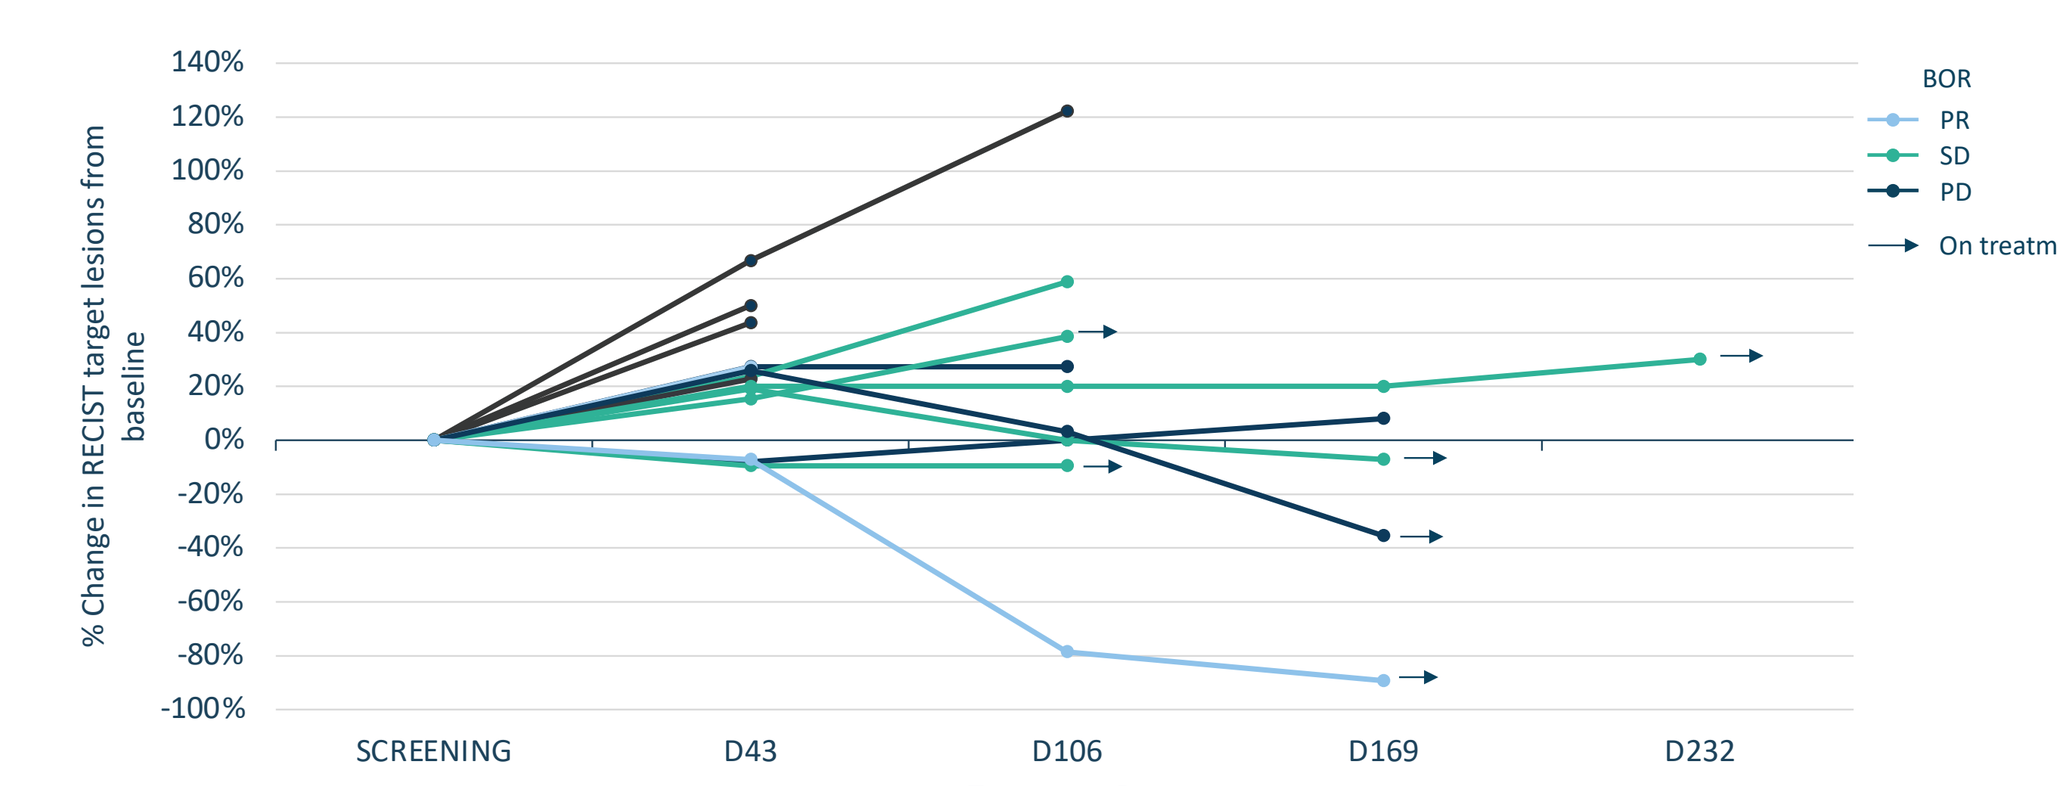
% Change in RECIST target lesions from baseline
140% 120% 100% 80% 60% 40% 20% 0% -20% -40% -60% -80% -100%
SCREENING D43 D106 D169 D232
Treatment Day
BOR
PR
SD
PD
On treatment

