The amphiphatic ß(2,2)-amino acid LTX-401 induces complete regression of experimental hepatocellular carcinoma

2347

BRYNJAR MAUSETH[1,2,5], JI-HUA SHI[2], KETIL ANDRÉ CAMILIO[3,5], ØYSTEIN REKDAL[4,5], BALDUR SVEINBJØRNSSON[4,5] AND PÅL-DAG LINE[1,2]
[1]Institute of Clinical Medicine, University of Oslo, Oslo, Norway
[2]Division of Cancer, Surgery and Transplantation, Oslo University Hospital, Rikshospitalet, Oslo, Norway
[3]Institute for Cancer Research, Department of Tumor Biology, Oslo University Hospital, Oslo, Norway
[4]Department of Medical Biology, University of Tromsø, Norway
[5]Lytix Biopharma, Oslo, Norway

Lytix Biopharma

Introduction

Hepatocellular carcinoma is the sixth most common cancer in the world and third most common cause of cancer mortality. While standard treatment includes surgical resection or transplantation, patients having unresectable tumors or widespread metastatic disease typically receive systemic chemotherapy. However, current pharmacological therapies have severe side effects and are seldom curative, thus illustrating the impending need to pursue new and improved therapeutic approaches.

The amphipathic ß(2,2)-amino acid LTX-401 is a novel cytolytic compound designed for intratumoral administration. Cancer cells treated with LTX-401 suffer irreversible damage to plasma membranes, resulting in cell death by necrosis and subsequent loss of intracellular content, including immune stimulating 'danger signals' or damage-associated molecular patterns molecules (DAMPs). Earlier in vivo studies have demonstrated potent anticancer effect against syngeneic murine B16 melanomas, presumably by engagement of antitumor immune responses (Eike et al. 2016).

Aim

The aim of the present study was to investigate the *in vitro* mechanism of action by LTX-401 followed by an assessment of the *in vivo* anti cancer efficacy against experimental hepatocellular JM1 carcinoma.

LTX-401

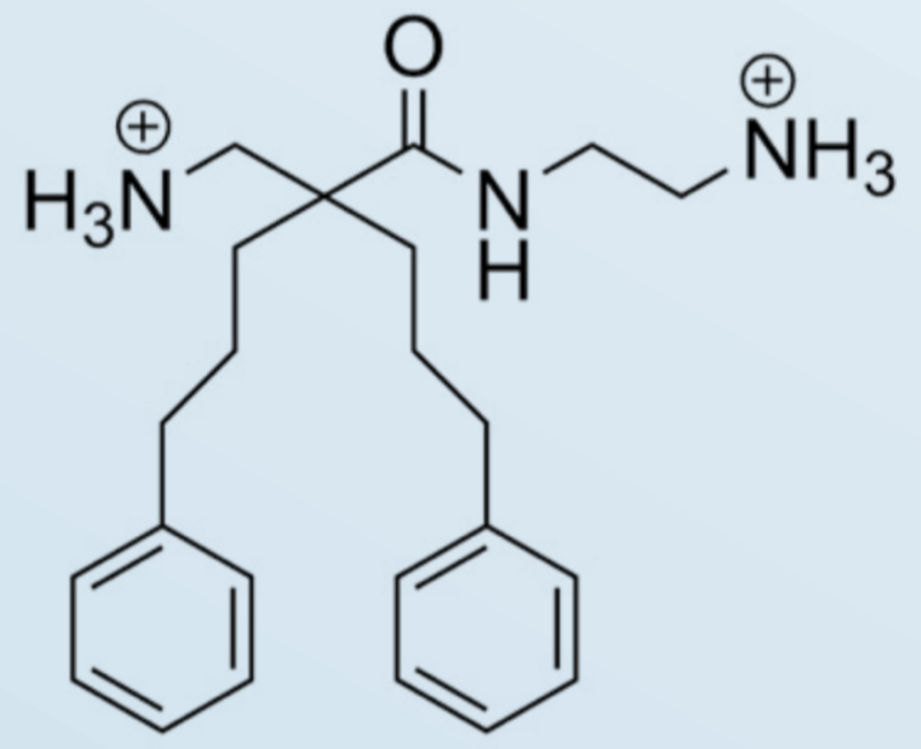

Chemical structure of LTX-401 (N-(2-aminoethyl)-2-(aminomethyl)-5-phenyl-2-(3-phenylpropyl) pentanamide). The molecule has an overall net charge of +2 with two lipophilic side chains connected to the ß-carbon

Results

Table 1 - LTX-401 displays high cytotoxic activity against a wide range of cancer cell lines and displays selectivity compared to human red blood cells.