RESPONSE ASSESSMENTS PER RECIST V1.1

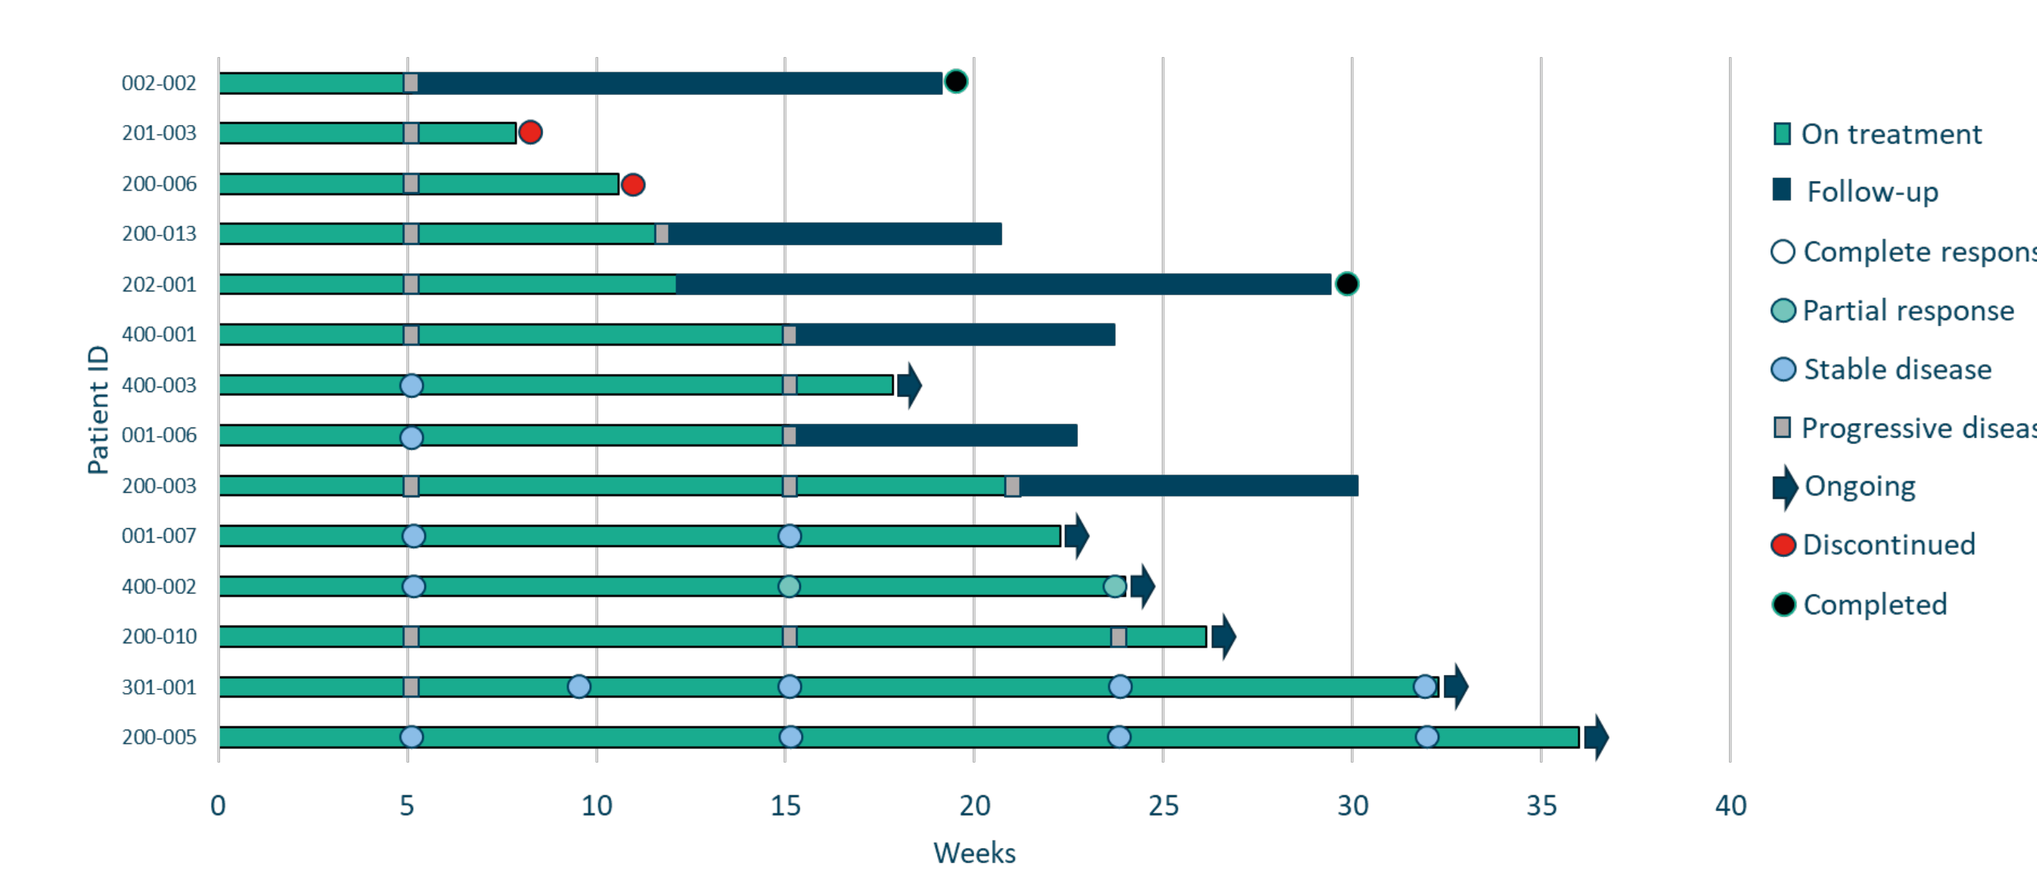
Patient ID
002-002
201-003
200-006
200-013
202-001
400-001
400-003
001-006
200-003
001-007
400-002
200-010
301-001
200-005
0 5 10 15 20 25 30 35 40
Weeks
On treatment
Follow-up
Complete response
Partial response
Stable disease
Progressive disease
Ongoing
Discontinued
Completed
Patient 300-001: only assessments by ultrasound