Cell lines	LTX-401 IC_{50}^{a} ± SD (µM)
JM1 (Rat hepatocellular carcinoma)	22.8 ± 3.3
HEPG2 (Human hepatocellular carcinoma)	35.4 ± 0.6
BEL7402 (Human hepatocellular carcinoma)	26.7 ± 0.4
B16F1 (Murine skin malignant melanoma)	23.3 ± 3.9
MDA-MB-435S (Human malignant melanoma)	13.5 ± 1.4
Malme-3M (Human malignant melanoma)	19.3 ± 0.4
HT-29 (Human colorectal adenocarcinoma)	31.7 ± 2.9
A375 (Human skin malignant carcinoma)	30 ± 0.9
SK-N-AS (Human neuroblastoma)	30.6 ± 0
RBC (Human)	>1087

[a] The peptide concentration needed to kill 50 % of the cells

Fig. 3 - JM1 cells treated with LTX-401 release DAMPs, such as High Mobility Group Box 1 protein (HMGB1), ATP and cytochrome C in vitro

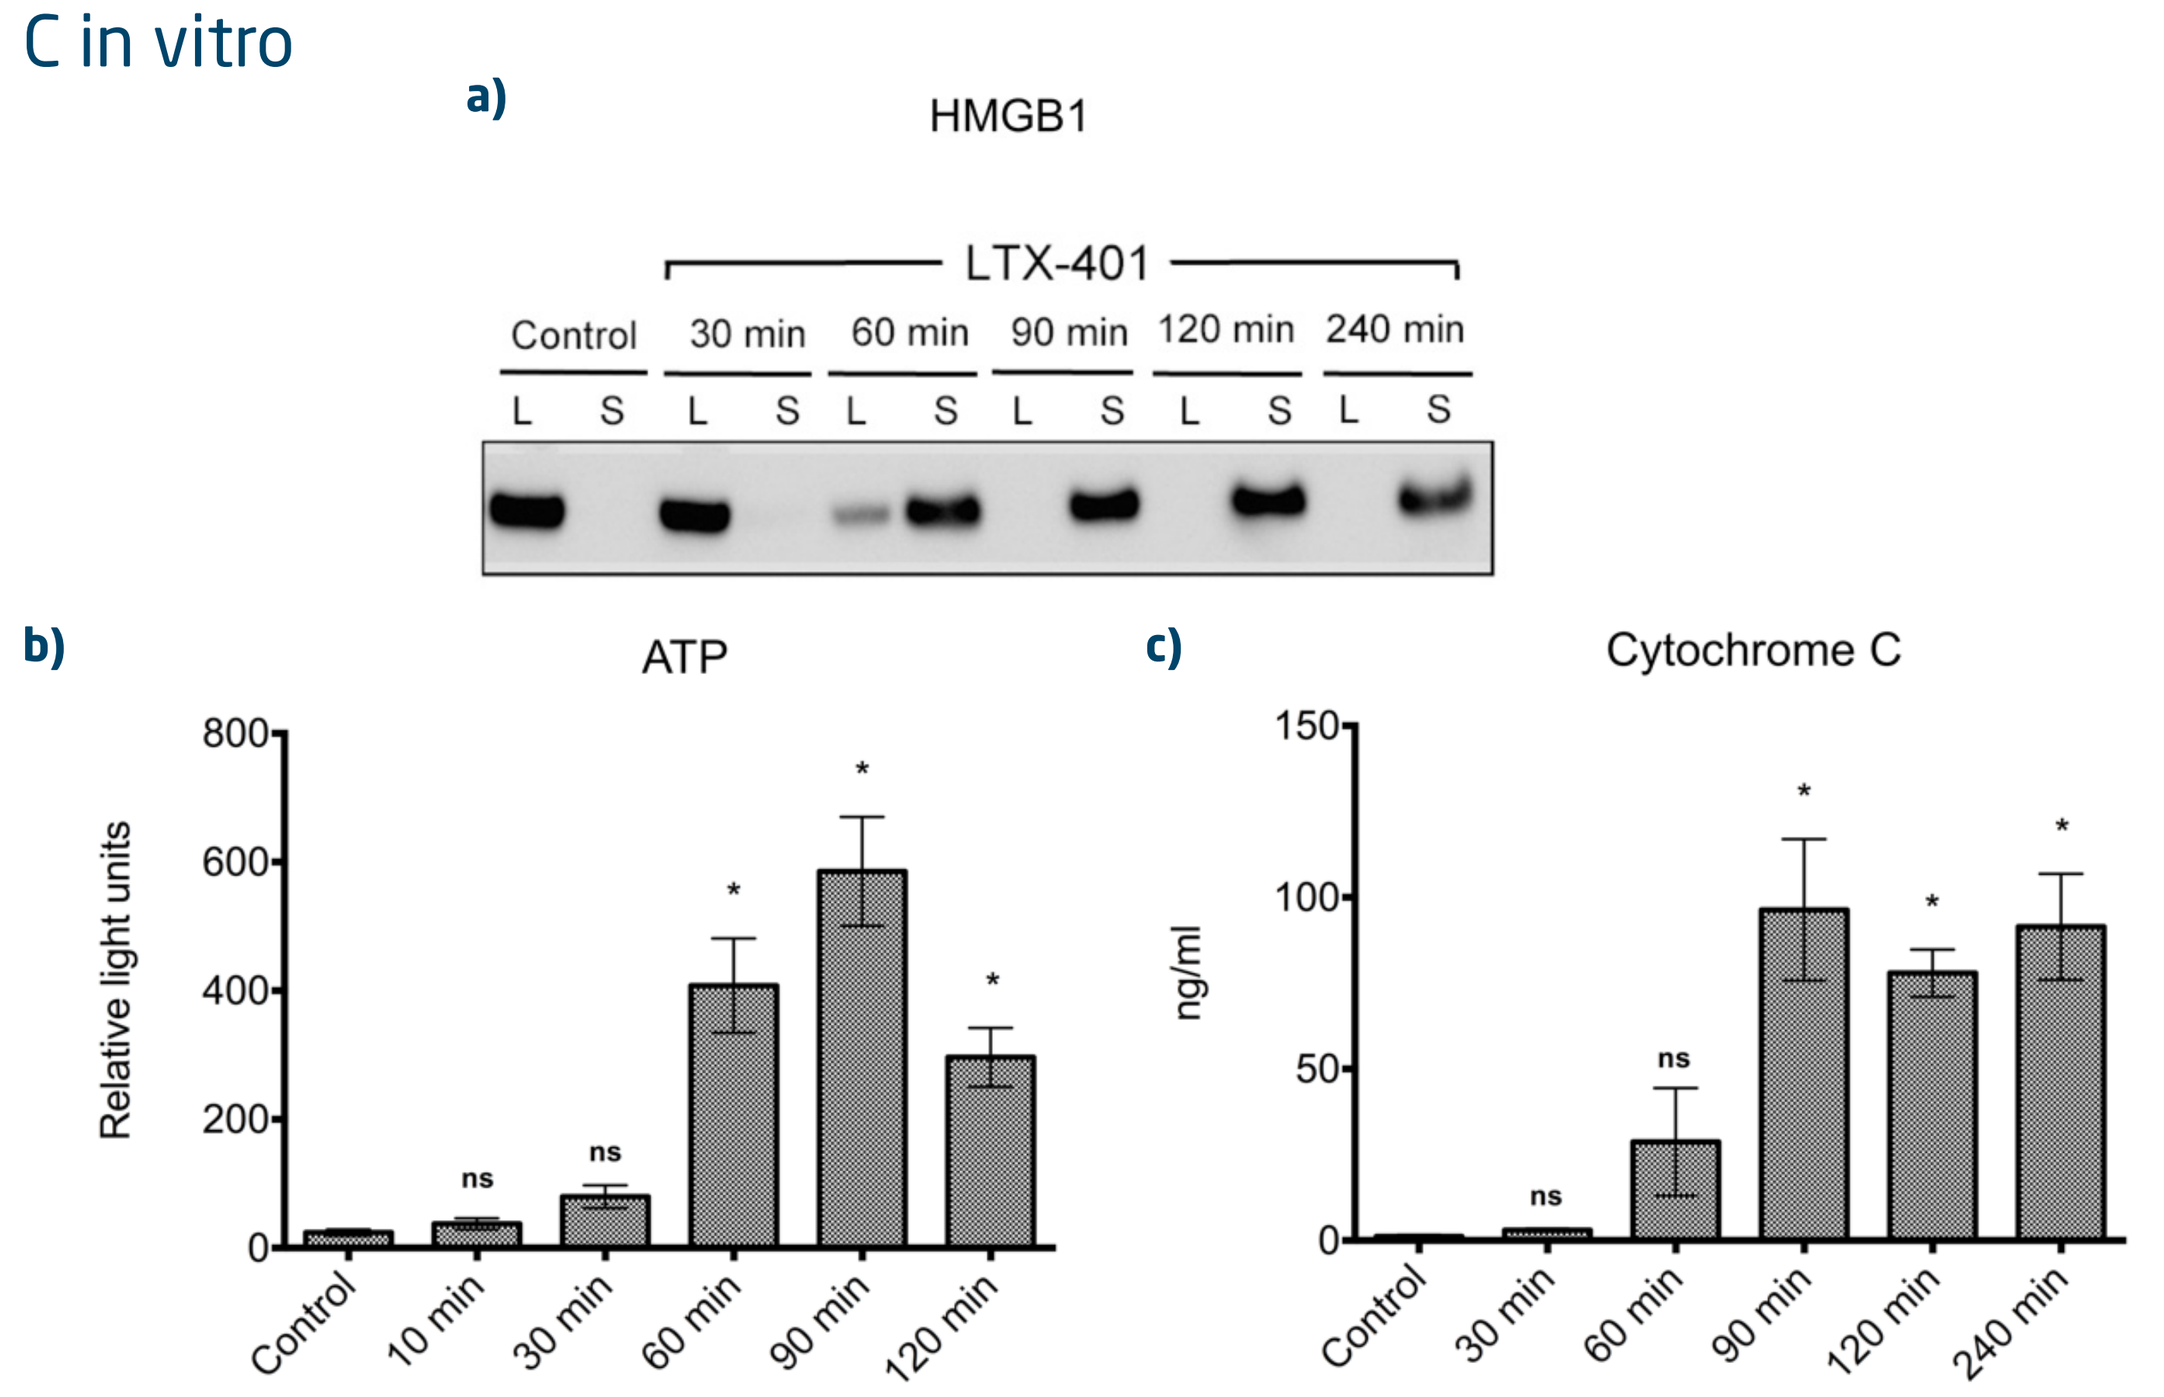
a)
HMGB1
LTX-401
Control 30 min 60 min 90 min 120 min 240 min
L S L S L S L S L S L S
b)
ATP
Relative light units
800 600 400 200 0
Control 10 min 30 min 60 min 90 min 120 min
ns ns * * *
c)
Cytochrome C
ng/ml
150 100 50 0
Control 30 min 60 min 90 min 120 min 240 min
ns ns * * *