RESPONSE IN INJECTED LESIONS

- 9 out of 21 (43%) evaluable injected lesions showed complete regression by CT scan as best response after start of treatment*
- Complete regression was shown in 3 out of 11 (27%) evaluable patients by CT scan

*Analysis includes EAS population, except:
- Injected lesions that were not injected per injection plan
- Injected lesions without baseline assessment of lesion size

BASELINE	DAY 8 - POST LTX-315 DOSING	DAY 43 - POST LTX-315 DOSING
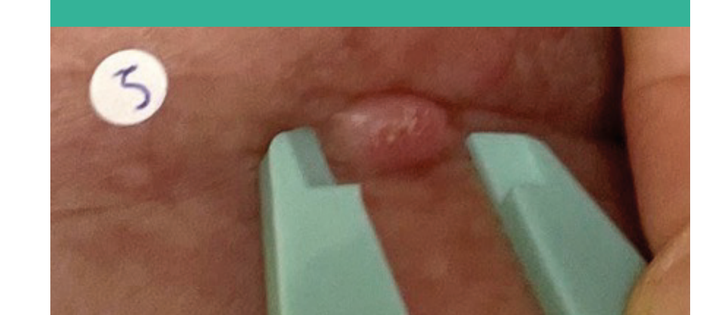	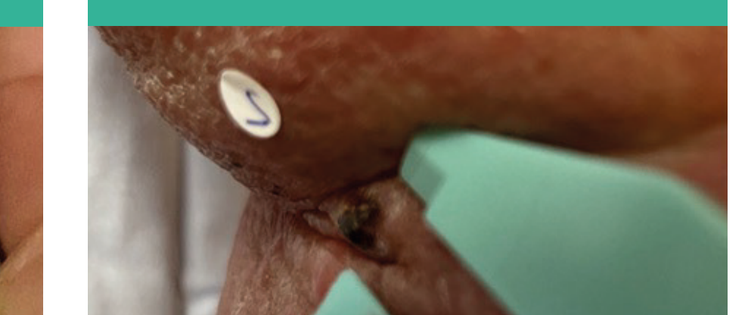	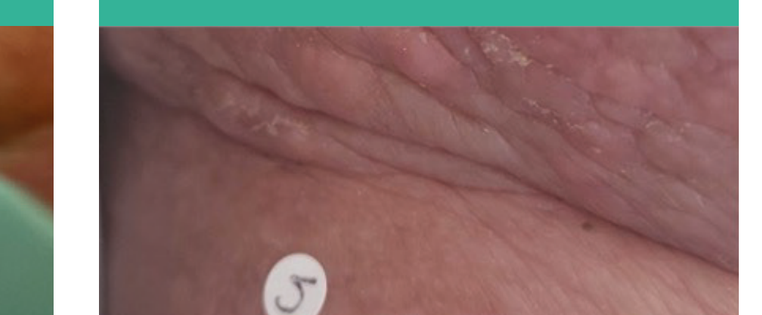
Mean of two longest perpendicular diameters **12 mm**	**NE**	**0 mm**

OVERVIEW OF TREATMENT EMERGENT ADVERSE EVENTS (>10%) IN SAS

Preferred AE term	Patients n (%)
Injection site pain	15 (75%)
Asthenia	5 (25%)
Pruritus	5 (25%)
Anemia	5 (25%)
Fatigue	4 (20%)
Injection site erythema	4 (20%)
Injection site swelling	3 (15%)
Hypertension	3 (15%)

LTX-315 TREATMENT-RELATED ADVERSE EVENTS (>10%) IN SAS

Preferred AE term	Grade 1-2	Grade 3	Grade 4	Grade 5	Patients - n (%)
Injection site pain	10 (50%)	5 (25%)	0	0	15 (75%)
Injection site erythema	3 (15%)	1 (5%)	0	0	4 (20%)
Injection site swelling	3 (15%)	0	0	0	3 (15%)

- The most common (>10%) LTX-315 treatment-related adverse events were related to injections and mostly mild, self-limiting and manageable in clinical practice.
- There was no increase in immune-related adverse events.
- No grade 4-5 treatment-related adverse events were reported

CASE - MELANOMA PATIENT WITH CLINICALLY RELEVANT SYSTEMIC RESPONSE

- 75-year-old male with Stage IVm1a, nodular melanoma (BRAF positive)
- Multiple metastases in lymph nodes and gluteal muscle at baseline
- Prior treatment with nivolumab (adjuvant setting) and BRAF/MEK inhibitor (metastatic setting)
- Treated with in total 20 intratumoral LTX-315 injections in 4 lesions on pre-scribed dosing days and 2 cycles (200 mg) + 3 cycles (400 mg) pembrolizumab
- Non-injected RECIST target lesion in left gluteal muscle
- Partial response as best overall response at cutoff date with RECIST target lesion shrinkage of 89%