(**a**) JM1 cells release HMGB1 from the lysate (L) to supernatant (S) after being stimulated with the 4 x IC_{50}^{4h} value of LTX-401 (108 µM). Protein is retained within lysates of control cells. (**b**) ATP is released from JM1 cells into supernatant following treatment with 2 x IC_{50}^{4h} value of LTX-401 (54 µM). (**c**) Release of cytochrome C into supernatant of LTX-401 treated JM1 cells (4 x IC_{50}^{4h}).*= $P<0.05$, ns = not significant

Fig. 1 - LTX-401 rapidly induces cell death in JM1 cells

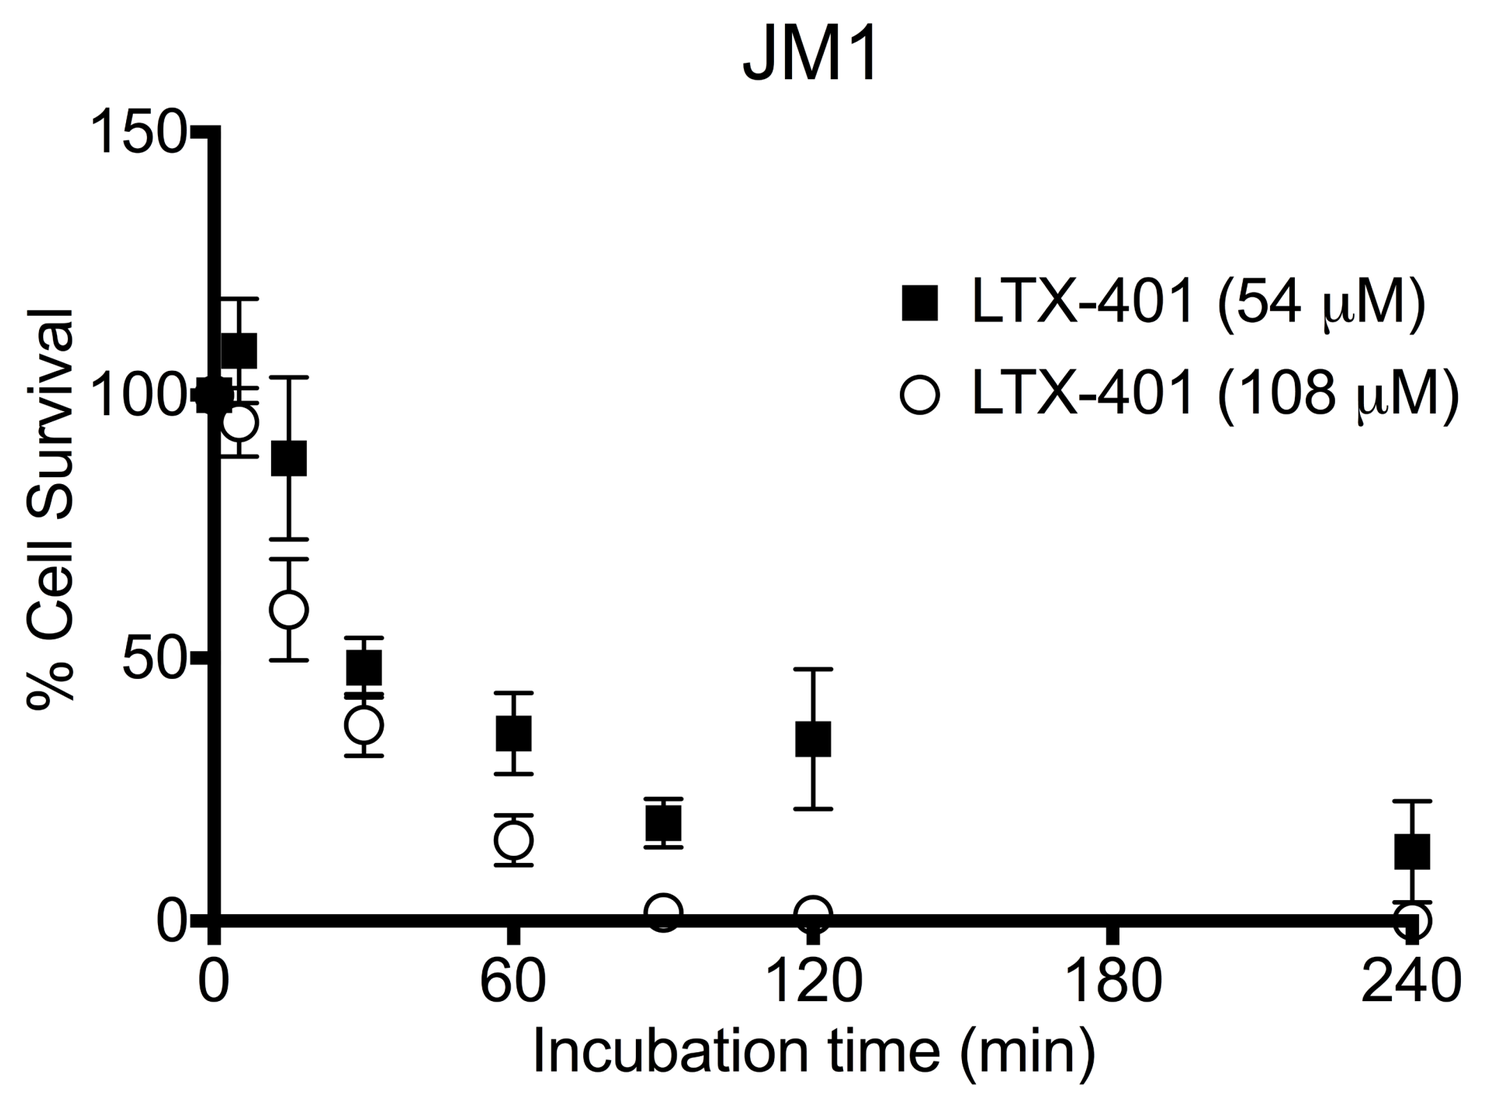
JM1
% Cell Survival
150 100 50 0
LTX-401 (54 µM)
LTX-401 (108 µM)
0 60 120 180 240
Incubation time (min)

Fig. 4 - Tumor treatment schedule

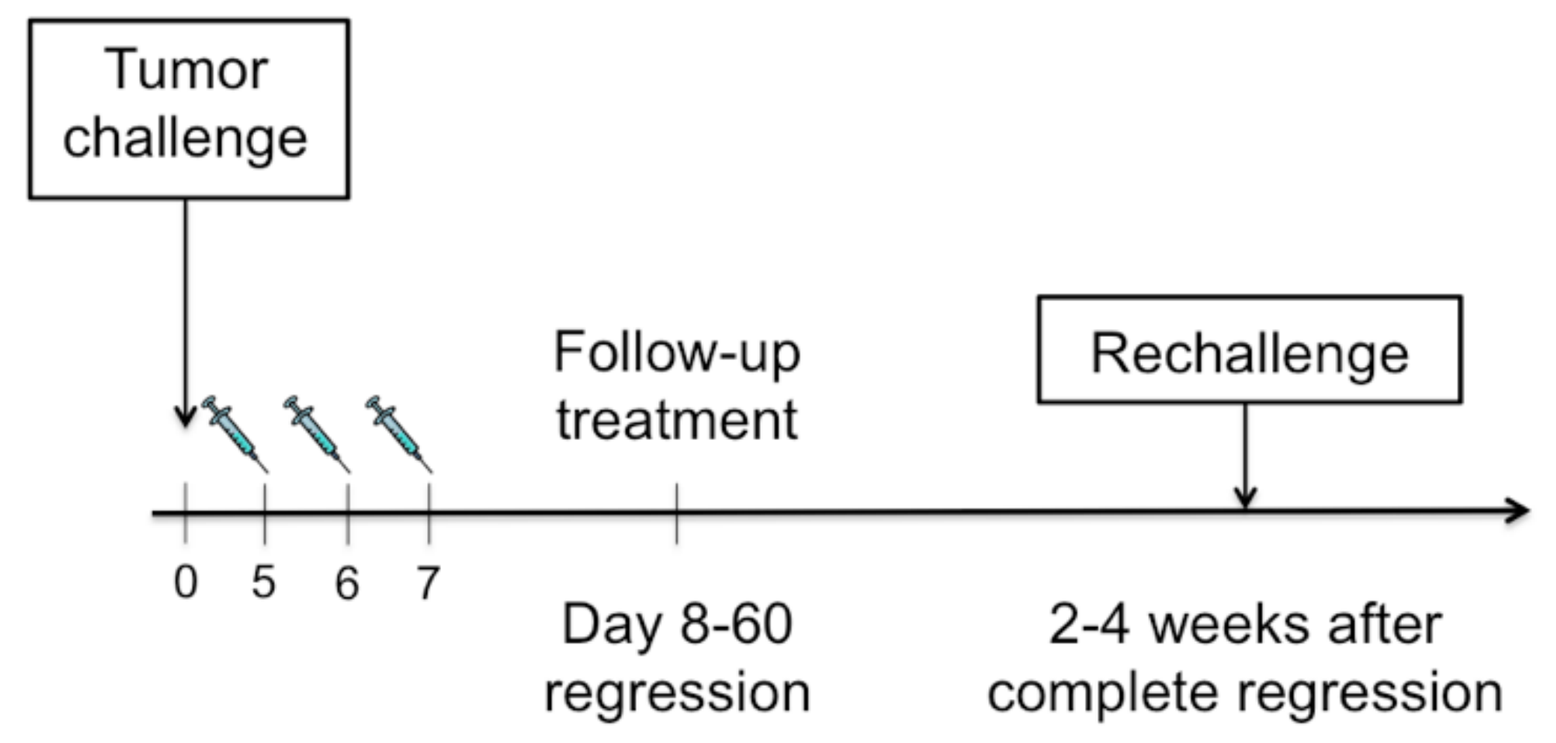
Tumor challenge
Follow-up treatment
Rechallenge
0 5 6 7
Day 8-60 regression
2-4 weeks after complete regression