NON-INJECTED LESION

	RECIST target lesion
BASELINE 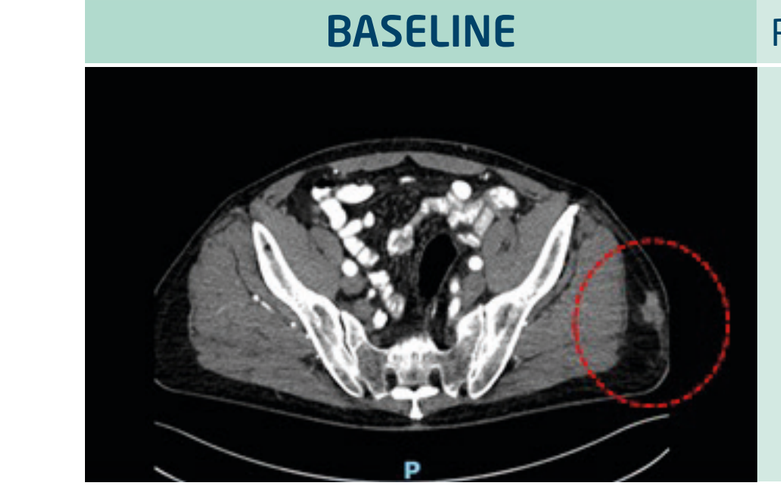	28 mm
DAY 43 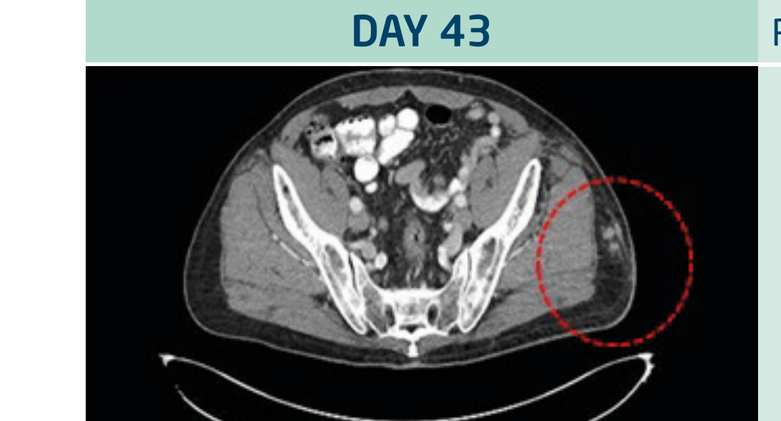	26 mm
DAY 169 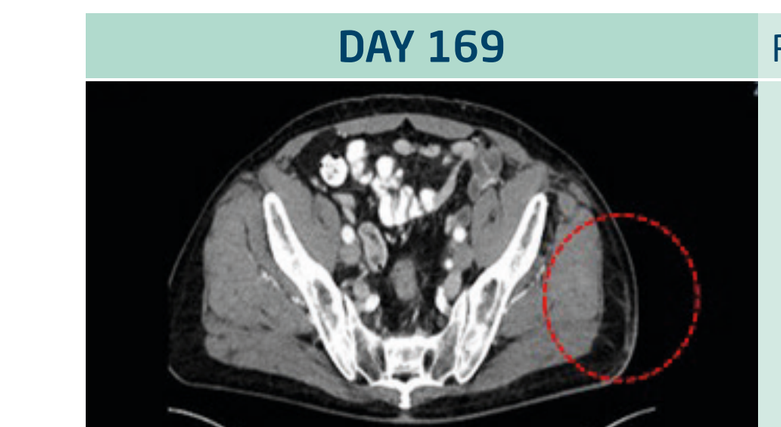	3 mm **89% reduction**