Tumors were established in syngeneic Fisher 344 rats either by subcutaneous (s.c.) injection or direct intrahepatic (i.h.) injection. Palpable tumors were injected intratumorally with either 0.20 or 0.40 mg LTX-401 in 50 µl (or vehicle only, 0.9 % NaCl) once a day for three consecutive days. Alternatively, animals received a fourth and/or a fifth injection if there still was viable tumor tissue (follow-up treatment). Animals displaying complete regression were given a rechallenge with live JM1 cells, both by s.c. inoculation and direct i.h. injection

Fig. 2 - Treatment with LTX-401 leads to ultrastructural changes in JM1 cells

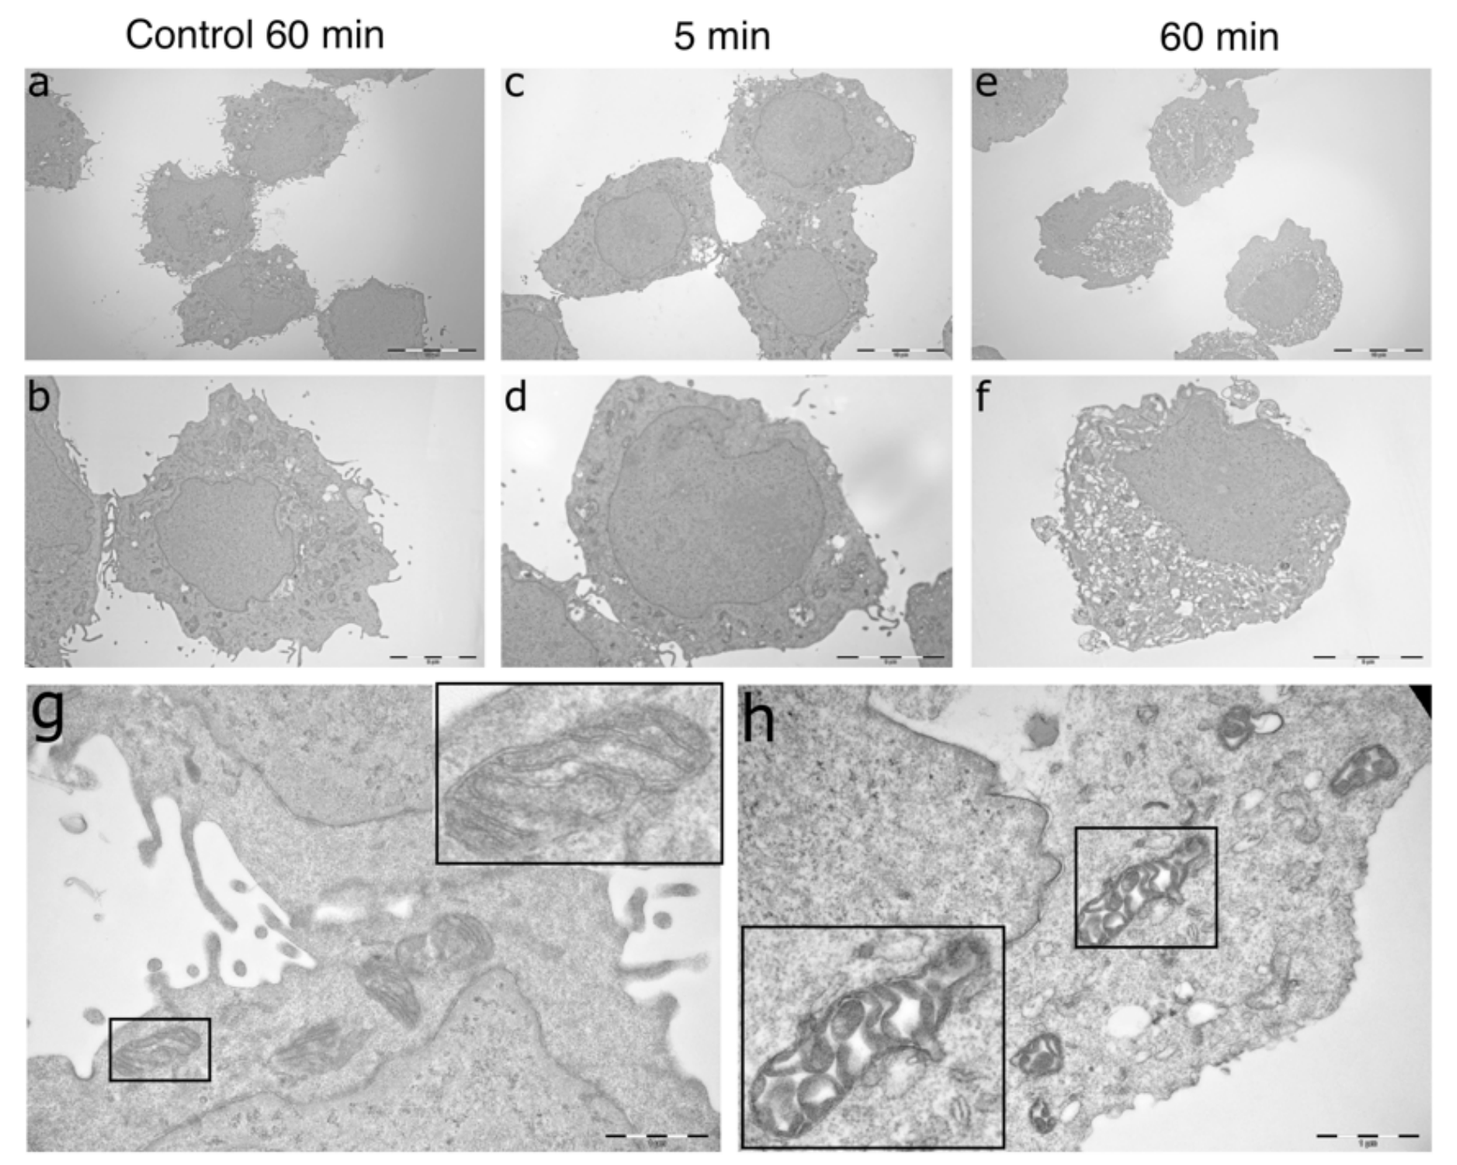
Control 60 min
5 min
60 min
a
c
e
b
d
f
g
h