Superficial lesion located in gluteus muscle

INJECTED LESIONS

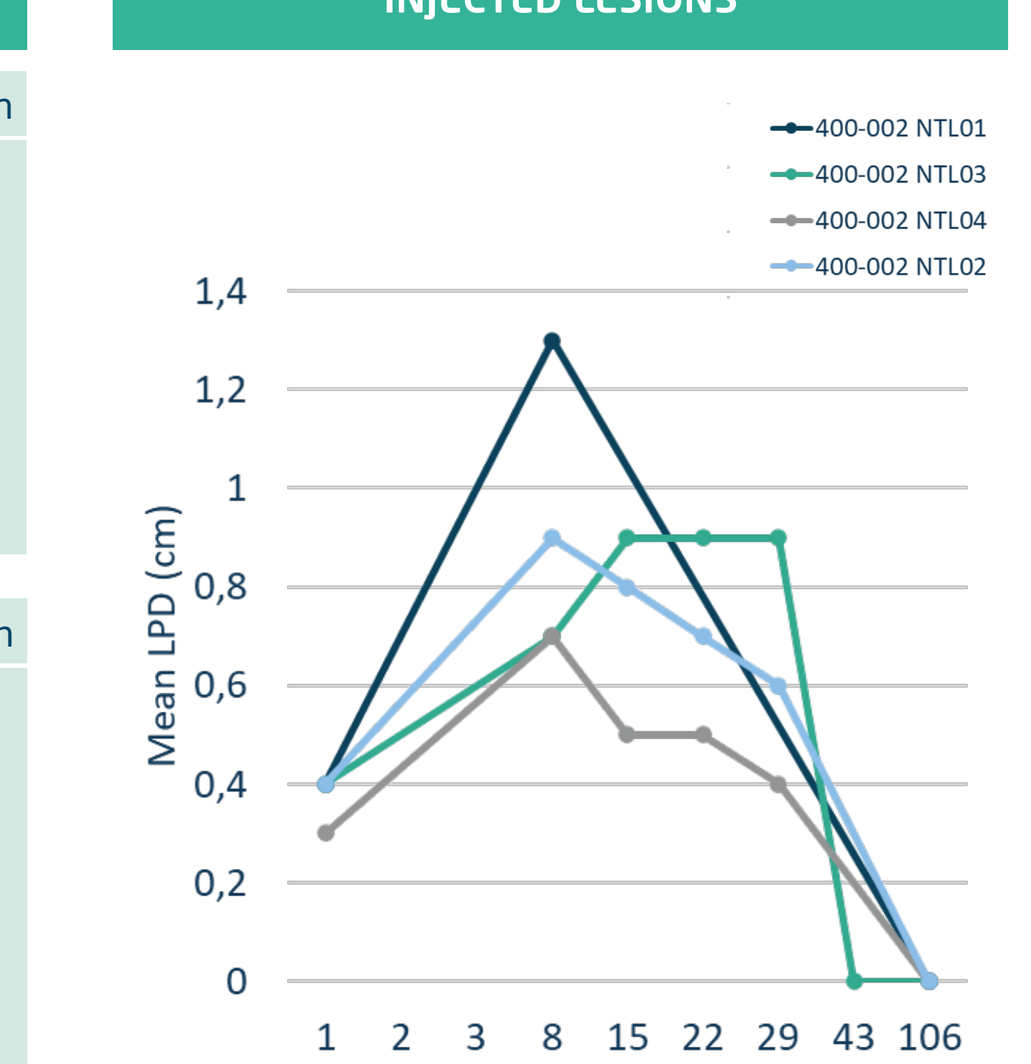
400-002 NTL01
400-002 NTL03
400-002 NTL04
400-002 NTL02
Mean LPD (cm)
1,4 1,2 1 0,8 0,6 0,4 0,2 0
1 2 3 8 15 22 29 43 106
Study Day

Longest perpendicular diameter (LPD) measurements based on caliper/ultrasound from Day 1-29; thereafter on CT scan.

CONCLUSION

- The combination regimen demonstrates preliminary signs of tumor shrinkage and prolonged stabilization in heavily pre-treated patients with PD-1/PD-L1 inhibitor refractory metastatic melanoma.
 - → Enrolled patients had generally poor prognostic factors and some patients had also failed BRAF/MEK inhibition.
- The efficacy signal is encouraging with a disease control rate of 43% and 1 patient achieving a partial response to date.
- There is evidence of tumor shrinkage in both injected and in non-injected lesions.
- Intratumoral treatment with LTX-315 is well-tolerated with mild to moderate treatment-related adverse events.
- Adverse events related to the intratumoral injections were generally self-limited and easily manageable in clinical practice.
- The trial is currently ongoing and data are considered immature – further details will be shared in a future presentation.

REFERENCES

1. Haug, B.E. et al; Journal of Medicinal Chemistry (2016).
2. Sveinbjørnsson, B. et al.; Future Medicinal Chemistry (2017).
3. Zhou, H. et al.; Oncotarget (2015).
4. Eike, L-M. et al.; Oncotarget (2015).
5. Forveille, S. et al.; Cell Cycle (2015).
6. Zhou, H. et al.; Cell Death Disease (2016).
7. Yamazaki, T. et al.; Cell Death and Differentiation (2016).
8. Camilio, K. et al.; Cancer Immunology Immunotherapy (2014).
9. Camilio, K. et al.; Oncoimmunology (2014).
10. Nestvold, J. et al.; Oncoimmunology (2017).
11. Jebsen, N et al.; Journal of Medical Case Reports (2019).
12. Spicer, J. et al.; Clin Cancer Res (2021).
13. Nielsen, M et al. Journal of Clinical Oncology (2022).

DoI Stephane Dalle: research grants and advisory board participation fees paid to institution by MSD, BMS, Pierre Fabre; SD spouse is a Sanofi employee.

Patient accrual (SAS): CHU Lyon 5; UPMC 3; MD Anderson 3; CU Navarra 3; CHRU Lille 2; Radiumhospital 2; Gustave Roussy 1; Mount Sinai 1.

Contact info: post@lytixbiopharma.com