Cells were treated with the 4 x IC504h of LTX-401 (108 µM) for various time points (5, 15, 30 and 60 min) before being fixed in a PHEM buffered malachite green fixative and prepared for electron microscopy studies. Untreated cells (**a**, **b** and **g**) were preserved in serum-free RPMI 1640 until experimental end-point (60 min) and compared with cells treated for 5 min (**c** and **d**) and 60 min (**e**, **f** and **h**). LTX-401-induced necrotic cell death was preceded by massive vacuolization of the cytoplasm and mitochondrial swelling with disarrangement of cristae structure.

Table 2 - *In vivo* efficacy of LTX-401

Group	n	[LTX-401]	Nr. of injections	Complete regression	Rechallenge (s.c. + i.h.)
Subcutaneous tumors					
1	7	0.20 mg	5	7/7	7/7
2	4	0.40 mg	3	3/4	3/3
Control	5	Vehicle	5	0/5	-
Intrahepatic tumors					
1	7	0.20 mg	3	0/7	-
2	7	0.40 mg	2	5/7	5/5
Control	3	Vehicle	3	0/5	-

Animals cured by LTX-401-treatment (i.e displaying complete regression) were protected against subsequent tumor growth when rechallenged with JM1 hepatocellular carcinoma cells. n = number of animals.

Conclusion

- LTX-401 displays potent anticancer activity against several cancer cell lines with selectivity compared to human red blood cells
- By inducing necrotic cell death, cancer cells are rendered immunogenic due to the release of DAMPs and tumor antigens
- Intratumoral treatment with LTX-401 induced complete regression of both subcutaneous and intrahepatic JM1 tumors followed by systemic protective immune responses, and thus represents a promising therapeutic approach against hepatocellular carcinoma

Reference

Eike et al. 2016. The Cytolytic Amphipathic ß(2,2)-Amino Acid LTX-401 Induces DAMP Release in Melanoma Cells and Causes Complete Regression of B16 Melanoma, PLoS One. 2016 Feb 16;11(2)

Oslo University Hospital

UiO : University of Oslo

University of Tromsø

CHILI. © WWW.CHILIGROUP.NO | 2016

Lytix Biopharma AS | P.O. Box 6447 | NO-9294 Tromsø, Norway | E-mail: post@lytixbiopharma.com | Phone: +47 77 67 55 00 | Fax: +47 77 67 55 01